IDWeek 2019 | Poster #1582

Delafloxacin Activity against Drug-Resistant *Streptococcus pneumoniae*, *Haemophilus influenzae*, *Haemophilus parainfluenzae*, and *Moraxella catarrhalis* from US Medical Centers (2014–2018)

Dee Shortridge, Jennifer M. Streit, Michael D. Huband, Rodrigo E. Mendes, Robert K. Flamm

JMI Laboratories, North Liberty, IA, USA

Introduction

- Delafloxacin is an anionic fluoroquinolone (FQ) antimicrobial that was approved in 2017 by the United States (US) Food and Drug Administration for the treatment of acute bacterial skin and skin structure infections
- Delafloxacin recently completed a successful clinical trial for treatment of community-acquired bacterial pneumonia (CABP)
- In the present study, *in vitro* susceptibility (S) results for delafloxacin and comparator agents were determined for CABP pathogens, including *Streptococcus pneumoniae*, *Haemophilus influenzae*, *Haemophilus parainfluenzae*, and *Moraxella catarrhalis* clinical isolates from US hospitals participating in the SENTRY Antimicrobial Surveillance Program during 2014–2018
- We also examined the activity of delafloxacin and quinolone comparators against various antibiotic-resistant phenotypes, including erythromycin and levofloxacin-resistant (R) *S. pneumoniae* and *S. pneumoniae* nonsusceptible to 3 or more first line therapies

Materials and Methods

- A total of 1,978 *S. pneumoniae*, 1,128 *H. influenzae*, 684 *M. catarrhalis*, and 43 *H. parainfluenzae* isolates were collected from community-acquired respiratory tract infections (CARTI) during 2014–2018 from US hospitals
- Sites included only 1 isolate/patient/infection episode, and isolate identifications were confirmed at JMI Laboratories
- Susceptibility testing was performed according to CLSI broth microdilution methodology, and CLSI (2019) breakpoints were applied, where applicable
- Other antimicrobials tested included levofloxacin and moxifloxacin (not tested in 2015)
- Multidrug-resistant (MDR) *S. pneumoniae* isolates were categorized as being nonsusceptible (NS) to amoxicillin-clavulanate, erythromycin, and tetracycline
 - Other resistant phenotypes were erythromycin-resistant, tetracycline-resistant, levofloxacin-NS, or penicillin-NS
- β-lactamase presence was determined for *Haemophilus* spp. and *M. catarrhalis*
- Levofloxacin-R *S. pneumoniae* isolates were sequenced and the quinolone-resistance determining region (QRDR) was examined to identify the mutations in gyrase (GyrA/B) and topoisomerase (ParC/E) genes responsible for the levofloxacin resistance
 - Select isolates at the levofloxacin-susceptible breakpoint of 2 mg/L were also sequenced

Table 1 Activity of delafloxacin, levofloxacin, and moxifloxacin against *S. pneumoniae*, *H. influenzae*, *H. parainfluenzae*, and *M. catarrhalis* with various resistant phenotypes

Organism (n) Phenotype (n)	Delafloxacin $MIC_{50/90}$ (mg/L)	Levofloxacin $MIC_{50/90}$ (mg/L)	Moxifloxacin $MIC_{50/90}$ (mg/L, n[a])
S. pneumoniae (1,978)	0.015/0.03	1/1	≤0.12/0.25 (1,687)
ERY-R (933)	0.015/0.03	1/1	≤0.12/0.25 (804)
MDR (85)	0.03/0.03	1/2	≤0.12/0.25 (75)
PEN-NS[b] (746)	0.015/0.03	1/1	≤0.12/0.25 (638)
LEV-R (14)	0.12/0.12	>4/>4	2/2 (11)
H. influenzae (1,128)	≤0.001/0.002 (1,069)	≤0.015/0.03	0.03/0.06 (965)
BL positive (363)	≤0.001/0.002 (346)	≤0.015/0.03	0.03/0.06 (318)
LEV-R (5)	0.25/-	>2/-	>2/-
H. parainfluenzae (43)	0.008/0.015 (42)	0.03/0.12	0.12/0.25 (10)
M. catarrhalis (684)	0.004/0.008 (682)	0.03/0.06	0.06/0.06 (598)
BL positive (589)	0.004/0.008 (587)	0.03/0.06	0.06/0.06 (585)

ERY-R, erythromycin-resistant; MDR, multidrug-resistant (amoxicillin-clavulanate, erythromycin, and tetracycline-NS); PEN-NS, penicillin nonsusceptible; LEV-R, levofloxacin resistant; BL, β-lactam.
[a]Number of isolates shown for moxifloxacin, not tested in 2015.
[b]Oral breakpoints: susceptible ≤0.06; intermediate 0.12-1.0; resistant ≥2 mg/L (CLSI M100, 2019).

Results

- The activities of the 3 fluoroquinolones against CARTI pathogens are shown in Table 1
 - Figure 1 shows the MIC distribution for delafloxacin against all isolates
- The most active fluoroquinolone against *S. pneumoniae* was delafloxacin with $MIC_{50/90}$ values of 0.015/0.03 mg/L
- Only 14 *S. pneumoniae* isolates were levofloxacin resistant (0.7%)
 - Levofloxacin-resistant isolates had delafloxacin $MIC_{50/90}$ values of 0.12/0.12 mg/L (Table 1)
 - All isolates with levofloxacin MIC values >4 mg/L had both gyrase and topoisomerase mutations, suggesting that levofloxacin resistance requires at least 2 mutations (Table 2)
 - The most common mutations were in GyrA and ParC as previously described
 - GyrB and ParE mutations were less common
 - 15 isolates with a susceptible levofloxacin MIC of 2 mg/L were also sequenced (data not shown)
 - 14 lacked any QRDR mutations; 1 isolate had a ParC substitution (S79Y)
 - Delafloxacin $MIC_{50/90}$ values were 0.03/0.06 mg/L
- Delafloxacin activities were similar when tested against the MDR, erythromycin, or penicillin-NS for *S. pneumoniae* phenotypes (Table 1)
 - Erythromycin susceptibility was 52.1% and clindamycin susceptibility was 84.4%
 - Tetracycline susceptibility was 78.1%
 - 37.7% of *S. pneumoniae* isolates were penicillin-nonsusceptible
 - The rate of MDR *S. pneumoniae* was 4.3%
- Distribution of *S. pneumoniae* resistant to erythromycin or tetracycline or nonsusceptible to penicillin by US census region is shown in Figure 2
 - All 3 resistant phenotypes were most frequently found in the West South Central region and were the lowest in the Pacific region
- Delafloxacin was the most active fluoroquinolone against *H. influenzae*, *H. parainfluenzae*, and *M. catarrhalis* (Table 1 and Figure 1)
 - Only 5 *H. influenzae* (0.4%) and 1 *H. parainfluenzae* were levofloxacin-NS with a delafloxacin MIC_{50} value of 0.25 mg/L (Table 1)
 - There were no levofloxacin-NS *M. catarrhalis*
 - 363 *H. influenzae* were β-lactamase positive (32.2%)
 - Only 2/43 (4.7%) *H. parainfluenzae* isolates were β-lactamase positive
 - Most *M. catarrhalis* were β-lactamase positive (86.1%)
 - β-lactamase presence did not affect fluoroquinolone MIC values (Table 1)

Conclusions

- Delafloxacin demonstrated potent *in vitro* antibacterial activity against *S. pneumoniae*, *H. influenzae*, *H. parainfluenzae*, and *M. catarrhalis* from the United States
- Delafloxacin was active against MDR *S. pneumoniae* isolates that were NS to the agents commonly used as treatments for CABP (erythromycin, amoxicillin/clavulanate, and tetracycline)
 - Erythromycin resistant *S. pneumoniae* was common at 47.2%
 - Tetracycline resistance was 21.6%
- Delafloxacin had excellent activity against levofloxacin-R *S. pneumoniae* and levofloxacin-NS *H. influenzae* isolates
 - Levofloxacin resistance in *S. pneumoniae* was 0.7%
 - Levofloxacin resistance in *H. influenzae* was 0.4%
- These data support delafloxacin as a potential treatment for CABP

Figure 1 Delafloxacin MIC distribution for CARTI pathogens

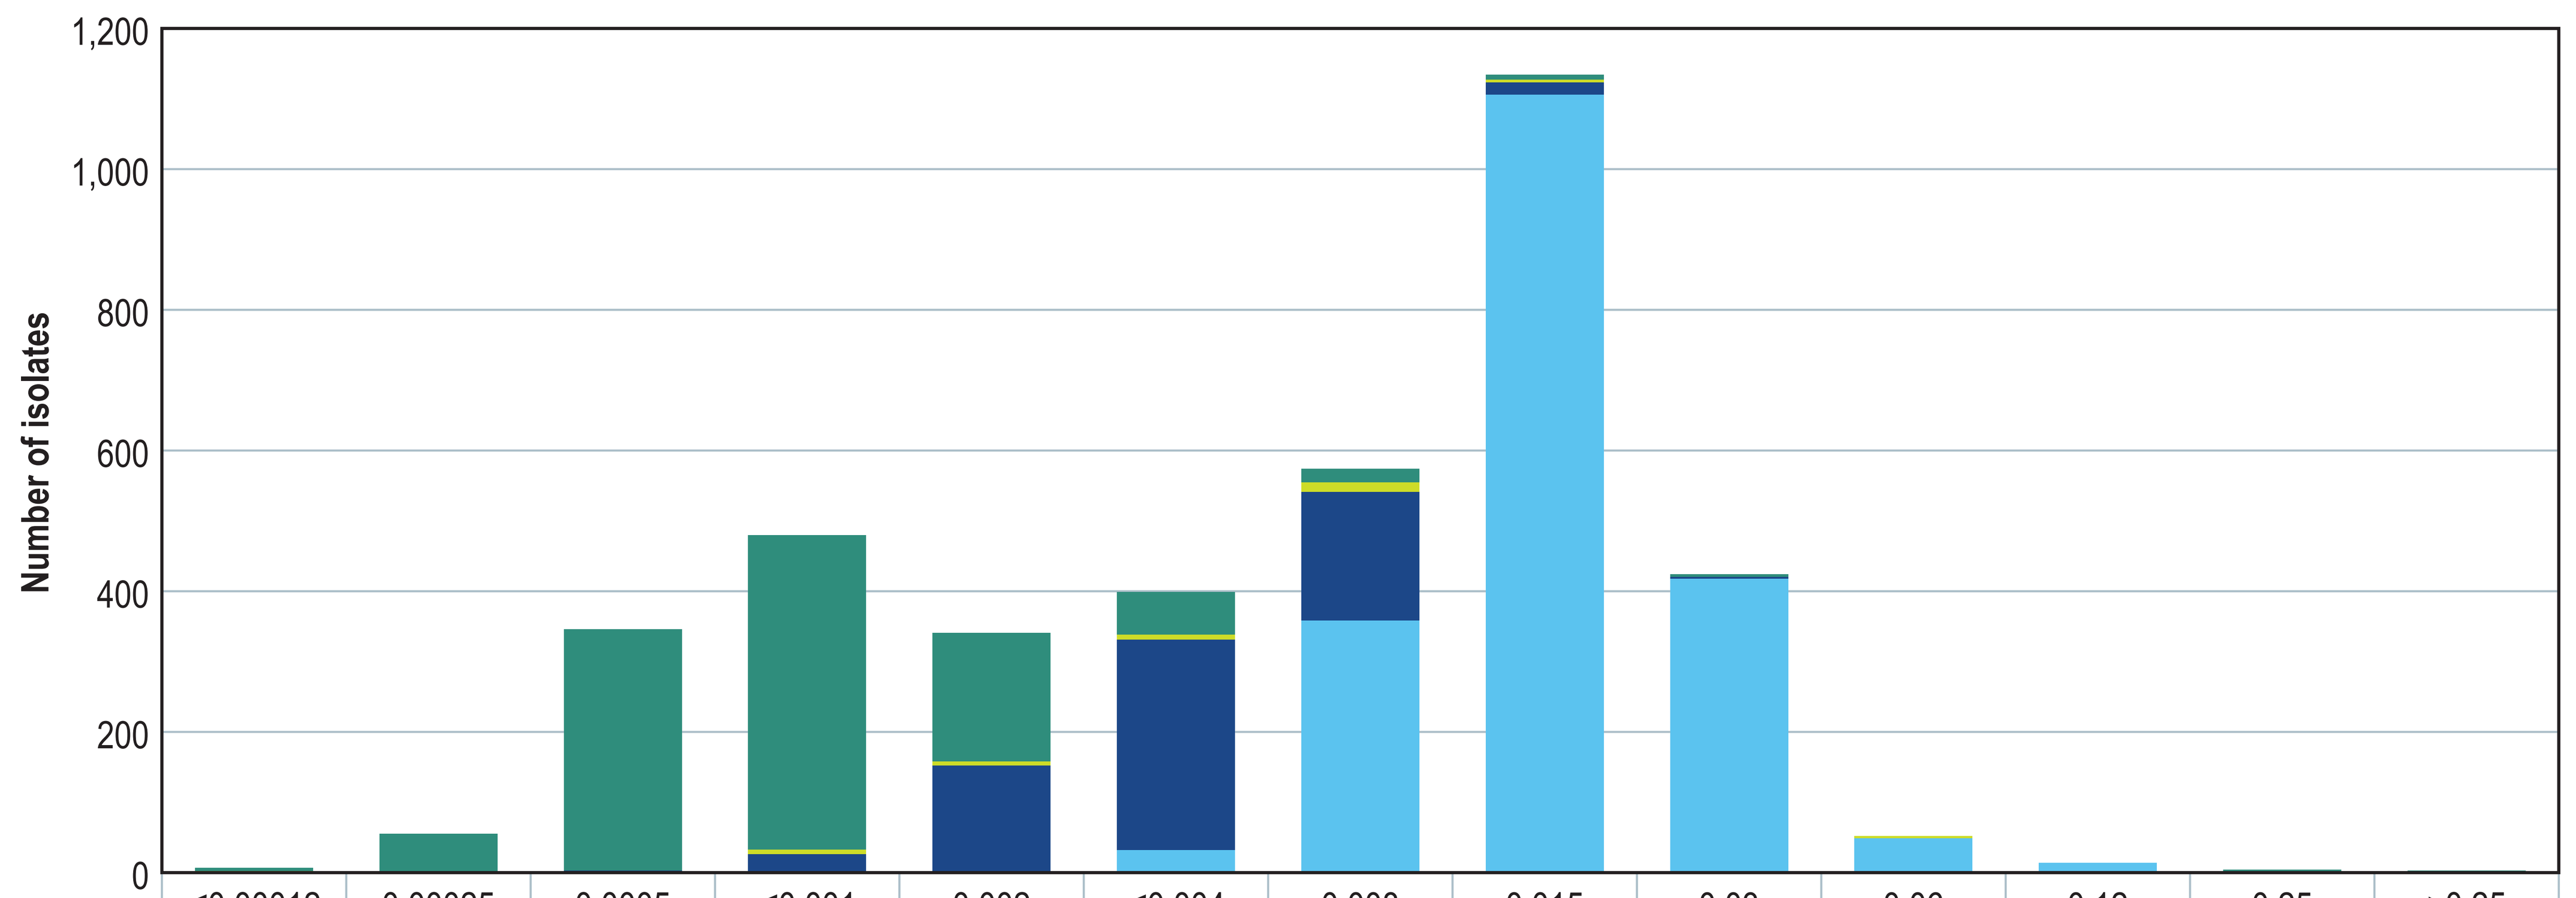

Number of isolates
1,200
1,000
800
600
400
200
0

	≤0.00012	0.00025	0.0005	≤0.001	0.002	≤0.004	0.008	0.015	0.03	0.06	0.12	0.25	>0.25
Haemophilus influenzae	5	54	343	447	183	61	19	7	4			3	2
Haemophilus parainfluenzae	1			7	6	7	14	4		3			1
Moraxella catarrhalis	1	1	3	26	152	299	183	17	2				
Streptococcus pneumoniae						32	358	1106	418	49	14	1	

Delafloxacin MIC (mg/L)

CARTI, community-acquired respiratory tract infection

Figure 2 *S. pneumoniae* resistant phenotypes by census division

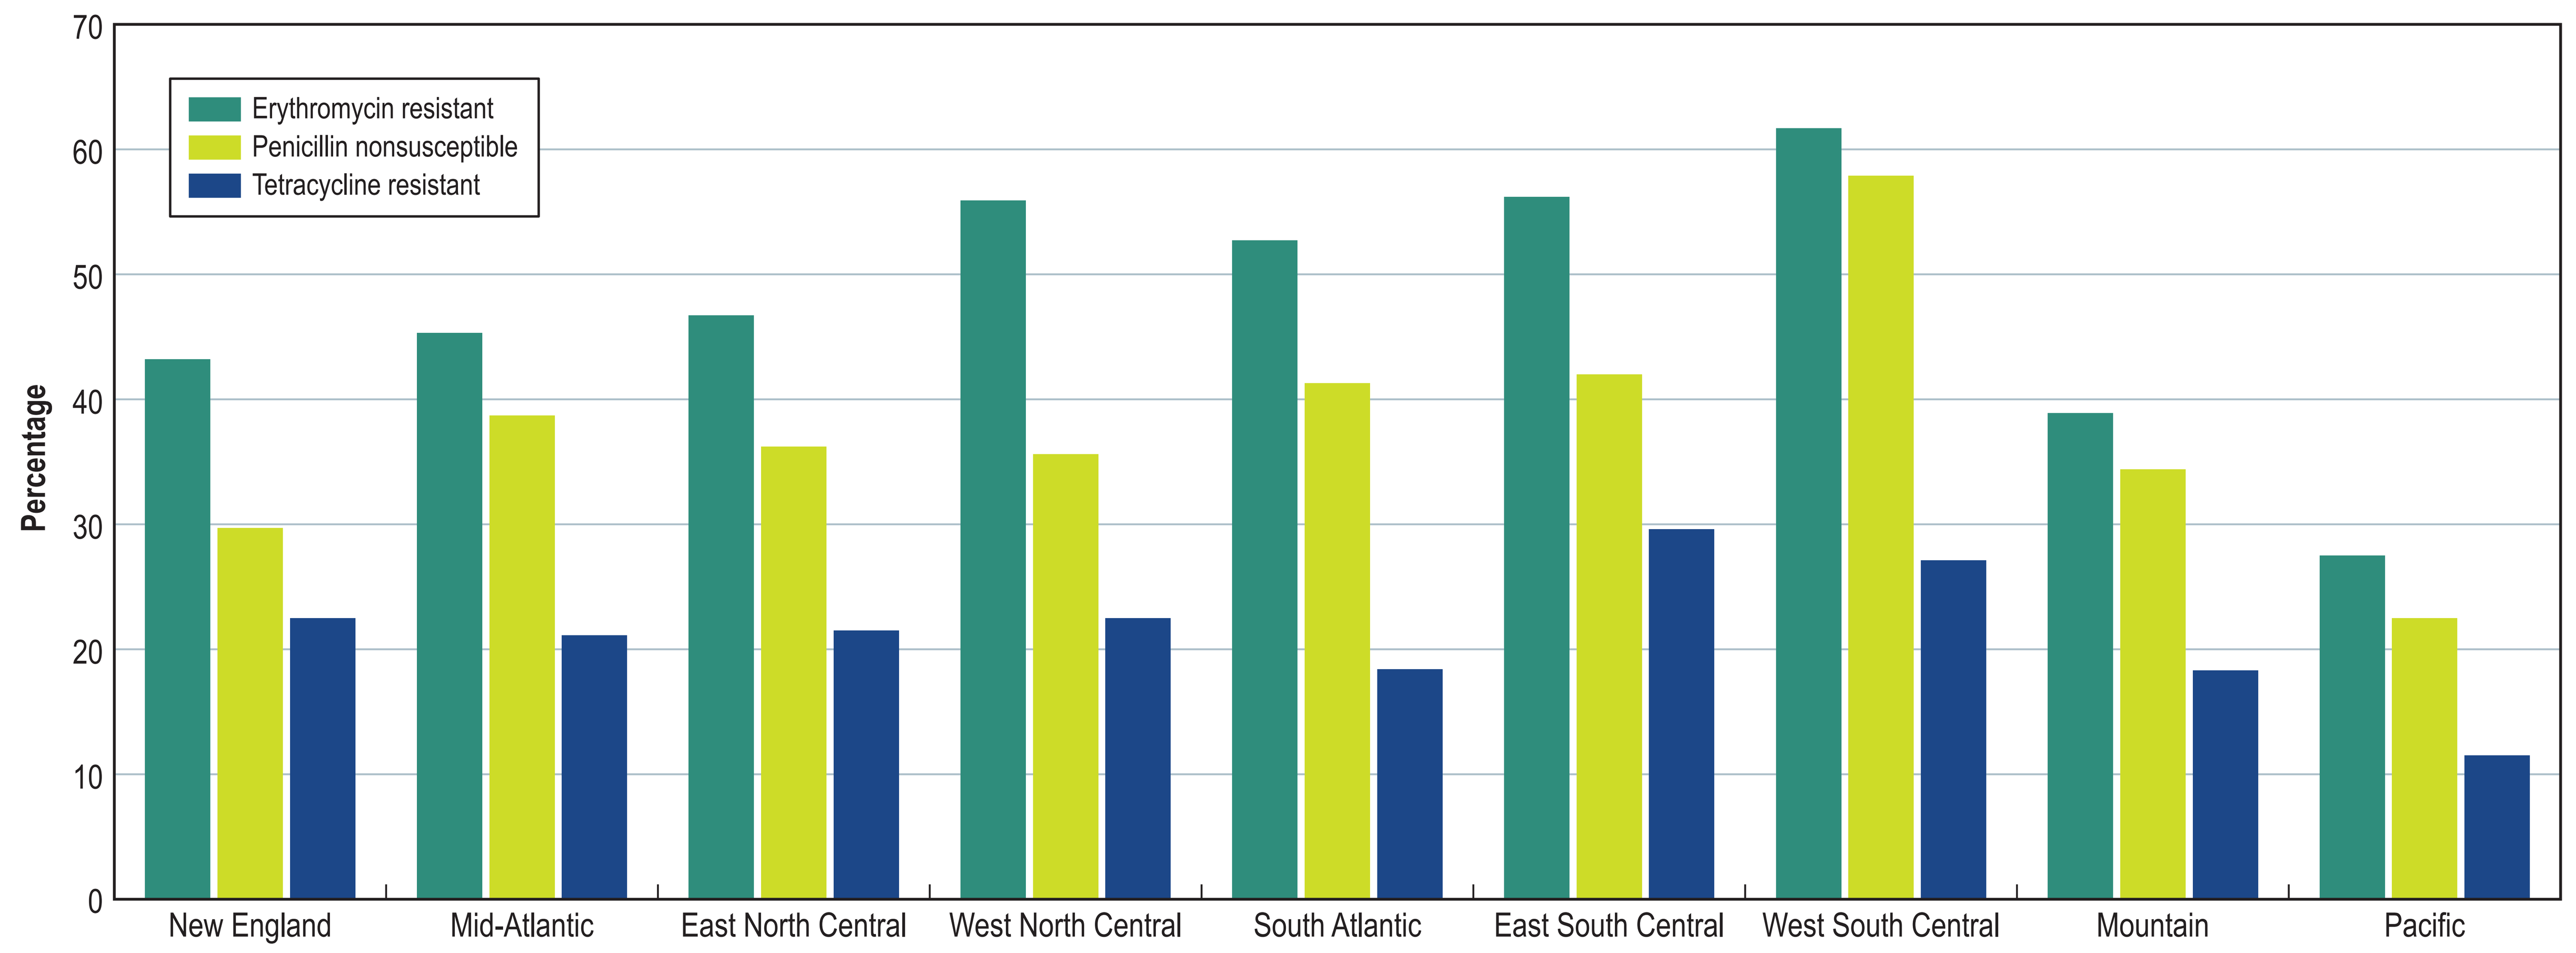

Erythromycin resistant
Penicillin nonsusceptible
Tetracycline resistant
Percentage
70
60
50
40
30
20
10
0
New England
Mid-Atlantic
East North Central
West North Central
South Atlantic
East South Central
West South Central
Mountain
Pacific

Table 2 Levofloxacin-resistant *S. pneumoniae* isolates with delafloxacin MIC values and quinolone-resistance mutations

Collection number	Organism	Delafloxacin MIC (mg/L)	Levofloxacin MIC (mg/L)	GyrA	GyrB	ParC	ParE
853694	*S. pneumoniae*	0.12	>4	S81F	WT	D83Y	WT
900190	*S. pneumoniae*	0.12	>4	S81Y	WT	S79F	WT
901469	*S. pneumoniae*	0.06	>4	S81F	WT	D83Y	WT
912320	*S. pneumoniae*	0.12	>4	S81F	WT	S79F	WT
935440	*S. pneumoniae*	0.12	>4	S81Y	WT	WT	D435N
936185	*S. pneumoniae*	0.12	>4	S81F	WT	S79F	WT
939459	*S. pneumoniae*	0.25	>4	WT	D435N	S79F	WT
942156	*S. pneumoniae*	0.12	>4	E85K	WT	S79Y	WT
960803	*S. pneumoniae*	0.12	>4	S81F	WT	S79F	WT
987525	*S. pneumoniae*	0.06	>4	S81F	WT	S79F	WT
993907	*S. pneumoniae*	0.06	>4	S81F	WT	D83N	WT
994209	*S. pneumoniae*	0.06	>4	WT	D435N	S79Y	WT
1025770	*S. pneumoniae*	0.12	>4	S81F	WT	S79F	WT
1049825	*S. pneumoniae*	0.12	>4	S81F	WT	S79F	WT

GyrA, gyrase A; GyrB, gyrase B; ParC, topoisomerase C; ParE, topoisomerase E; WT, wild type

Acknowledgements

This study was supported by Melinta Therapeutics, Inc.

References

Clinical and Laboratory Standards Institute (2018). *M07 Eleventh edition. Methods for dilution antimicrobial susceptibility testing for bacteria that grow aerobically*, Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2015). *M45 Third edition. Methods for antimicrobial dilution and disk susceptibility testing of infrequently isolated or fastidious bacteria*, Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement.* Wayne, PA: CLSI.

Murata M, Kosai K, Yamauchi S, Sasaki D, Kaku N, Uno N, Morinaga Y, Hasegawa H, Miyazaki T, Izumikawa K, Mukae H, Yanagihara, K (2018). In vitro activity of lascufloxacin against *Streptococcus pneumoniae* with mutations in the quinolone resistance-determining regions. *Antimicrob Agents Chemother* 62:e01971-17.

Contact

Dee Shortridge, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: dee-shortridge@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2019-delafloxacin-CARTI.pdf

Charges may apply. No personal information is stored.

IDWeek 2019, October 2–6, 2019, Washington, DC