IDWeek 2023 | Poster #2117

In Vitro Antimicrobial Activity of Taurolidine against *Candida auris* Bloodstream Isolates from Global Sources

Antony Pfaffle[1], Leonard Duncan[2], Mariana Castanheira[2], Bruce Reidenberg[3], Cecilia Carvalhaes[2], Jessica Vaughn[1], Phoebe Mounts[1]

[1] CorMedix Inc., Berkeley Heights, New Jersey, USA; [2] JMI Laboratories, North Liberty, Iowa, USA;
[3] Weill Cornell Medicine, New York, New York, USA

Introduction

- The Centers for Disease Control and Prevention (CDC) have recently warned that the fungus *Candida auris* is an emerging, resistant pathogen of concern [1], and recent epidemiological reports point to the increasing global spread of *C. auris* [2, 3].
 - Recent reports highlight *C. auris* as an emerging cause of invasive bloodstream infections (BSI) [4-6].
 - Central venous catheter use is associated with *C. auris* BSI [4].
- Taurolidine (Figure 1) has been marketed in Europe as a catheter lock solution component with the goal of reducing catheter-related bloodstream infections (CRBSI) [7].
- Taurolidine is a novel antimicrobial with broad spectrum antibacterial/antifungal activity and two mechanisms of action that does not lend itself to clinically relevant microbial resistance at concentrations contained within a central venous catheter.
- Taurolidine exerts its activity through damage to microbial cell walls by denaturing surface proteins and chemically altering membrane lipids, as well as inhibiting adherence of microorganisms to biological surfaces.
- A large, double-blind, randomized comparator-controlled study was conducted that evaluated the safety and efficacy of a catheter lock solution containing taurolidine/heparin to prevent CRBSIs in patients with advanced kidney disease on hemodialysis (*Clin J Am Soc Nephrol*, in press, https://journals.lww.com/cjasn/abstract/9900/taurolidine_heparin_lock_solution_and.236.aspx).
- The marketing application for use in the United States is currently under review by the United States Food and Drug Administration (US FDA).
- To provide additional evidence for the potential utility of taurolidine to reduce CRBSI, this study investigated the *in vitro* antimicrobial activity of taurolidine against a diverse set of *C. auris* strains/isolates using reference testing methods.

Materials and Methods

- 41 *C. auris* strains/isolates were collected from various sources during 2008–2019 (Table 1 and Figure 2), and the species were confirmed by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany) or other standard methods:
 - The CDC and the US FDA Antimicrobial Resistance Isolate Bank (CDC AR) (n=11) [8].
 - Several clades were represented (Table 1).
 - The Westerdijk Fungal Biodiversity Institute (n=4) [9].
 - JMI Laboratories' SENTRY Antimicrobial Surveillance Program (n=16) [10].
 - A special collection of *C. auris* isolates obtained by JMI Laboratories from a hospital in Nairobi, Kenya (n=10) [11].
- Most *C. auris* isolates (90%; 37/41) were from bloodstream infections (Table 1).
- The *C. auris* set was tested for antifungal susceptibility using Clinical and Laboratory Standards Institute (CLSI) broth microdilution guidelines [12, 13].
- The test medium was Roswell Park Memorial Institute 1640 broth buffered with MOPS (morpholinepropanesulfonic acid) and 0.2% (w/v) glucose [12].
- JMI Laboratories produced the minimal inhibitory concentration (MIC) panels.
- CLSI-recommended quality control strains were also tested (*Candida krusei* ATCC 6258 and *Candida parapsilosis* ATCC 22019).
- MIC values were read after 24 hours at 35°C in ambient air.
- Taurolidine MIC values were read at both 50% and 100% growth inhibition.
 - No CLSI reading criteria for taurolidine have been published.
- Amphotericin B and fluconazole were also tested as control antifungal agents.
 - Tentative CDC breakpoints were applied to *C. auris* [14].

IDWeek 2023, October 11–15, 2023, Boston, Massachusetts

Results

- Taurolidine exhibited *in vitro* antimicrobial activity against all *C. auris* strains and isolates regardless of geographic source, year of isolation, or clade (Tables 1–3).
 - Overall $MIC_{50/90}$ values were 256/512 mg/L using the 50% inhibition reading criterion and 512/512 mg/L using the 100% inhibition reading criterion (Table 3).
 - The taurolidine MIC range was 128–512 mg/L and 256–1,024 mg/L using the 50% and 100% inhibition reading criteria, respectively (Table 3).
- In general, taurolidine MIC values were slightly more potent when read using the 50% inhibition criterion (Tables 1–2).
- The full set of *C. auris* isolates was 53.7% resistant to amphotericin B and 85.4% resistant to fluconazole using tentative CDC MIC breakpoints (Table 3; [14]).
- Where applicable, MIC data for amphotericin B and fluconazole tested against these strains/isolates agreed well with previously published data (Table 1).
 - Because these isolates were not randomly selected, however, these resistance rates cannot be extrapolated to other sets of *C. auris* isolates.
- Taurolidine maintained activity against the amphotericin B and fluconazole-resistant subsets (Table 4).

Figure 1. Two-dimensional structure of taurolidine

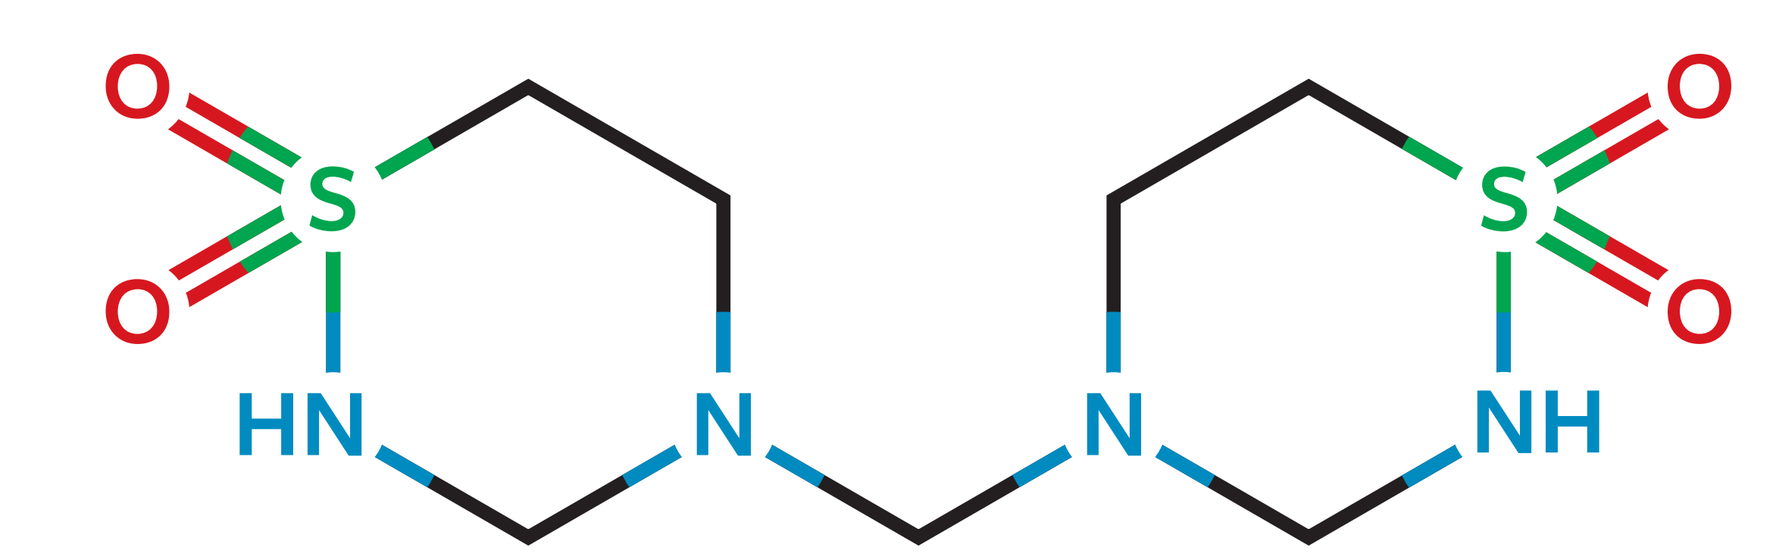

Figure 2. Geographical sources of *Candida auris* strains/isolates tested in this study

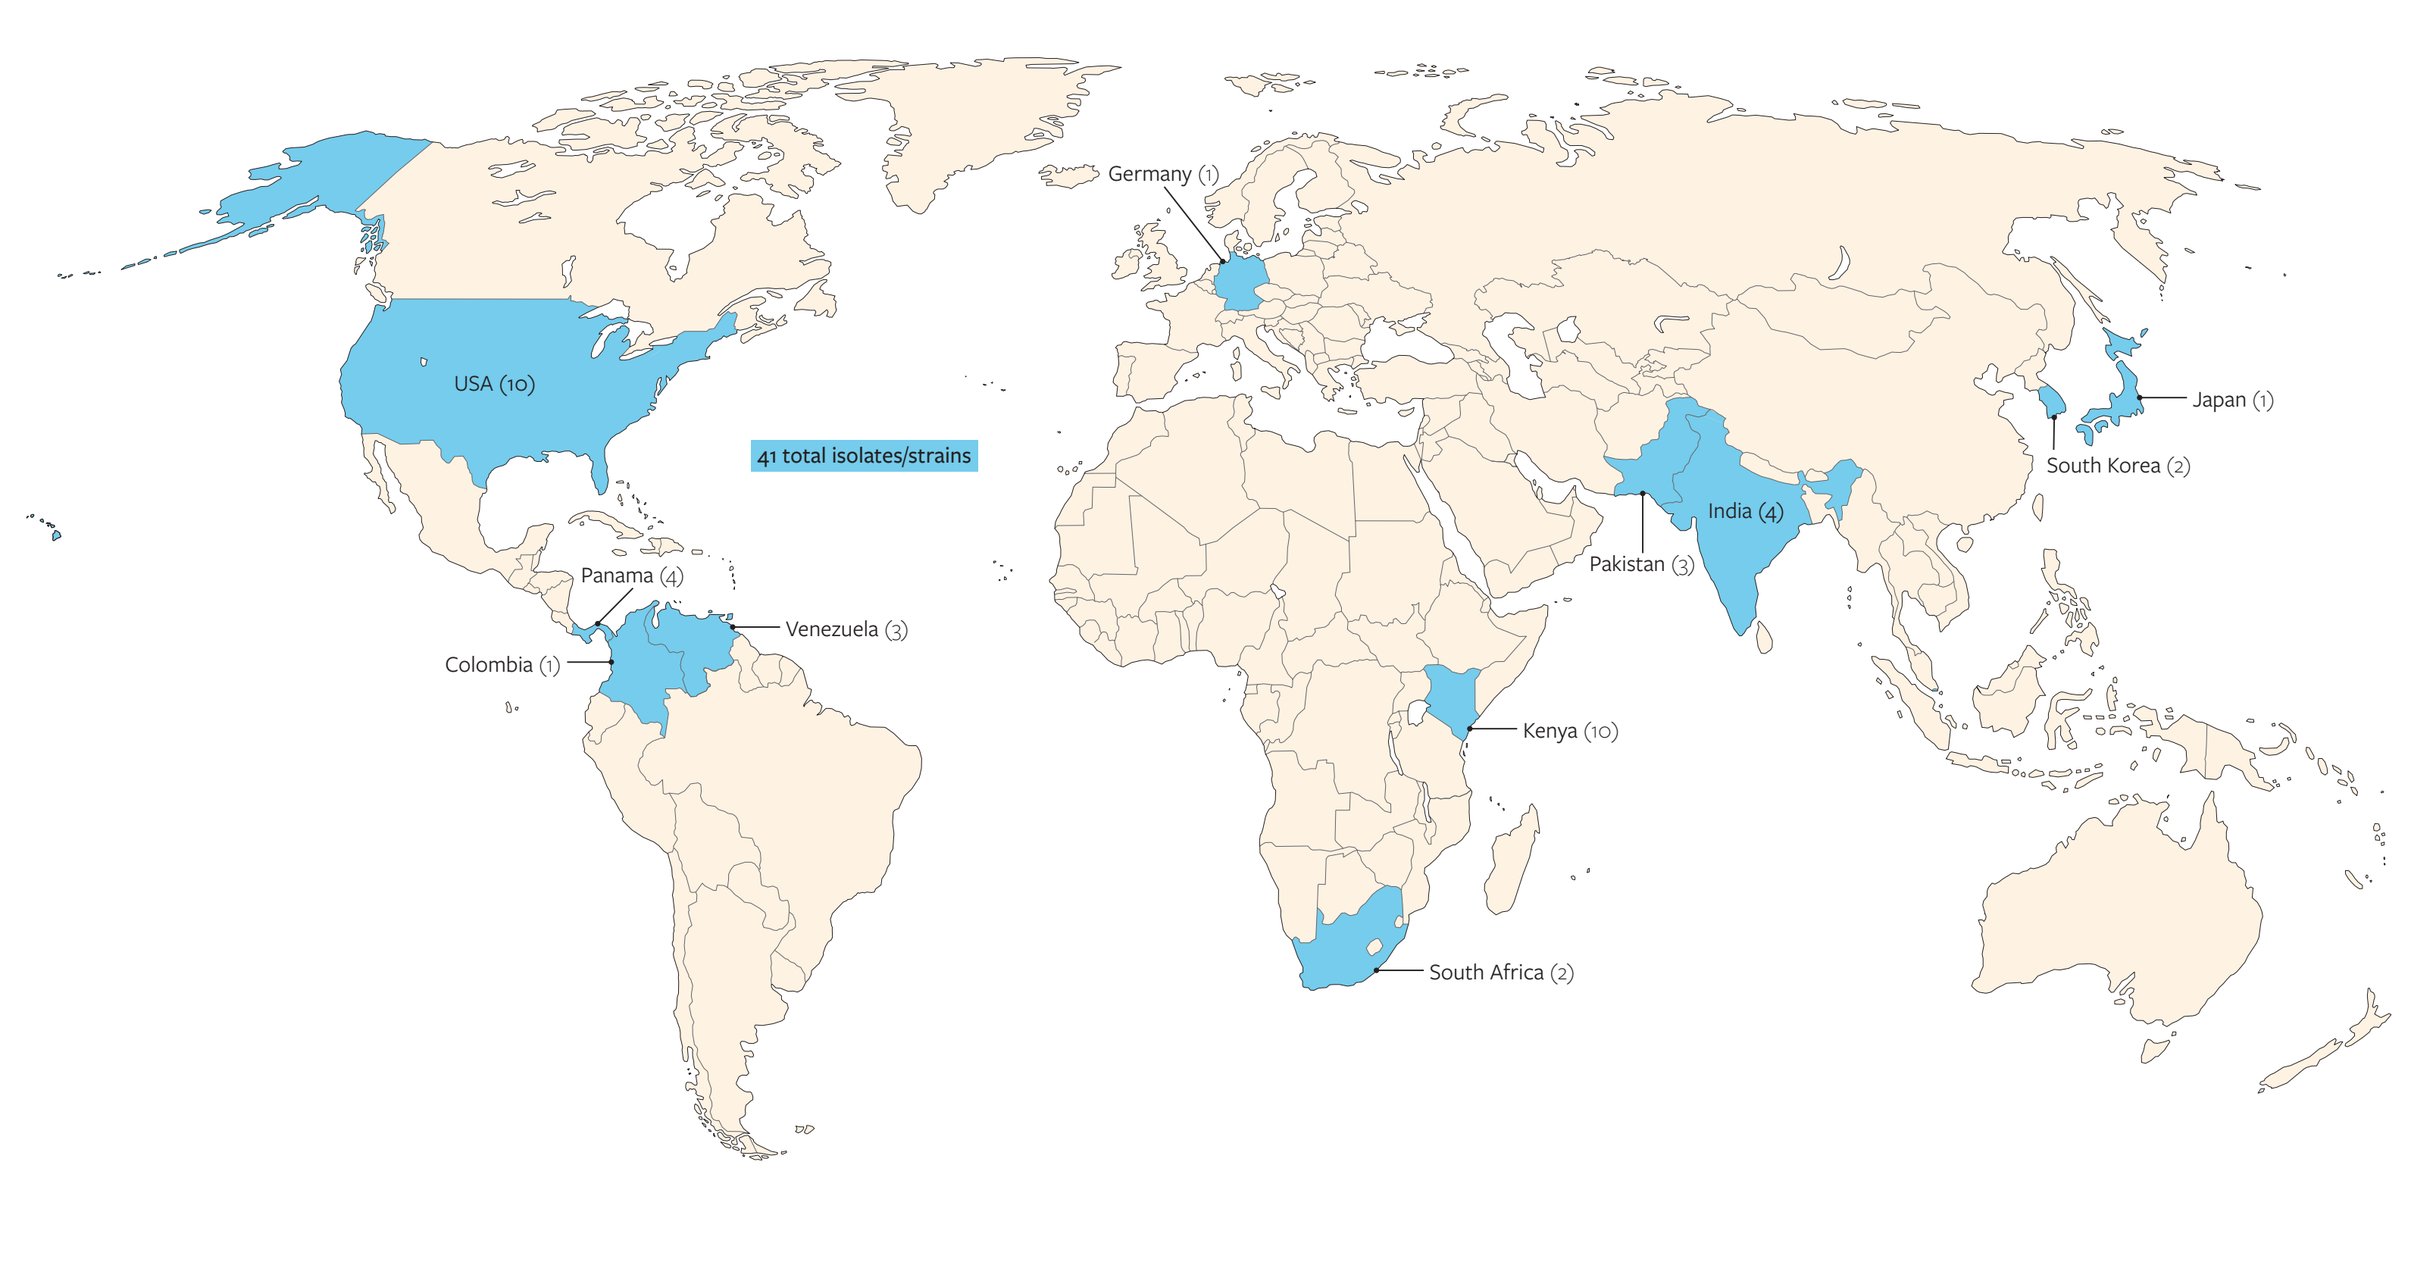

USA (10)
41 total isolates/strains
Germany (1)
Panama (4)
Venezuela (3)
Colombia (1)
Kenya (10)
South Africa (2)
Pakistan (3)
India (4)
South Korea (2)
Japan (1)

Table 1. *Candida auris* isolates/strains and their associated MIC values

Collection No.	Source	Year	Country	Specimen type	Clade [a]	Alias	MIC (mg/L) Taurolidine (50% inhibition)	Taurolidine (100% inhibition)	Amphotericin B (published MIC value)	Fluconazole (published MIC value)
1	CDC AR	2019	Pakistan	Burn wound	South Asia	AR Bank# 0382	256	512	1 (0.4) [b]	4 (16) [b]
2	SENTRY	2019	Panama	Blood culture			256	512	2	64
3	SENTRY	2019	Panama	Blood culture			256	512	2	8
4	SENTRY	2019	USA	Blood culture			512	512	2	>128
5	SENTRY	2019	USA	Blood culture			256	512	2	>128
6	CDC AR	2018	India	BAL	South Asia	AR Bank# 0389	512	512	2 (4) [b]	>128 (256) [b]
7	SENTRY	2018	Panama	Blood culture			256	512	2	8
8	SENTRY	2018	Panama	Blood culture			512	512	2	4
9	SENTRY	2018	USA	Blood culture			512	512	2	>128
10	SENTRY	2018	USA	Blood culture			512	512	2	>128
11	SENTRY	2018	USA	Blood culture			512	512	2	>128
12	SENTRY	2017	USA	Blood culture			256	512	2	>128
13	SENTRY	2016	USA	Blood culture			512	512	1	>128
14	SENTRY	2016	USA	Blood culture			512	512	1	>128
15	SENTRY	2015	USA	Blood culture			512	512	1	>128
16	SENTRY	2014	Colombia	Blood culture			512	512	1	64
17	CDC AR	2014	Pakistan	Blood culture	South Asia	AR Bank# 0388	512	512	2 (1.5) [b]	>128 (>256) [b]
18	Kenya	2013	Kenya [c]	Blood culture			256	512	1	32
19	Kenya	2013	Kenya [c]	Blood culture			256	256	2	>128
20	Kenya	2013	Kenya [c]	Blood culture			256	512	1	128
21	Kenya	2013	Kenya [c]	Blood culture			512	512	1	>128
22	SENTRY	2013	USA	Blood culture			256	512	2	>128
23	CDC AR	2013	Venezuela	Blood culture	South America	AR Bank# 0931	512	1024	2 (0.75) [b]	>128 (>256) [b]
24	Kenya	2012	Kenya [c]	Blood culture			256	512	1	128
25	Kenya	2012	Kenya [c]	Blood culture			256	512	1	>128
26	Kenya	2012	Kenya [c]	Blood culture			256	512	1	>128
27	Kenya	2012	Kenya [c]	Blood culture			256	512	1	128
28	Kenya	2012	Kenya [c]	Blood culture			128	512	2	128
29	CDC AR	2012	South Africa	Blood culture	Africa	AR Bank# 0383	256	512	1 (0.4) [b]	>128 (128) [b]
30	CDC AR	2012	South Africa	Blood culture	Africa	AR Bank# 0384	256	512	1 (0.5) [b]	>128 (128) [b]
31	CDC AR	2012	Venezuela	Blood culture	South America	AR Bank# 0385	512	512	1 (0.5) [b]	>128 (>256) [b]
32	CDC AR	2012	Venezuela	Blood culture	South America	AR Bank# 0386	512	512	1 (0.5) [b]	>128 (>256) [b]
33	Kenya	2011	Kenya [c]	Blood culture			256	512	2	>128
34	SENTRY	2009	Germany	Blood culture			256	512	2	128
35	CDC AR	2009	Japan	Ear	East Asia	AR Bank# 0381	256	512	1 (0.4) [b]	2 (4) [b]
36	CDC AR	2008	Pakistan	Blood culture	South Asia	AR Bank# 0387	256	512	1 (0.75) [b]	4 (8) [b]
37	CDC AR	NR	India	Wound	South Asia	AR Bank# 0390	512	512	2 (4) [b]	>128 (>256) [b]
38	Westerdijk	NR	India	Blood culture		CBS 12768 [d]	512	1024	2 (4) [e]	>128 (32) [e]
39	Westerdijk	NR	India	Blood culture		CBS 12766 [d]	512	1024	2 (4) [e]	>128 (32) [e]
40	Westerdijk	NR	South Korea	Blood culture		CBS 12372 [d]	256	512	1 (1) [e]	128 (2) [e,f]
41	Westerdijk	NR	South Korea	Blood culture		CBS 12373 [d]	256	512	2 (1) [e]	128 (16) [e,f]

Abbreviations: BAL, bronchoalveolar lavage; CDC AR, Centers for Disease Control and Prevention and US FDA Antimicrobial Resistance Isolate Bank; MIC, minimal inhibitory concentration; NR, not reported.
[a] Clade categorization as reported by the CDC and FDA Antibiotic Resistance (CDC AR) isolate bank [8].
[b] MIC values for amphotericin B (Etest) and fluconazole from the CDC AR bank website are shown in parentheses.
[c] The Kenyan isolates are described by Adam et al. [11].
[d] CBS isolates are from the Westerdijk Fungal Biodiversity Institute [9].
[e] 24-hour MIC values for amphotericin B and fluconazole from Larkin et al. [15] are shown in parentheses. The 48-hour fluconazole MIC values were all >64 mg/L.
[f] The fluconazole 24-hour MIC values were measured twice for strain #40 (repeat MIC value, 64 mg/L) and strain #41 (repeat MIC value, 128 mg/L).

Table 2. Cumulative distributions of taurolidine MIC values against various *Candida auris* subsets

Candida auris set (no. of isolates)	No. and cumulative % of isolates inhibited at MIC (mg/L) of: ≤0.5	1	2	4	8	16	32	64	128	256	512	1024	MIC_{50}	MIC_{90}
All *Candida auris*														
Taurolidine 50% inhibition (41)								0 0.0	1 2.4	22 56.1	18 100.0		256	512
Taurolidine 100% inhibition (41)									0 0.0	1 2.4	37 92.7	3 100.0	512	512
SENTRY *Candida auris*														
Taurolidine 50% inhibition (16)									0 0.0	7 43.8	9 100.0		512	512
Taurolidine 100% inhibition (16)										0 0.0	16 100.0		512	512
Kenyan *Candida auris*														
Taurolidine 50% inhibition (10)								0 0.0	1 10.0	8 90.0	1 100.0		256	256
Taurolidine 100% inhibition (10)									0 0.0	1 10.0	9 100.0		512	512
CDC AR bank *Candida auris*														
Taurolidine 50% inhibition (11)									0 0.0	5 45.5	6 100.0		512	512
Taurolidine 100% inhibition (11)										0 0.0	10 90.9	1 100.0	512	512
Westerdijk *Candida auris*														
Taurolidine 50% inhibition (4)									0 0.0	2 50.0	2 100.0		256	
Taurolidine 100% inhibition (4)										0 0.0	2 50.0	2 100.0	512	

Abbreviations: CDC AR, Centers for Disease Control and Prevention and US FDA Antimicrobial Resistance Isolate Bank; MIC, minimal inhibitory concentration.

Table 3. Activity of taurolidine and comparators against the full *Candida auris* set (n=41)

Antimicrobial agent	MIC_{50}	MIC_{90}	Range	CLSI [a] %S	%I	%R
Taurolidine 50% inhibition	256	512	128 to 512			
Taurolidine 100% inhibition	512	512	256 to 1024			
Amphotericin B	2	2	1 to 2	46.3		53.7
Fluconazole	>128	>128	2 to >128	14.6		85.4

[a] Using tentative CDC antifungal susceptibility breakpoints [14].

Table 4. Activity of taurolidine against resistant *Candida auris* subsets

Resistant subset [a] / Taurolidine criterion	No. of isolates	mg/L MIC_{50}	MIC_{90}	MIC range
Amphotericin B MIC ≥2 mg/L				
Taurolidine 50% criterion	22	256	512	128 to 512
Taurolidine 100% criterion	22	512	1024	256 to 1024
Fluconazole MIC ≥32 mg/L				
Taurolidine 50% criterion	35	256	512	128 to 512
Taurolidine 100% criterion	35	512	512	256 to 1024

[a] Using tentative CDC antifungal susceptibility breakpoints [14].

Conclusions

- Taurolidine activity was similar for all *C. auris* subsets tested regardless of source or clade.
 - Overall $MIC_{50/90}$ values were 256/512 mg/L using the 50% inhibition reading criterion and 512/512 mg/L using the 100% inhibition reading criterion.
- There was no evidence that taurolidine activity was affected by resistance to amphotericin B or fluconazole.
- Based on these data, catheter lock solutions containing the broad-spectrum antimicrobial taurolidine at 13,500 mg/L have the potential to prevent CRBSI caused by *C. auris,* including clinical isolates that are resistant to amphotericin B and fluconazole.
- Taurolidine MIC values are being measured against additional *C. auris* isolates to further explore the activity of this broad-spectrum antimicrobial.

Disclosures

JMI Laboratories received compensation for services related to the preparation of this poster.

References

1. CDC, *Antibiotic resistance threats in the United States*. 2019, Centers for Disease Control and Prevention: Atlanta, GA. Available at: https://www.cdc.gov/drugresistance/pdf/threats-report/2019-ar-threats-report-508.pdf.
2. Lyman, M., Forsberg, K., Sexton, D.J., Chow, N.A., Lockhart, S.R., Jackson, B.R., et al., *Worsening Spread of Candida auris in the United States, 2019 to 2021.* Ann Intern Med, 2023. 176(4): 489-495.
3. Ademe, M. and Girma, F., *Candida auris: From Multidrug Resistance to Pan-Resistant Strains.* Infect Drug Resist, 2020. 13: 1287–1294.
4. Benedict, K., Forsberg, K., Gold, J.A.W., Baggs, J., and Lyman, M., *Candida auris–Associated Hospitalizations, United States, 2017–2022.* Emerg Infect Dis, 2023. 29(7): 1485-1487.
5. Koleri, J., Petkar, H.M., Rahman, S.A.S.H.A., and Rahman, S.A.M.A., *Candida auris Blood stream infection—a descriptive study from Qatar.* BMC Infect Dis, 2023. 23(1): 513.
6. Mohsin, J., Weerakoon, S., Ahmed, S., Puts, Y., Al Balushi, Z., Meis, J.F., et al., *A Cluster of Candida auris Blood Stream Infections in a Tertiary Care Hospital in Oman from 2016 to 2019.* Antibiotics (Basel), 2020. 9(10).
7. Reidenberg, B.E., Wanner, C., Polsky, B., Castanheira, M., Shelip, A., Stalleicken, D., et al., *Postmarketing experience with Neutrolin(R) (taurolidine, heparin, calcium citrate) catheter lock solution in hemodialysis patients.* Eur J Clin Microbiol Infect Dis, 2018. 37(4): 661-663.
8. CDC, *CDC & FDA Antibiotic Resistance Isolate Bank website*. 2023, Centers for Disease Control and Prevention. Available at: https://wwwn.cdc.gov/ARIsolateBank/.
9. *Westerdijk Fungal Biodiversity Institute website*. 2023: Available at: https://wi.knaw.nl/.
10. Fuhrmeister, A.S. and Jones, R.N., *The importance of antimicrobial resistance monitoring worldwide and the origins of SENTRY Antimicrobial Surveillance Program.* Open Forum Infect. Dis., 2019. 6(Suppl 1): S1-S4.
11. Adam, R.D., Revathi, G., Okinda, N., Fontaine, M., Shah, J., Kagotho, E., et al., *Analysis of Candida auris fungemia at a single facility in Kenya.* Int. J. Infect. Dis., 2019. 85: 182–187.
12. CLSI, *M27Ed4. Reference method for broth dilution antifungal susceptibility testing of yeasts*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.
13. CLSI, *M27M44S-Ed3. Performance standards for antifungal susceptibility testing of yeasts*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.
14. *Candida auris Antifungal Susceptibility Testing and Interpretation website.* Available at: https://www.cdc.gov/fungal/candida-auris/c-auris-antifungal.html. 2023, Centers for Disease Control and Prevention.
15. Larkin, E., Hager, C., Chandra, J., Mukherjee, P.K., Retuerto, M., Salem, I., et al., *The emerging pathogen Candida auris: Growth phenotype, virulence factors, activity of antifungals, and effect of SCY-078, a novel glucan synthesis inhibitor, on growth morphology and biofilm formation.* Antimicrob. Agents Chemother., 2017. 61(5): e02396.

Contact

element

Cecilia Carvalhaes, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia.carvalhaes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2023_23-CMX-02_P1_Taurolidine.pdf

Charges may apply. No personal information is stored.