IDWeek 2023 | Poster #1812

Comparative Activity of Oritavancin and Comparator Agents against *Staphylococcus aureus* and *Streptococcus* spp. Causing Skin and Soft Tissue Infections (SSTIs) in US Medical Centers between 2017–2019 and 2022

Cecilia G. Carvalhaes, Rodrigo E. Mendes, Dee Shortridge, Mariana Castanheira
Element Iowa City (JMI Laboratories)

Introduction

- There is a need for improved antibiotic formulations for the treatment of skin and soft tissue infections (SSTIs), especially with the rise of antimicrobial resistance among Gram-positive bacteria.
- Oritavancin is a lipoglycopeptide agent with a prolonged half-life and concentration-dependent bactericidal activity against clinically relevant Gram-positive pathogens.
- A new formulation of oritavancin (Kimyrsa™) was developed and approved by the US FDA to be infused over 1 hour and is indicated for the treatment of adult patients with acute bacterial skin and soft tissue infections (ABSSTIs) caused or suspected to be caused by susceptible isolates of designated Gram-positive microorganisms.
- This study evaluates the activity of oritavancin and comparators against *Staphylococcus aureus*, β-hemolytic streptococci (BHS), and Viridans group *Streptococcus* (VGS) isolates causing SSTIs in US medical centers in 2017–2019 and 2022.

Methods

- 6,110 Gram-positive SSTI pathogens were consecutively collected (1/patient) from 33 US medical centers in 2017–2019 and 2022—27/33 sites were included in both periods.
- Organisms from 2017–2019 and 2022 include 4,550 and 705 *S. aureus* isolates, 613 and 158 BHS isolates, and 57 and 27 VGS isolates, respectively (Figure 1).
- Isolates were identified by MALDI-TOF mass spectrometry and standard microbiology tests and susceptibility tested by CLSI broth microdilution.
- CLSI clinical breakpoints were applied.

Results

S. aureus

- Oritavancin activity against MSSA from the period 2017–2019 (*n*=2,671; MIC$_{50/90}$, 0.03/0.03 mg/L; 99.9%S) was similar to that during 2022 (*n*=450; MIC$_{50/90}$, 0.015/0.03 mg/L; 100%S; Table 1).
- The MRSA rate (36.2%) was slightly lower in 2022 than previous years (41.3%), but the oritavancin susceptibility rate remained 100% in 2022 (Table 2; Figure 2).
- Vancomycin, daptomycin, and linezolid also remained active (≥99.9% S) against *S. aureus* and the MRSA subset from both periods.

BHS

- Equivalent activity was observed for oritavancin against the BHS group between 2017–2019 (MIC$_{50/90}$, 0.06/0.25 mg/L; 96.7%S) and 2022 (MIC$_{50/90}$, 0.06/0.25 mg/L; 94.3%S), but variation among species was noted (Tables 1 and 2).
 - Five isolates showed oritavancin MIC values ≥0.5 mg/L in 2017–2019 (97.2%S), while all *S. agalactiae* were inhibited by oritavancin at ≤0.25 mg/L in 2022 (100%S).
 - 10 and 2 *S. pyogenes* displayed oritavancin MICs ≥0.5 mg/L in 2017–2019 (97.4%S) and 2022 (97.3%S), respectively.
 - Oritavancin MICs ≥0.5 mg/L were noted for 5/47 and 7/19 *S. dysgalactiae* in 2017–2019 (89.4%S) and 2022 (63.2%S), respectively.
- Penicillin, vancomycin, daptomycin, and linezolid inhibited all BHS isolates at their respective breakpoints, regardless of the period or BHS species.

VGS

- All VGS were inhibited by oritavancin, vancomycin, daptomycin, and linezolid in 2017–2019 and 2022, except for 1 *Streptococcus salivarius/vestibularis* group in 2022 (oritavancin MIC, 1 mg/L; 96.3%S; Table 2).
- Penicillin inhibited 93.0% and 92.6% of VGS isolates in 2017–2019 and 2022 periods, respectively.

Figure 1. Distribution of Gram-positive pathogens causing SSTIs in US medical centers (2017–2019 and 2022)

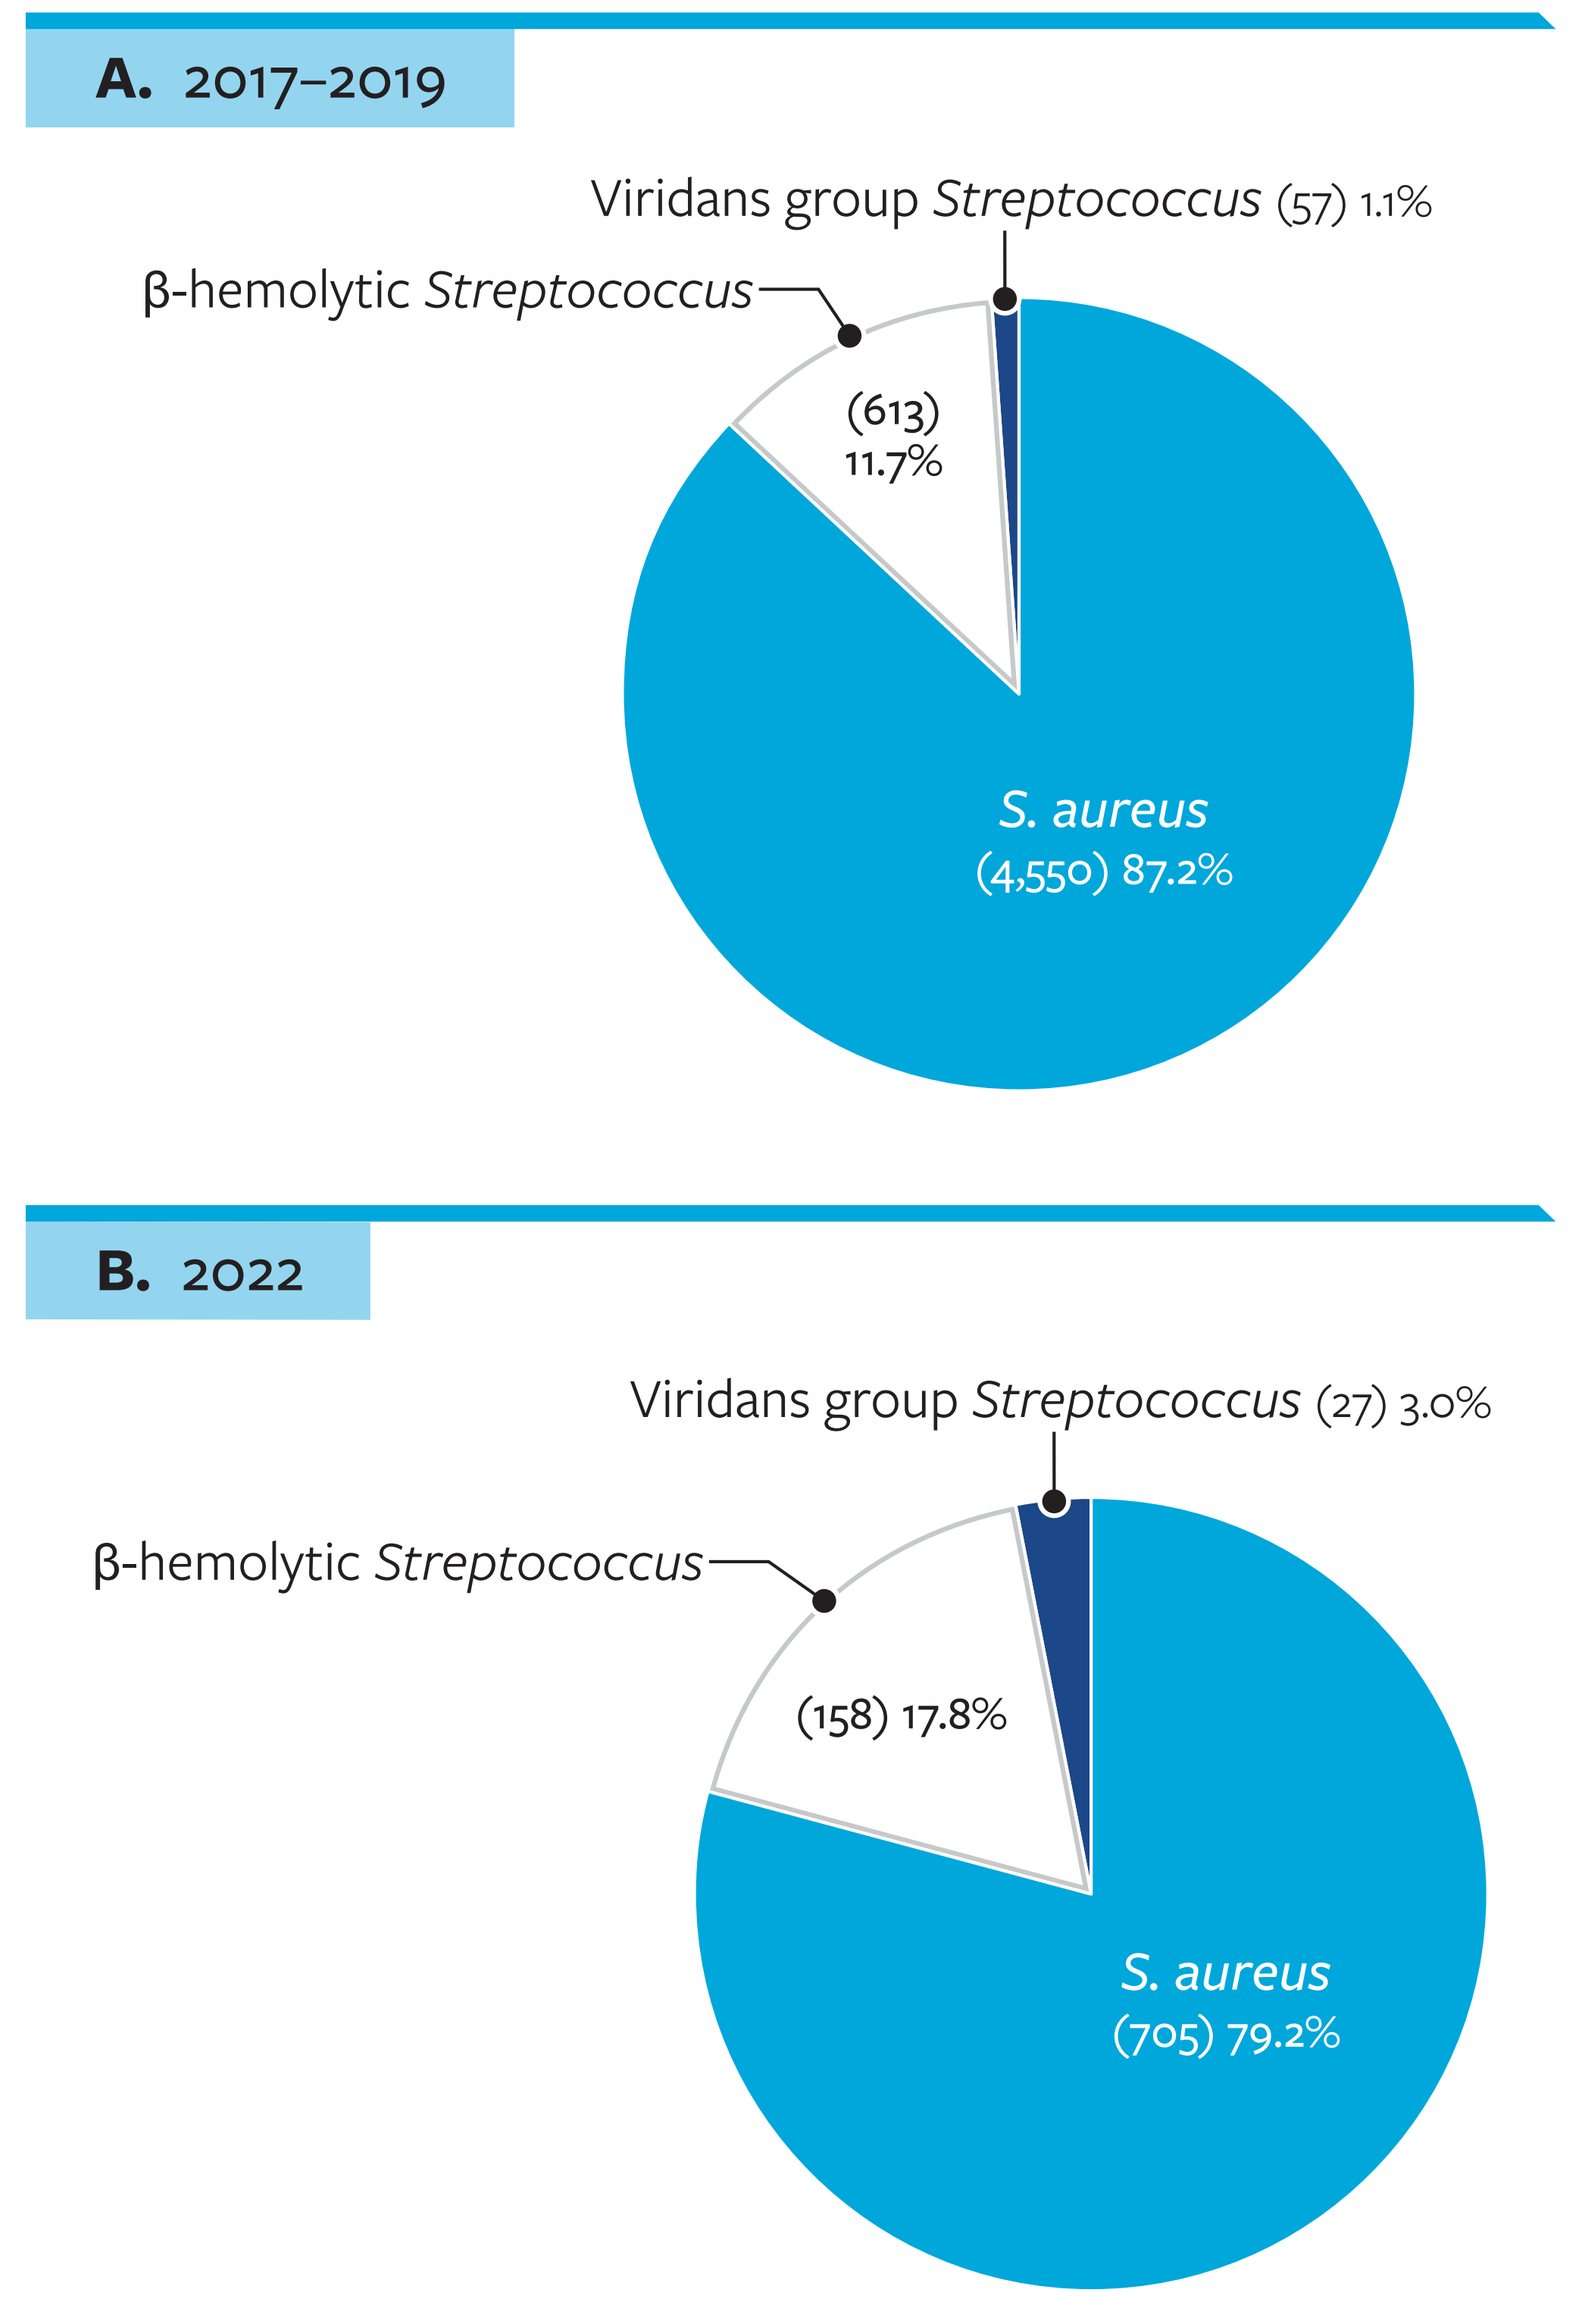
A. 2017–2019
Viridans group Streptococcus (57) 1.1%
β-hemolytic Streptococcus
(613) 11.7%
S. aureus (4,550) 87.2%
B. 2022
Viridans group Streptococcus (27) 3.0%
β-hemolytic Streptococcus
(158) 17.8%
S. aureus (705) 79.2%

Figure 2. Activity of oritavancin and comparator agents against MSSA and MRSA causing SSTIs in US medical centers (2017–2019 and 2022)

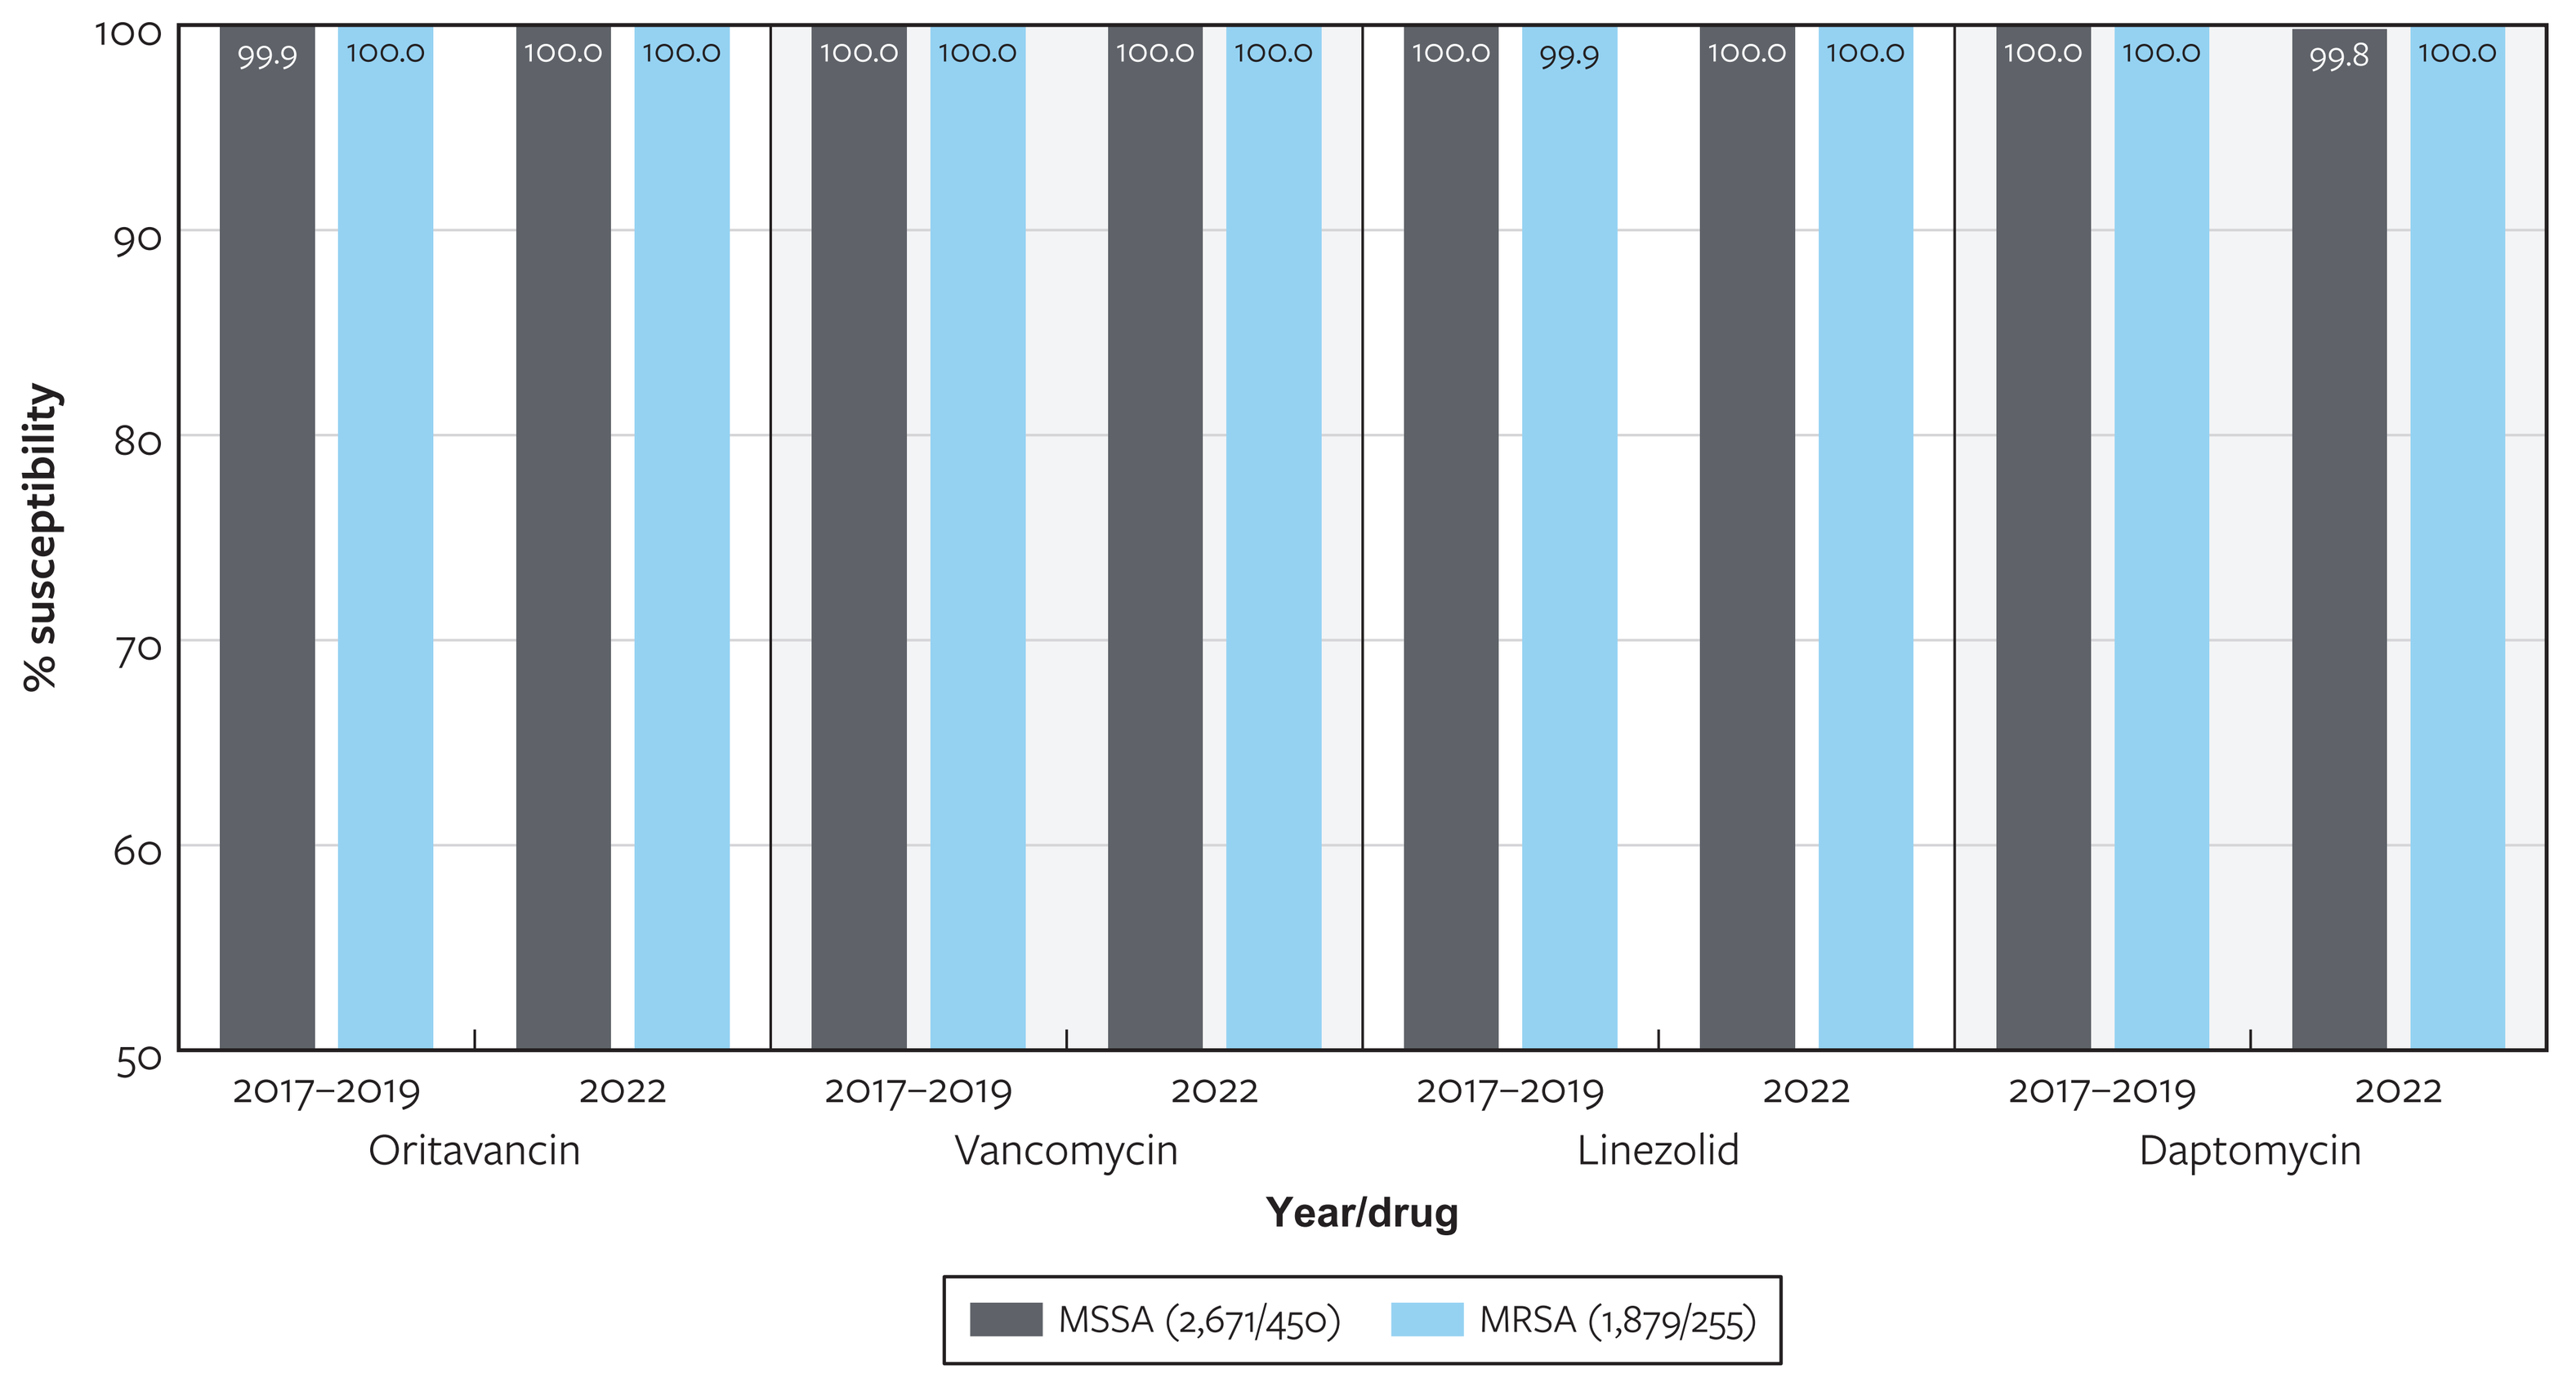
% susceptibility
100
90
80
70
60
50
99.9
100.0
100.0
100.0
100.0
100.0
100.0
100.0
100.0
99.9
100.0
100.0
100.0
100.0
99.8
100.0
2017–2019
2022
Oritavancin
Vancomycin
Linezolid
Daptomycin
Year/drug
MSSA (2,671/450)
MRSA (1,879/255)

Table 1. Activity of oritavancin and comparator agents against Gram-positive pathogens causing SSTIs in US medical centers (2017–2019 and 2022)

Organism group (no. of isolates 2017–2019/2022)	2017–2019						2022					
	ORI	VAN	LZD	DAP	OXA	PEN	ORI	VAN	LZD	DAP	OXA	PEN
	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$	MIC$_{50/90}$
S. aureus (4,550/705)	0.03/0.03	1/1	1/2	0.25/0.25	0.5/>2	NT	0.015/0.03	0.5/1	1/2	0.25/0.5	0.5/>2	NT
MSSA (2,671/450)	0.03/0.03	1/1	1/2	0.25/0.25	0.5/1	NT	0.015/0.03	0.5/1	1/2	0.25/0.5	0.5/1	NT
MRSA (1,879/255)	0.03/0.03	1/1	1/2	0.25/0.25	>2/>2	NT	0.015/0.3	0.5/1	1/2	0.25/0.5	>2/>2	NT
BHS (613/158)	0.06/0.25	0.5/0.5	1/2	≤0.06/0.25	NT	0.015/0.06	0.06/0.25	0.25/0.5	1/1	≤0.06/0.25	NT	0.015/0.06
S. pyogenes (381/73)	0.06/0.25	0.5/0.5	1/2	≤0.06/≤0.06	NT	≤0.008/0.015	0.06/0.25	0.25/0.25	1/1	≤0.06/≤0.06	NT	0.015/0.015
S. agalactiae (181/63)	0.06/0.12	0.5/0.5	1/2	0.12/0.25	NT	0.06/0.06	0.03/0.06	0.5/0.5	1/2	0.25/0.25	NT	0.06/0.06
S. dysgalactiae (47/19)	0.12/0.5	0.25/0.5	1/2	≤0.06/≤0.06	NT	0.015/0.015	0.25/1	0.25/0.25	1/2	≤0.06/≤0.06	NT	0.015/0.015
VGS (57/27)	0.008/0.06	0.5/1	1/2	0.25/0.5	NT	0.03/0.06	0.008/0.06	0.5/1	1/1	0.25/0.5	NT	0.03/0.12

ORI, oritavancin; VAN, vancomycin; LZD, linezolid; DAP, daptomycin; OXA, oxacillin; PEN, penicillin; NT, not tested

Table 2. Oritavancin and comparator agents susceptibility rates (%S) against Gram-positive pathogens causing SSTIs in US medical centers (2017–2019 and 2022)

Organism group (no. of isolates 2017–2019/2022)	2017–2019 CLSI[a] %S						2022 CLSI[a] %S					
	ORI	VAN	LZD	DAP	OXA	PEN	ORI	VAN	LZD	DAP	OXA	PEN
S. aureus (4,550/705)	>99.9	100.0	>99.9	100.0	58.7	NT	100.0	100.0	100.0	99.9	63.8	NT
MSSA (2,671/450)	99.9	100.0	100.0	100.0	100.0	NT	100.0	100.0	100.0	99.8	100.0	NT
MRSA (1,879/255)	100.0	100.0	99.9	100.0	0.0	NT	100.0	100.0	100.0	100.0	0.0	NT
BHS (613/158)	96.7	100.0	100.0	100.0	NT	100.0	94.3	100.0	100.0	100.0	NT	100.0
S. pyogenes (381/73)	97.4	100.0	100.0	100.0	NT	100.0	97.3	100.0	100.0	100.0	NT	100.0
S. agalactiae (181/63)	97.2	100.0	100.0	100.0	NT	100.0	100.0	100.0	100.0	100.0	NT	100.0
S. dysgalactiae (47/19)	89.4	100.0	100.0	100.0	NT	100.0	63.2	100.0	100.0	100.0	NT	100.0
VGS (57/27)	100.0	100.0	100.0	100.0	NT	93.0	96.3	100.0	100.0	100.0	NT	92.6

ORI, oritavancin; VAN, vancomycin; LZD, linezolid; DAP, daptomycin; OXA, oxacillin; PEN, penicillin
[a] CLSI M100Ed33E (2023)

Conclusions

- Isolates from SSTIs at US medical centers from 2017–2019 and 2022 remained highly susceptible to oritavancin, including 100% of MRSA.
- Vancomycin, daptomycin, and linezolid also exhibited stable activity against these pathogens over time.
- A small number of *S. dysgalactiae* was recovered in both periods, but changes in susceptibility rates were noted. Further evaluation with larger numbers of *S. dysgalactiae* isolates is necessary to assess the relevance of this finding.

Funding

This study was supported by Melinta Therapeutics. Cecilia Carvalhaes, Rodrigo Mendes, and Mariana Castanheira are employees of JMI Laboratories, which was paid consultant to Melinta in connection with the development of this poster.

Acknowledgments

The authors thank all the participant centers for their work in providing isolates.

References

Carvalhaes CG, Sader HS, Streit JM, Castanheira M, Mendes RE. Activity of Oritavancin against Gram-Positive Pathogens Causing Bloodstream Infections in the United States over 10 Years: Focus on Drug-Resistant Enterococcal Subsets (2010-2019). Antimicrob Agents Chemother. 2022 Feb 15;66(2):e0166721.

CLSI. 2023. M100Ed33. Performance standards for antimicrobial susceptibility testing: 33rd informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.

Hoover RK, Krsak M, Molina KC, Shah K, Redell M. Kimyrsa, An Oritavancin-Containing Product: Clinical Study and Review of Properties. Open Forum Infect Dis. 2022 Mar 23;9(5):ofac090.

Contact

Cecilia Carvalhaes, MD, Ph.D., D(ABMM)
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia.carvalhaes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2023_23-MEL-04_P4_Orita_SSSI.pdf

Charges may apply. No personal information is stored.

IDWeek 2023, October 11–15, 2023, Boston, Massachusetts