IDWeek 2023 | Poster #2754

Delafloxacin and Comparator Fluoroquinolones *In Vitro* Resistance Trends in Isolates from Skin and Skin Structure Infections in the USA (2017–2022)

Michael D. Huband, Dee Shortridge, Cecilia G. Carvalhaes, Leonard Duncan, Mariana Castanheira
JMI Laboratories, North Liberty, Iowa, United States

Introduction

- Delafloxacin (DLX) is a broad-spectrum fluoroquinolone approved in 2017 for the treatment of acute bacterial skin and skin structure infection (ABSSSI) caused by multiple Gram-positive pathogens, including *Staphylococcus aureus* (methicillin-susceptible [MSSA] and methicillin-resistant [MRSA]), *S. haemolyticus*, *S. lugdunensis*, *Streptococcus agalactiae*, *S. anginosus* group, *S. pyogenes*, *Enterococcus faecalis* (vancomycin-susceptible), and Gram-negative organisms, including *Enterobacter cloacae* species complex, *Escherichia coli*, *Klebsiella pneumoniae*, and *Pseudomonas aeruginosa*.
- Delafloxacin is also approved to treat community-acquired bacterial pneumonia (CABP) caused by MSSA, *Streptococcus pneumoniae*, *Haemophilus influenzae*, *H. parainfluenzae*, *E. coli*, *K. pneumoniae*, and *P. aeruginosa*.
- In this study, the *in vitro* susceptibilities of delafloxacin and comparator fluoroquinolones, levofloxacin (LEV), moxifloxacin (MOX), and ciprofloxacin (CIP) were determined for US clinical isolates collected from skin and skin structure infections (SSSI) during 2017–2022.

Materials and Methods

- 16,039 bacterial isolates from SSSI were consecutively collected at 77 US medical centers participating in the SENTRY Antimicrobial Surveillance Program (2017–2022). The prevalence of SSSI pathogens by species are shown in Figure 1.
- Medical centers submitted 1 isolate per patient per infection episode.
- Isolate identification was determined at each site and confirmed at JMI Laboratories using MALDI-TOF MS.
- Susceptibility testing was performed according to CLSI M07 (2018) broth microdilution methodology.
- Current (2023) FDA breakpoint interpretive criteria were applied for delafloxacin; CLSI (2023) interpretive criteria were applied to comparator agents. Moxifloxacin was tested against Gram-positives and ciprofloxacin against Gram-negative bacterial species.

Results

- The most common SSSI organisms were *S. aureus* (n=8,540), including 4,967 MSSA (42%) and 3,573 MRSA (58%), *P. aeruginosa* (n=1,085), *E. coli* (n=840), *S. pyogenes* (n=781), *Klebsiella* spp. (n=781), enterococci (n=571), and *Enterobacter* spp. (n=473) (Figure 1).
- The percent susceptible (%S) by year (2017–2022 and all years combined) for delafloxacin, levofloxacin, moxifloxacin, and ciprofloxacin are shown in Table 1.
- Susceptibility of MSSA to delafloxacin remained high (96.9%–98.3%) throughout the surveillance period.
- For MRSA, %S to delafloxacin increased from 81.0% to 87.0% over 6 years (Figure 2), whereas %S to levofloxacin and moxifloxacin declined from 36.5% to 34.7% and 36.8% to 34.7%, respectively (Table 1).
- 100% of *Streptococcus agalactiae*, *S. anginosus* group, and *S. pyogenes* isolates were susceptible to delafloxacin (2017–2022) (Table 1 and Figure 3).
- Overall susceptibility of *S. lugdunensis* isolates to delafloxacin was 98.9% (Table 1 and Figure 3).
- Susceptibility of *S. haemolyticus* isolates to fluoroquinolones (DLX, LEV, MOX) was low, averaging 56.2% for the surveillance period (Table 1 and Figure 3).
- Delafloxacin susceptibility against *Enterococcus faecalis* increased from 64.6% to 80.3% from 2017–2022 (Figure 2) with smaller increases observed for levofloxacin (4.9%) and moxifloxacin (3.4%).
- Delafloxacin susceptibility against *Pseudomonas aeruginosa* increased from 64.9% to 73.7% from 2017 to 2022 (Figure 2), as did susceptibility to levofloxacin (69.5% to 74.7%) (Table 1). *P. aeruginosa* susceptibility to ciprofloxacin declined slightly from 82.5% to 79.8% from 2017 to 2022 (Table 1).
- Delafloxacin susceptibility against *E. coli* increased from 59.4% to 74.4% from 2017 to 2022 (Figure 2), and also increased for levofloxacin (66.7% to 75.2%) and ciprofloxacin (61.8% to 76.0%) (Table 1).
- Overall susceptibility of *Enterobacter cloacae* species complex isolates to delafloxacin, levofloxacin, and ciprofloxacin (2017 to 2022) was 84.2%, 92.3%, and 91.2%, respectively (Table 1 and Figure 4).

Conclusions

- An increase in fluoroquinolone susceptibility (2017–2022) was observed for SSSI pathogens including MRSA, *E. faecalis*, *E. coli*, and *P. aeruginosa*.
- Susceptibility of MSSA to delafloxacin remained high (96.9%–98.3%) from 2017–2022.
- Susceptibility of MRSA to delafloxacin increased from 81.0% to 87.0% over the period of 2017 to 2022, whereas susceptibility to levofloxacin and moxifloxacin declined.
- High delafloxacin susceptibility rates (>98.9% susceptible) were observed among SSSI pathogens from the following organism groups: *S. lugdunensis*, *S. agalactiae*, *S. anginosus* group, and *S. pyogenes*.
- The decreased use of FQs may have led to improved susceptibility for DLX and as such it remains a good treatment option for SSSI.

Funding

This study was supported by Melinta Therapeutics. MD Huband, D Shortridge, CG Carvalhaes, L Duncan, and M Castanheira are/were employees of JMI Laboratories at the time of this study and were paid consultants to Melinta in connection with the development of this poster.

Acknowledgments

The authors thank the participating medical centers for providing the bacterial isolates.

Figure 1. Prevalence of SSSI pathogens in the United States (2017–2022)

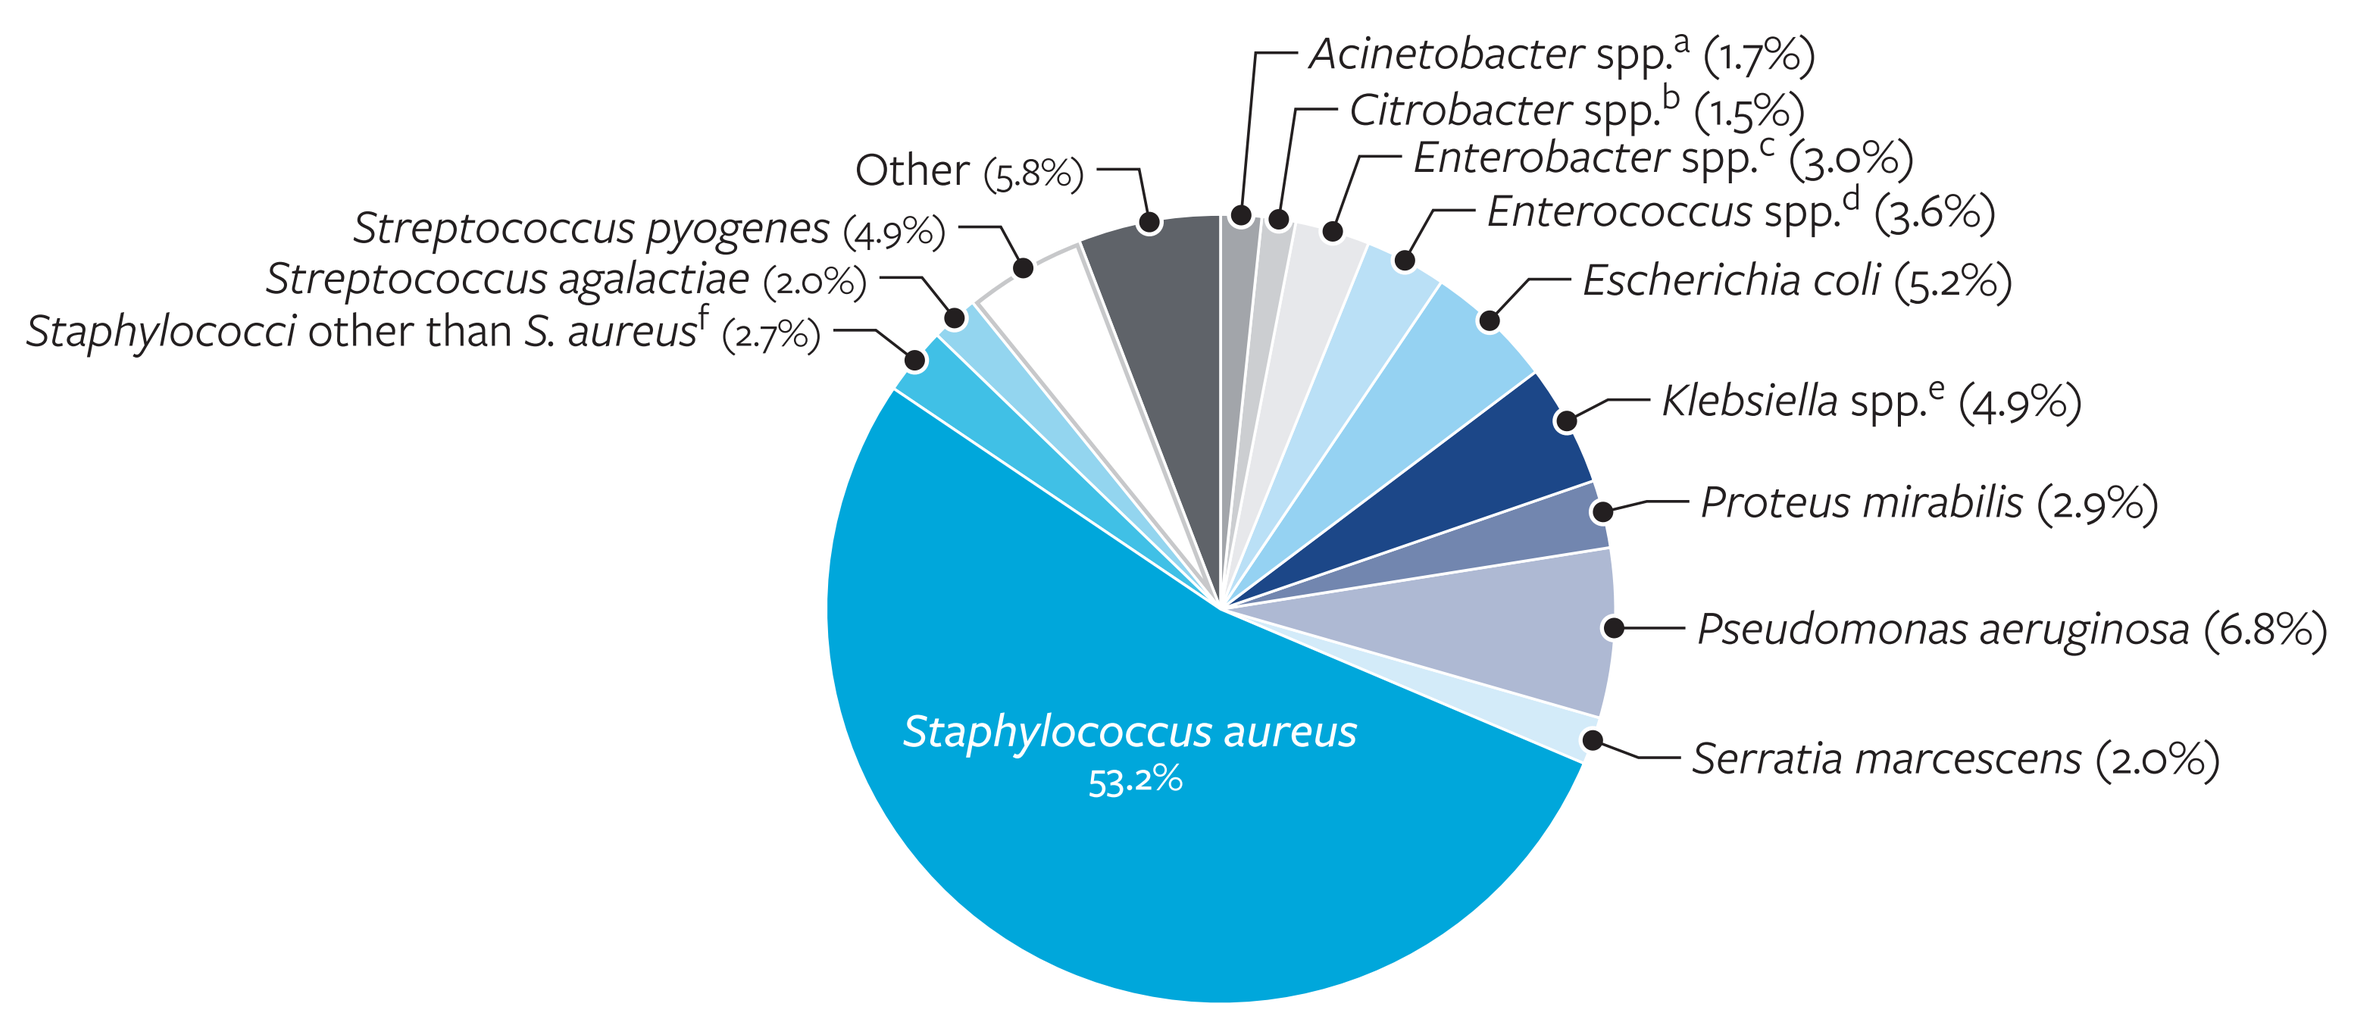
Acinetobacter spp.[a] (1.7%)
Citrobacter spp.[b] (1.5%)
Enterobacter spp.[c] (3.0%)
Enterococcus spp.[d] (3.6%)
Escherichia coli (5.2%)
Klebsiella spp.[e] (4.9%)
Proteus mirabilis (2.9%)
Pseudomonas aeruginosa (6.8%)
Serratia marcescens (2.0%)
Staphylococcus aureus 53.2%
Staphylococci other than S. aureus[f] (2.7%)
Streptococcus agalactiae (2.0%)
Streptococcus pyogenes (4.9%)
Other (5.8%)

[a] Contains *Acinetobacter baumannii calcoaceticus* species complex (229); *A berezinae* (6), *A courvalinii* (3), *A. haemolyticus* (2), *A. johnsonii* (2), *A junii* (2), *A. lwoffii* (7), *A. proteolyticus* (1), *A. radioresistens* (10), *A. ursingii* (12), *A. variabilis* (1).
[b] Contains *Citrobacter amalonaticus* (1), *C. amalonaticus/farmeri* (5), *C. braakii* (1), *C. freundii* species complex (128), *C. koseri* (98).
[c] Contains *Enterobacter asburiae* (1), *E. cancerogenus* (1), *E. cloacae* species complex (443), *E. hormaechei* (3).
[d] Contains *Enterococcus avium* (19), *E. avium/raffinosus/gilvus* (1), *E. casseliflavus* (5), *E. faecalis* (44), *E. faecium* (117), *E. gallinarum* (3), *E. mundtii* (1), *E. raffinosus* (6).
[e] Contains *Klebsiella aerogenes* (105), *K. oxytoca* (222), *K. pneumoniae* (451), *K. variicola* (3).
[f] Contains *Staphylococcus capitis* (7), *S. caprae* (9), *S. epidermidis* (174), *S. haemolyticus* (16), *S. hominis* (9), *S. intermedius* (3), *S. lugdunensis* (188), *S. pettenkoferi* (1), *S. pseudintermedius* (13), *S. pseudintermedius/intermedius/delphini* (1), *S. saprophyticus* (2), *S. schleiferi* (2), *S. scuri* (2), *S. simulans* (17), *S. warneri* (4), *S. xylosus* (1).

Figure 2. Susceptibility of delafloxacin against Gram-positive and Gram-negative SSSI pathogens (2017–2022)

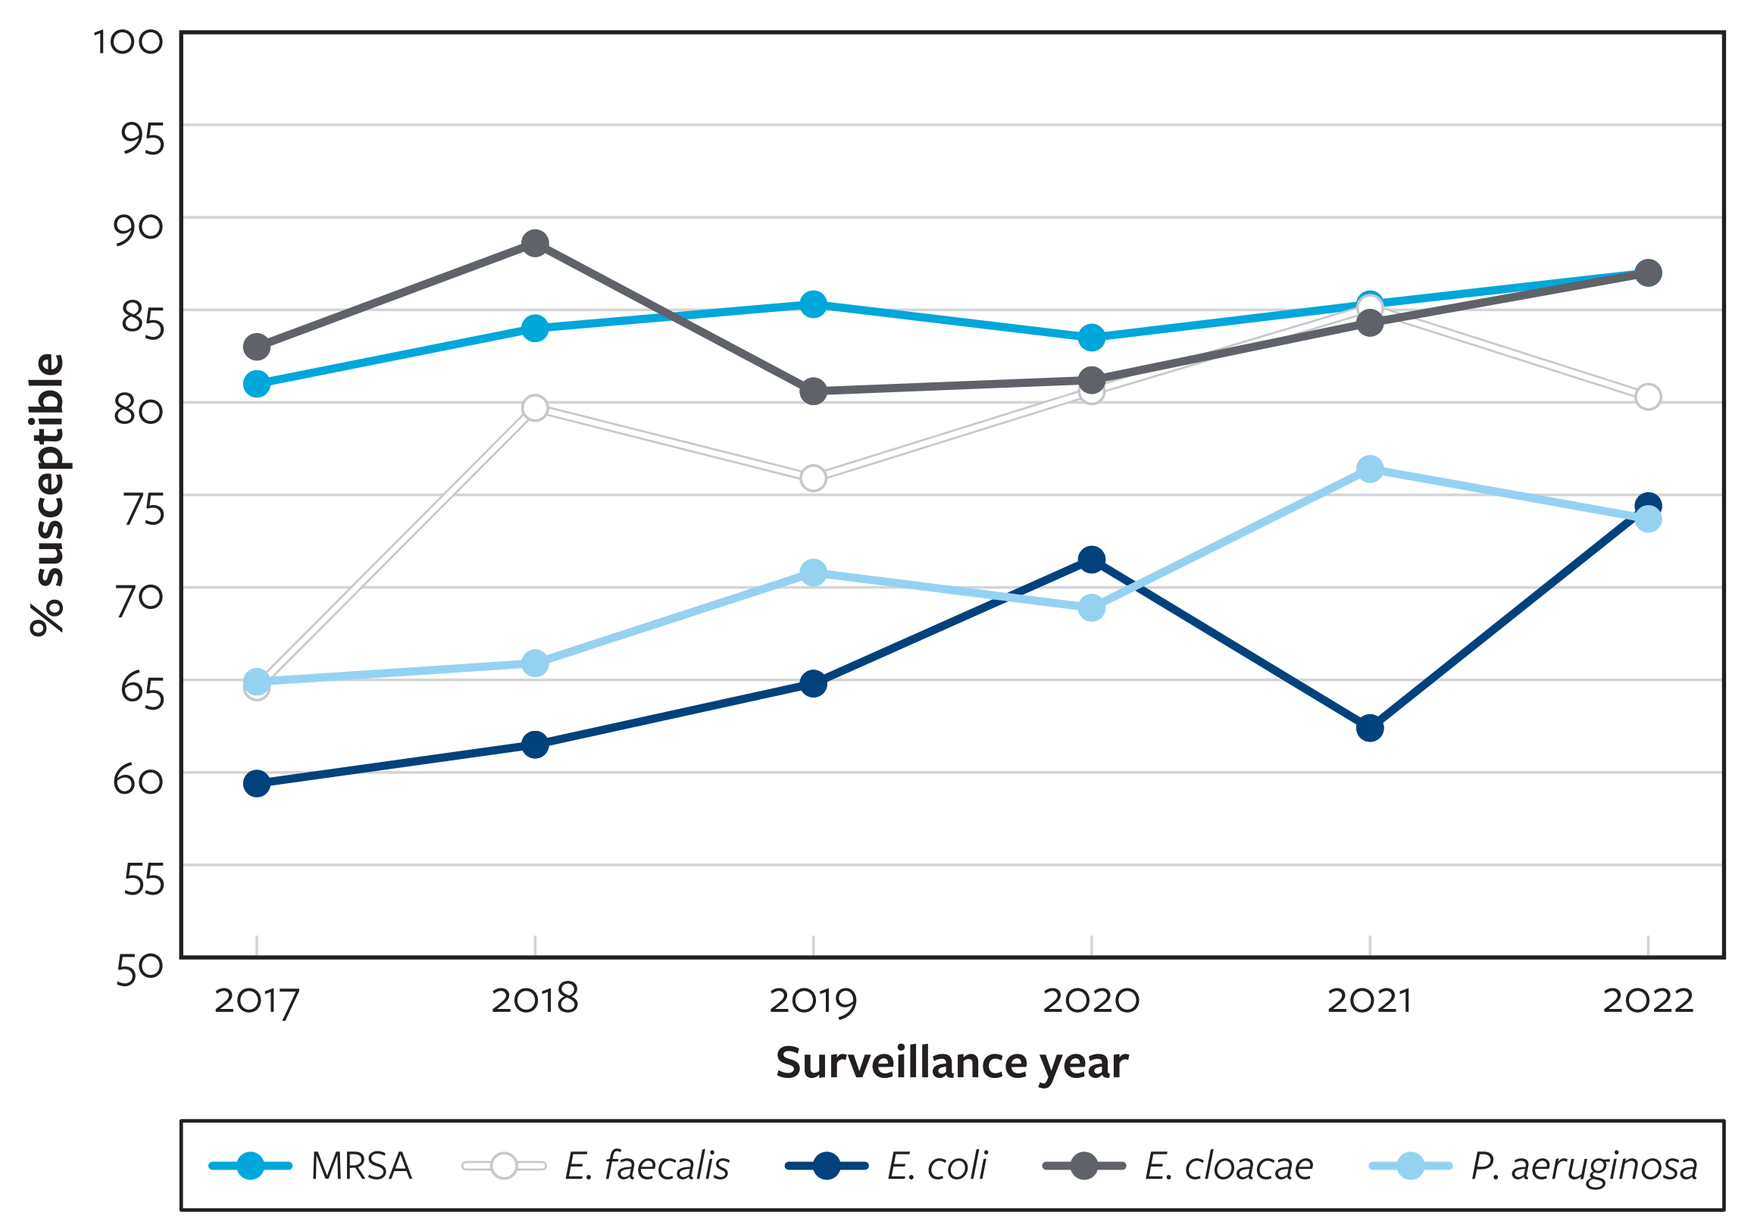
% susceptible
100
95
90
85
80
75
70
65
60
55
50
2017
2018
2019
2020
2021
2022
Surveillance year
MRSA
E. faecalis
E. coli
E. cloacae
P. aeruginosa

Figure 3. Susceptibility of delafloxacin and comparators against Gram-positive bacterial SSSI pathogens (2017–2022)

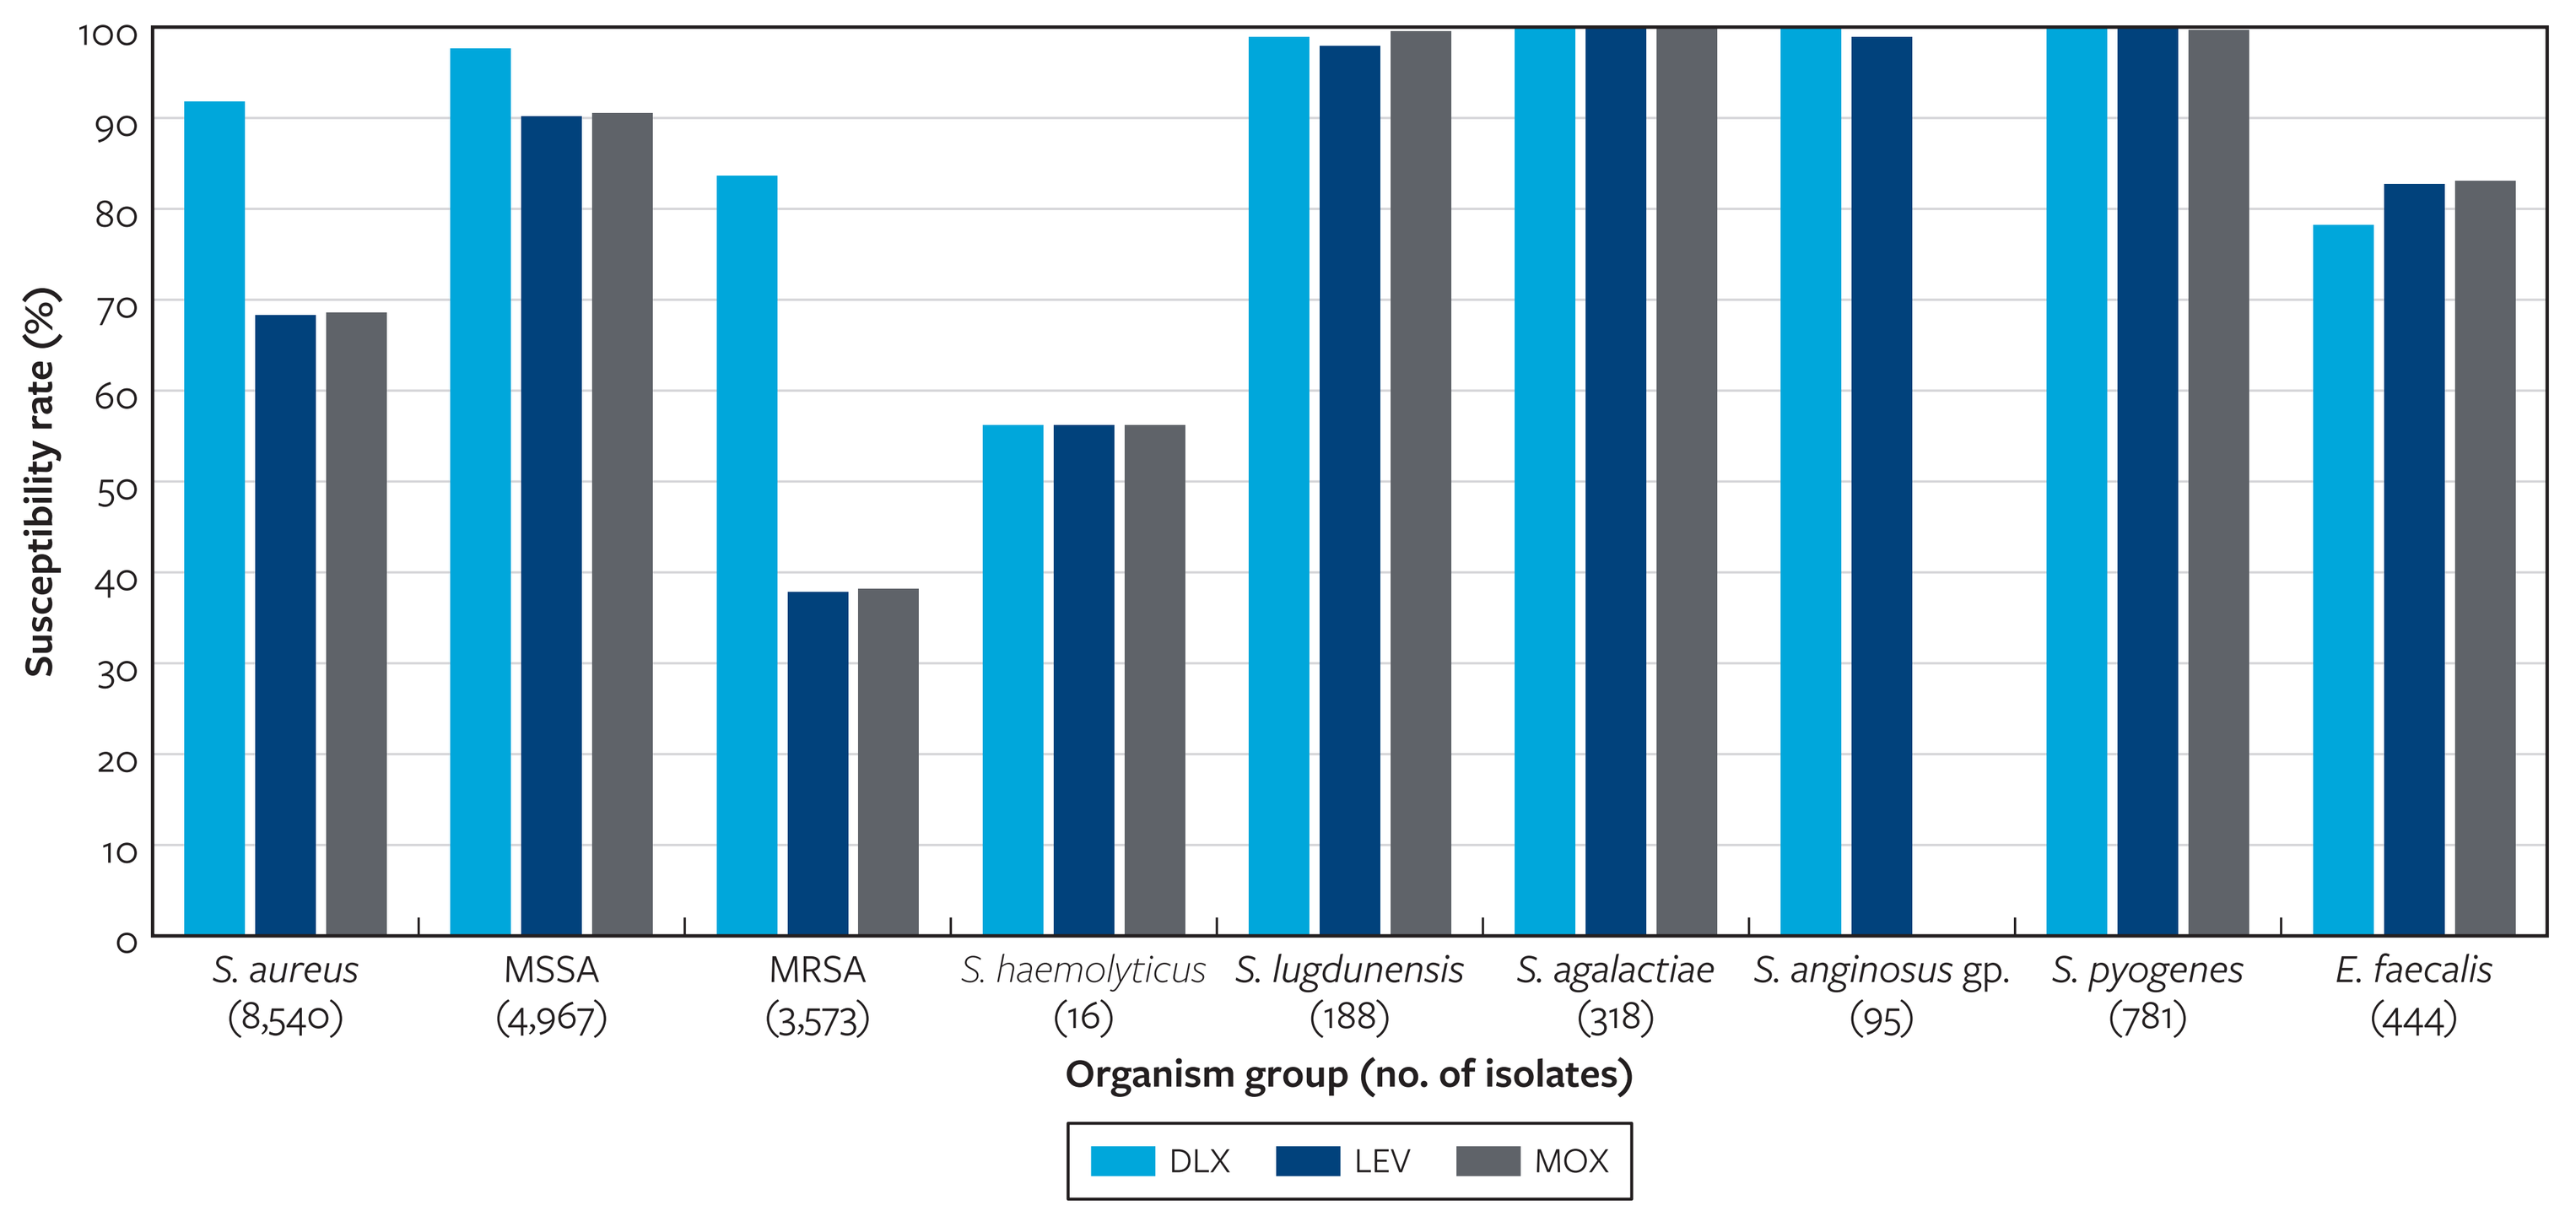
Susceptibility rate (%)
100
90
80
70
60
50
40
30
20
10
0
S. aureus (8,540)
MSSA (4,967)
MRSA (3,573)
S. haemolyticus (16)
S. lugdunensis (188)
S. agalactiae (318)
S. anginosus gp. (95)
S. pyogenes (781)
E. faecalis (444)
Organism group (no. of isolates)
DLX
LEV
MOX

Figure 4. Susceptibility of delafloxacin and comparators against Gram-negative bacterial SSSI pathogens (2017–2022)

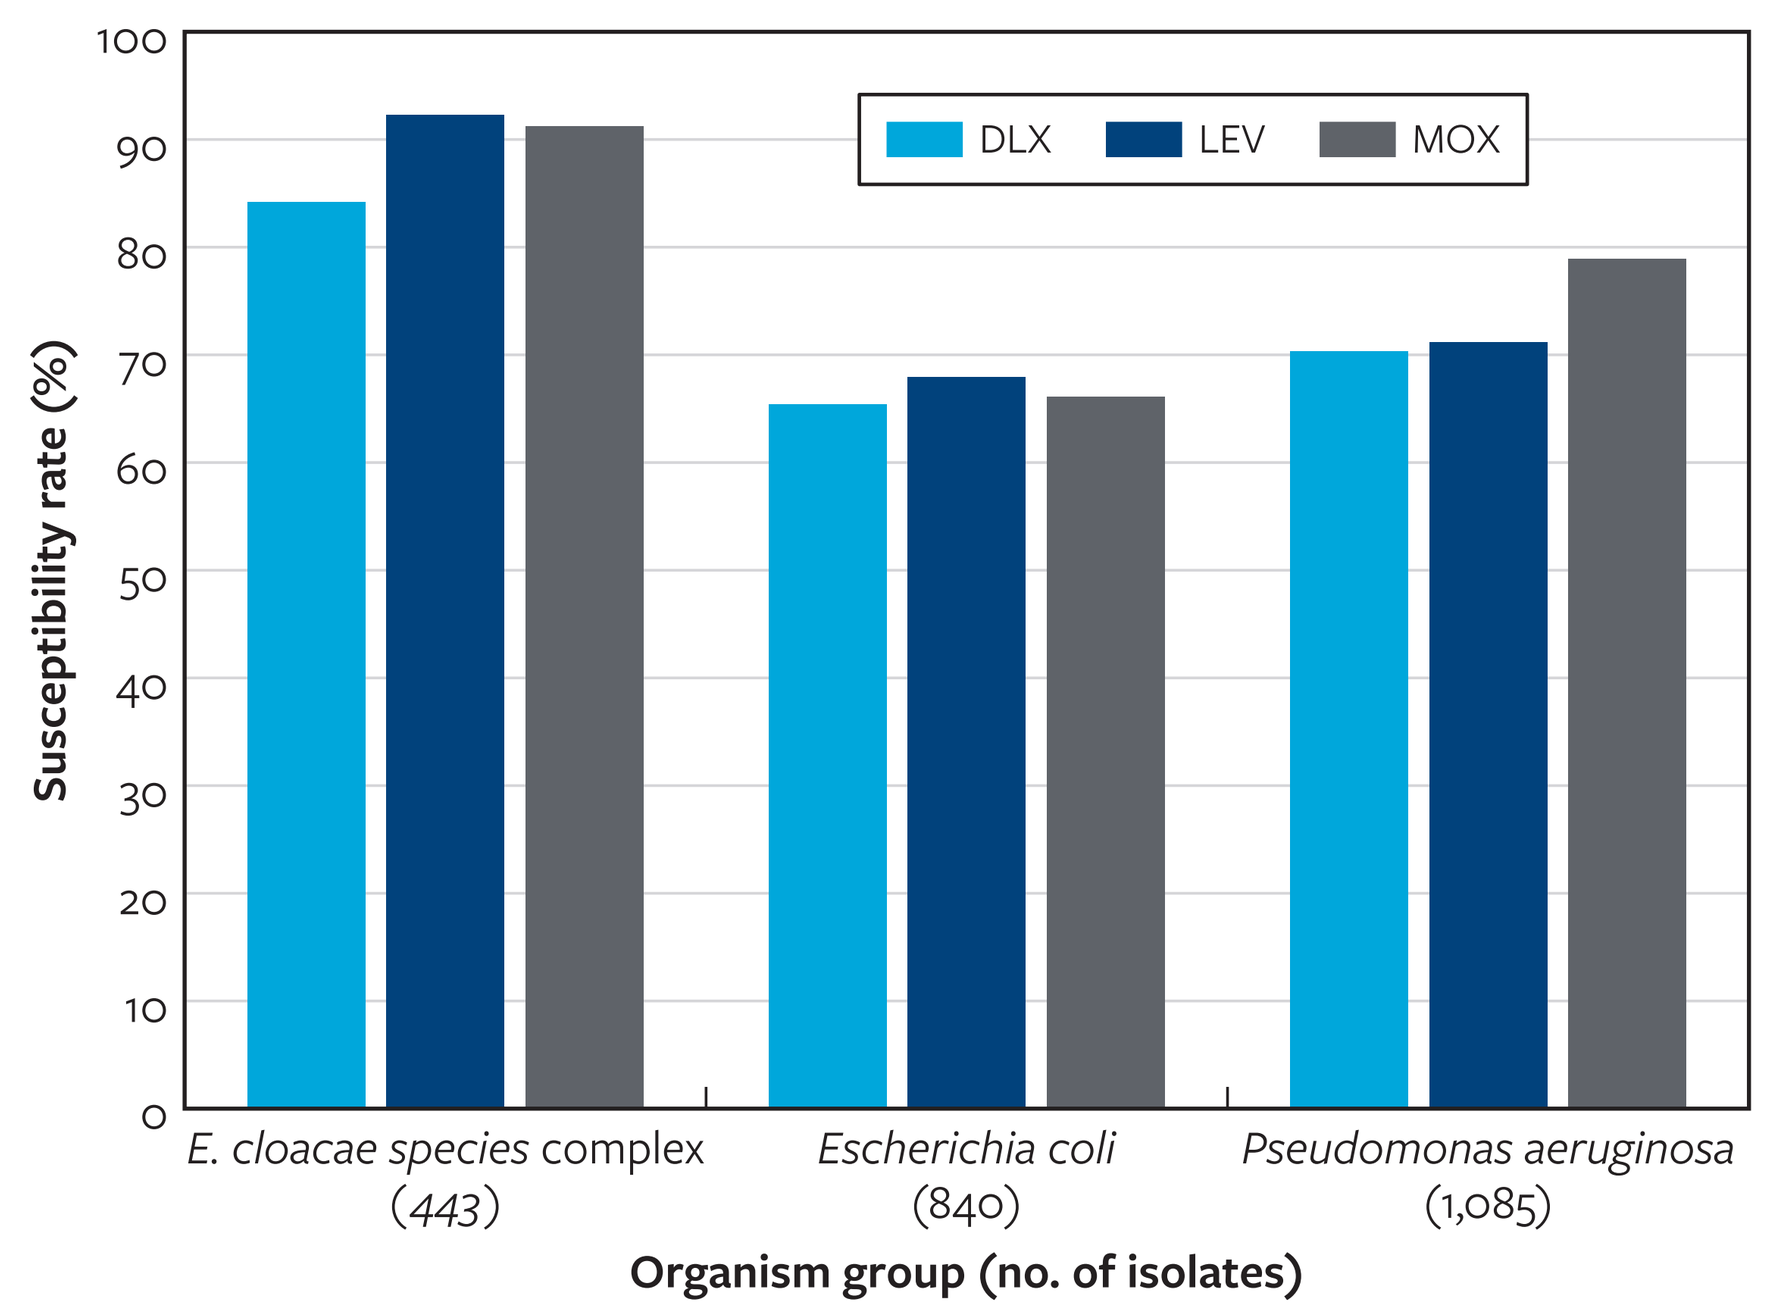
Susceptibility rate (%)
100
90
80
70
60
50
40
30
20
10
0
DLX
LEV
MOX
E. cloacae species complex (443)
Escherichia coli (840)
Pseudomonas aeruginosa (1,085)
Organism group (no. of isolates)

Table 1. Susceptibilities of delafloxacin and comparator agents tested against Gram-positive and Gram-negative SSSI pathogens from the United States (2017–2022)

	2017	2018	2019	2020	2021	2022	Total
S. aureus n	2,791	1,175	1,127	1,171	1,170	1,106	8,540
DLX[a]	**90.0%**	**92.1%**	**92.8%**	**91.5%**	**92.8%**	**93.9%**	**91.8%**
LEV[a]	66.5%	71.9%	69.2%	66.9%	68.5%	69.3%	68.3%
MOX[a]	66.8%	72.4%	69.5%	67.1%	68.9%	69.3%	68.6%
MSSA *n*	1,594	699	651	639	708	676	4,967
DLX	**96.9%**	**97.6%**	**98.3%**	**98.1%**	**97.7%**	**98.2%**	**97.6%**
LEV	89.0%	**90.7%**	**92.8%**	**90.9%**	88.4%	**91.3%**	**90.2%**
MOX	89.3%	**91.1%**	**93.1%**	**91.1%**	88.6%	**91.4%**	**90.5%**
MRSA *n*	1,197	476	476	532	462	430	3,573
DLX	81.0%	84.0%	85.3%	83.5%	85.3%	87.0%	83.6%
LEV	36.5%	44.3%	37.0%	38.0%	37.9%	34.7%	37.8%
MOX	36.8%	45.0%	37.2%	38.3%	38.7%	34.7%	38.2%
S. haemolyticus n	2	1	3	3	4	3	16
DLX	50.0%	0.0%	66.7%	66.7%	50.0%	66.7%	56.2%
LEV	50.0%	0.0%	66.7%	66.7%	50.0%	66.7%	56.2%
MOX	50.0%	0.0%	66.7%	66.7%	50.0%	66.7%	56.2%
S. lugdunensis n	41	29	27	32	27	32	188
DLX	**100%**	**100%**	**100%**	**93.8%**	**100%**	**100%**	**98.9%**
LEV	**97.6%**	**96.6%**	**100%**	**96.9%**	**100%**	**96.9%**	**97.9%**
MOX	**100%**	**100%**	**100%**	**96.9%**	**100%**	**100%**	**99.5%**
S. agalactiae n	70	51	51	47	45	54	318
DLX	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**
LEV	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**
MOX	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**
S. anginosus group *n*	12	14	11	18	18	22	95
DLX	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**
LEV	**100%**	**100%**	**100%**	**94.4%**	**100%**	**100%**	**98.9%**
MOX[c]	—	—	—	—	—	—	—

	2017	2018	2019	2020	2021	2022	Total
S. pyogenes n	324	122	110	88	71	66	781
DLX	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**	**100%**
LEV	**99.7%**	**100%**	**100%**	**100%**	**100%**	**100%**	**99.9%**
MOX[b]	**99.7%**	**99.2%**	**100%**	**100%**	**100%**	**100%**	**99.7%**
E. faecalis n	65	59	83	72	94	71	444
DLX	64.6%	79.7%	75.9%	80.6%	85.1%	80.3%	78.2%
LEV	75.4%	86.4%	77.1%	87.5%	88.3%	80.3%	82.7%
MOX[d]	76.9%	86.4%	77.1%	87.5%	89.4%	80.3%	83.1%
E. cloacae species complex *n*	94	79	62	69	70	69	443
DLX	83.0%	88.6%	80.6%	81.2%	84.3%	87.0%	84.2%
LEV	**93.6%**	**94.9%**	88.7%	**92.8%**	87.1%	**95.7%**	92.3%
CIP	**93.5%**	**93.7%**	87.1%	**92.8%**	85.7%	**92.8%**	91.2%
Escherichia coli n	165	148	128	137	133	129	840
DLX	59.4%	61.5%	64.8%	71.5%	62.4%	74.4%	65.4%
LEV	66.7%	63.5%	68.0%	72.3%	62.4%	75.2%	67.9%
CIP	61.8%	60.5%	66.9%	71.5%	61.7%	76.0%	66.1%
Klebsiella pneumoniae n	82	82	53	76	73	85	451
DLX	81.7%	62.2%	67.9%	78.9%	69.9%	64.7%	71.0%
LEV	89.0%	75.6%	75.5%	85.5%	78.1%	78.8%	80.7%
CIP	85.4%	65.0%	71.7%	78.9%	67.1%	72.9%	73.7%
Pseudomonas aeruginosa n	154	179	195	177	182	198	1,085
DLX	64.9%	65.9%	70.8%	68.9%	76.4%	73.7%	70.3%
LEV	69.5%	67.6%	70.8%	68.9%	75.3%	74.7%	71.2%
CIP	82.5%	73.6%	78.4%	76.8%	83.0%	79.8%	78.9%

Susceptibilities ≥90.0% listed in **bold**.
[a] Breakpoint interpretive criteria as published by the FDA for delafloxacin or CLSI (2023) for the comparator agents.
[b] Moxifloxacin has no CLSI or FDA breakpoint interpretive criteria for *S. agalactiae* and *S. pyogenes*, so EUCAST (2023) breakpoints were applied.
[c] Moxifloxacin has no CLSI, FDA, or EUCAST breakpoints for *Streptococcus anginosus* group isolates.
[d] Moxifloxacin has no CLSI breakpoint interpretive criteria for *E. faecalis*, so FDA breakpoints were applied.

References

CLSI 2018. M07Ed11. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. Clinical and Laboratory Standards Institute, Wayne, PA.

CLSI. 2023. M100Ed33. Performance standards for antimicrobial susceptibility testing: 33rd informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.

United States Food and Drug Administration (2023). Antibacterial susceptibility test interpretive criteria. Accessed July 2023. https://www.fda.gov/Drugs/DevelopmentApprovalProcess/DevelopmentResources/ucm575163.htm

Contact

element

Michael D. Huband, Associate Director
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
michael.huband@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2023_23-MEL-04_P1_Dela.pdf

Charges may apply. No personal information is stored.

IDWeek 2023, October 11–15, 2023, Boston, Massachusetts