IDWeek 2023 | Poster #734

Trend Analysis of Oritavancin and Comparator Agents' Activities against *Enterococcus* species Causing Infections in US Medical Centers between 2017–2019 and 2022

Cecilia G. Carvalhaes, Rodrigo E. Mendes, Dee Shortridge, Mariana Castanheira
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- *Enterococcus* infections, mainly caused by *E. faecium* and *E. faecalis* species, remain challenging because of intrinsic resistance to various antimicrobials and acquisition of resistance traits.
- Oritavancin is a lipoglycopeptide agent with a prolonged half-life and concentration-dependent bactericidal activity against clinically relevant Gram-positive pathogens, including *Enterococcus*.
- A new formulation of oritavancin (Kimyrsa™) that can be infused over 1 hour as a single dose for the treatment of acute bacterial skin and skin structure infections (ABSSSI) was approved in 2021 by the US FDA.
- Currently, oritavancin is indicated for the treatment of adult patients with infections caused or suspected to be caused by susceptible isolates of designated Gram-positive microorganisms, including vancomycin-susceptible *Enterococcus faecalis*.
- The activity of oritavancin and its comparators against *Enterococcus* spp. and resistant subsets from US medical centers was evaluated comparatively between 2017–2019 and 2022.

Methods

- 2,344 Enterococcus were consecutively collected (1/patient) from 34 US medical centers in 2017–2019 and 2022 periods—28/34 sites were included in both periods.
- Organisms included 1,283 and 246 *E. faecalis,* 573 and 148 *E. faecium*, and 80 and 14 other *Enterococcus* spp. from 2017–2019 and 2022 periods, respectively (Figure 1A).
- *Enterococcus* isolates were collected mainly from bloodstream infections in both periods (Figure 1B).
- Isolates were identified by MALDI-TOF MS and standard microbiology tests then were susceptibility tested by CLSI broth microdilution.
- CLSI clinical breakpoints and VanA/VanB phenotypes were used, as follows:
 - VanA: isolates non-susceptible (NS) to teicoplanin and vancomycin
 - VanB: isolates susceptible (S) to teicoplanin and vancomycin-NS
- Oritavancin breakpoints against vancomycin-susceptible *E. faecalis* were applied to all *Enterococcus*.

Results

All *Enterococcus*

- Overall, oritavancin activity against *Enterococcus* spp. from 2017–2019 ($MIC_{50/90}$, 0.015/0.06 mg/L) was similar to 2022 ($MIC_{50/90}$, 0.015/0.03 mg/L; Table 1).
- At the vancomycin *E. faecalis* susceptible breakpoint (≤0.12 mg/L), oritavancin inhibited 98.9% and 97.5% of all *Enterococcus* spp. isolates from 2017–2019 and 2022, respectively (Table 2).
- Vancomycin and linezolid inhibited 78.6%/76.2% and 99.6%/99.0% of *Enterococcus* spp. from 2017–2019/2022, at the respective breakpoints (Table 2).

E. faecalis

- Oritavancin, vancomycin, linezolid and daptomycin showed stable susceptibility rates (>96%) against *E. faecalis* (Table 2).
- Vancomycin-resistant (VAN-R) *E. faecalis* rates were low in both periods—only 2.5% in 2017–2019 and 2.8% in 2022.
- Oritavancin remained active against 53.1% and 57.1% of VAN-R *E. faecalis* from 2017–2019 and 2022, respectively (Table 2).
 - VanA rates in *E. faecalis* were 2.3% in 2017–2019 and 2.4% in 2022.
 - Oritavancin inhibited the 3 VanB *E. faecalis* isolates at ≤0.03 mg/L.
 - Linezolid and daptomycin remained active against VAN-R *E. faecalis* (100%).

E. faecium

- Notably, an increase in the number of VanB-*E. faecium* isolates was observed in 2022 (18 isolates) compared to an average of 10 isolates per year in 2017–2019 (29 isolates total).
- Overall, similar activity was noted for oritavancin against *E. faecium* from 2017–2019 ($MIC_{50/90}$, 0.015/0.06 mg/L) and 2022 ($MIC_{50/90}$, 0.008/0.06 mg/L; Table 1).
- The susceptibility rates to oritavancin (using VAN-S *E. faecalis* breakpoints) and linezolid remained stable between 2017–2019 (99.1%/99.6%) and 2022 (98.6%/98.0%, respectively), as well as rates of daptomycin susceptible dose-dependent (SDD; 99.5% in 2017–2019 and 96.6% in 2022; Table 2 and Figure 2).
- The highest oritavancin MIC value displayed by *E. faecium* was 0.5 mg/L in 2017–2019 and 0.25 mg/L in 2022, only 1–2 dilutions above the susceptible clinical breakpoint for vancomycin-S *E. faecalis* (≤0.12 mg/L).
- The *E. faecium* susceptibility rate to vancomycin was 35.1% in 2017–2019 and 39.2% in 2022 (Table 2).
 - The VanB phenotype increased from 7.8% in 2017–2019 to 20.0% in 2022.
 - Oritavancin inhibited all VanB and 97.2–98.5% of VanA *E. faecium* at ≤0.12 mg/L.
 - Linezolid inhibited 99.4%/98.6% of VanA *E. faecium* isolates in 2017–2019/2022, respectively.
- The daptomycin-SDD rate decreased against VanA *E. faecium* between 2017–2019 (99.7%) and 2022 (93.1%; Table 2).

Other *Enterococcus* spp.

- Oritavancin (100%S in 2017–2019 and 2022) and comparators (87.5%–100%S in 2017–2019 and 100%S in 2022) were active against other species of *Enterococcus* (Table 2).

Conclusions

- Oritavancin exhibited potent and stable activity against *Enterococcus* clinical isolates, including VAN-R *E. faecium* and *E. faecalis* in US medical centers.
- Van-R *E. faecalis* rates remain very low in US medical centers (<3%), while Van-R *E. faecium* rates are >60%.
- An increase in VanB *E. faecium* phenotype and a slight decrease in the daptomycin-SDD rates in VAN-R *E. faecium* subsets were noted over time.
- Oritavancin may represent a valuable treatment option for infections caused by *Enterococcus* including VAN-R strains.

Funding

This study was supported by Melinta Therapeutics. Cecilia Carvalhaes, Rodrigo Mendes, and Mariana Castanheira are employees of JMI Laboratories, which was paid consultant to Melinta in connection with the development of this poster.

Figure 1. Characterization of *Enterococcus* isolates causing infection in US medical centers (2017–2019 and 2022)

A. By species

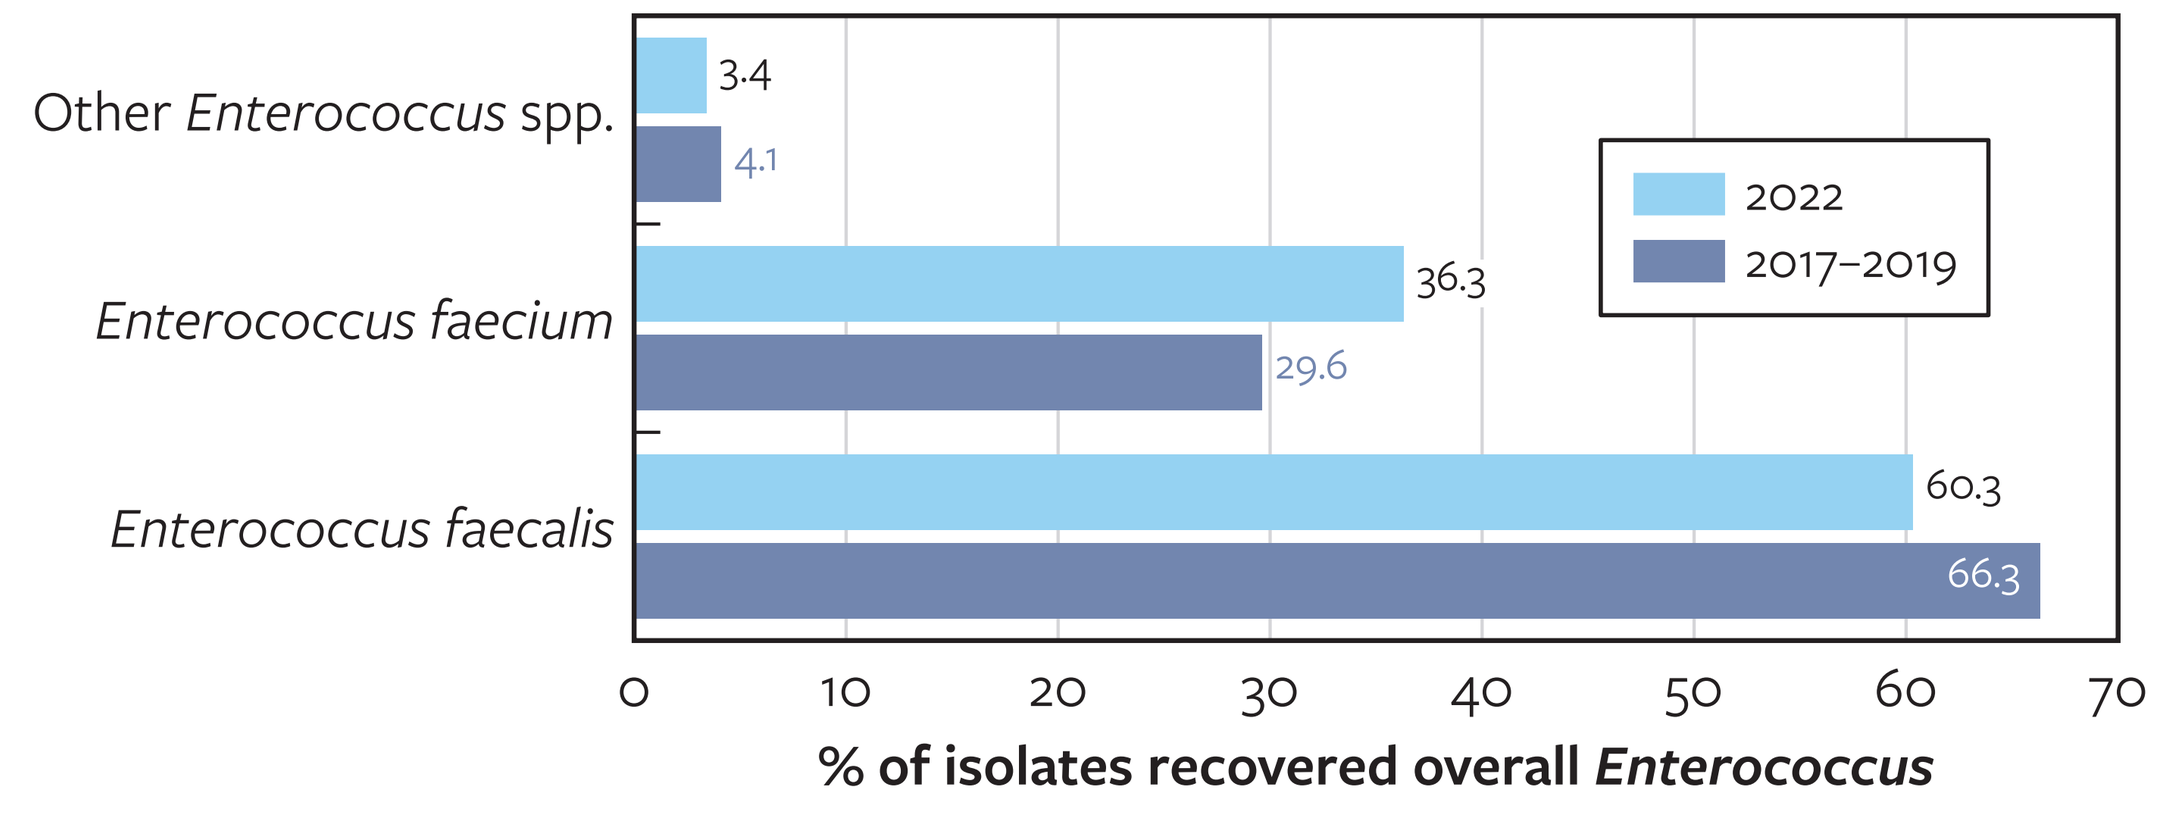
Other Enterococcus spp.
Enterococcus faecium
Enterococcus faecalis
3.4
4.1
36.3
29.6
60.3
66.3
2022
2017–2019
0
10
20
30
40
50
60
70
% of isolates recovered overall Enterococcus

B. By infection type

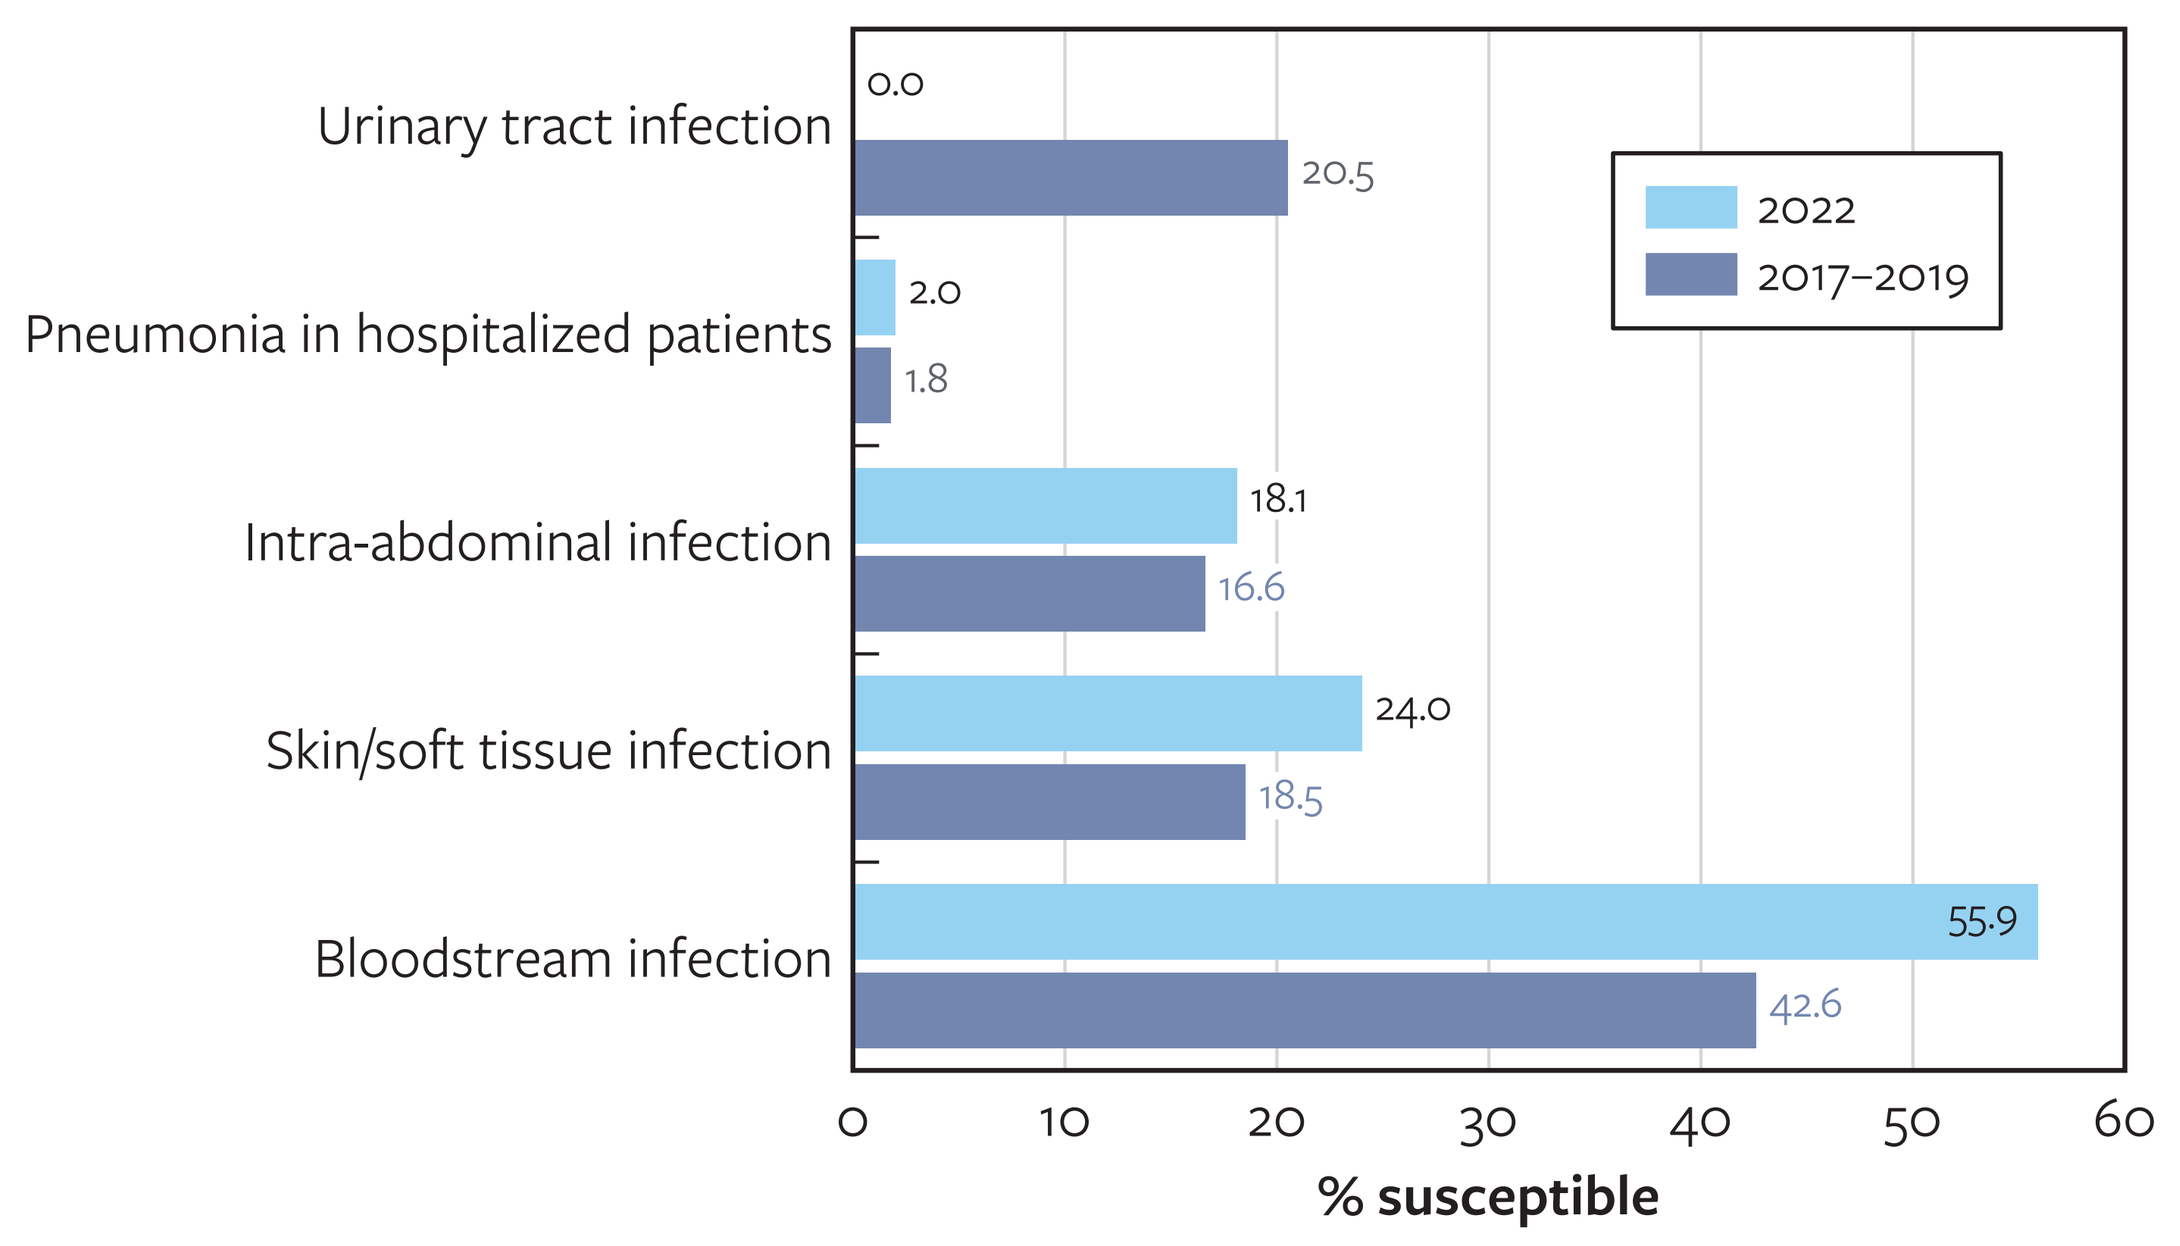
Urinary tract infection
Pneumonia in hospitalized patients
Intra-abdominal infection
Skin/soft tissue infection
Bloodstream infection
0.0
20.5
2.0
1.8
18.1
16.6
24.0
18.5
55.9
42.6
2022
2017–2019
0
10
20
30
40
50
60
% susceptible

Figure 2. Activity of oritavancin and comparator agents against *E. faecium* and resistant subgroups (2017–2019 and 2022)

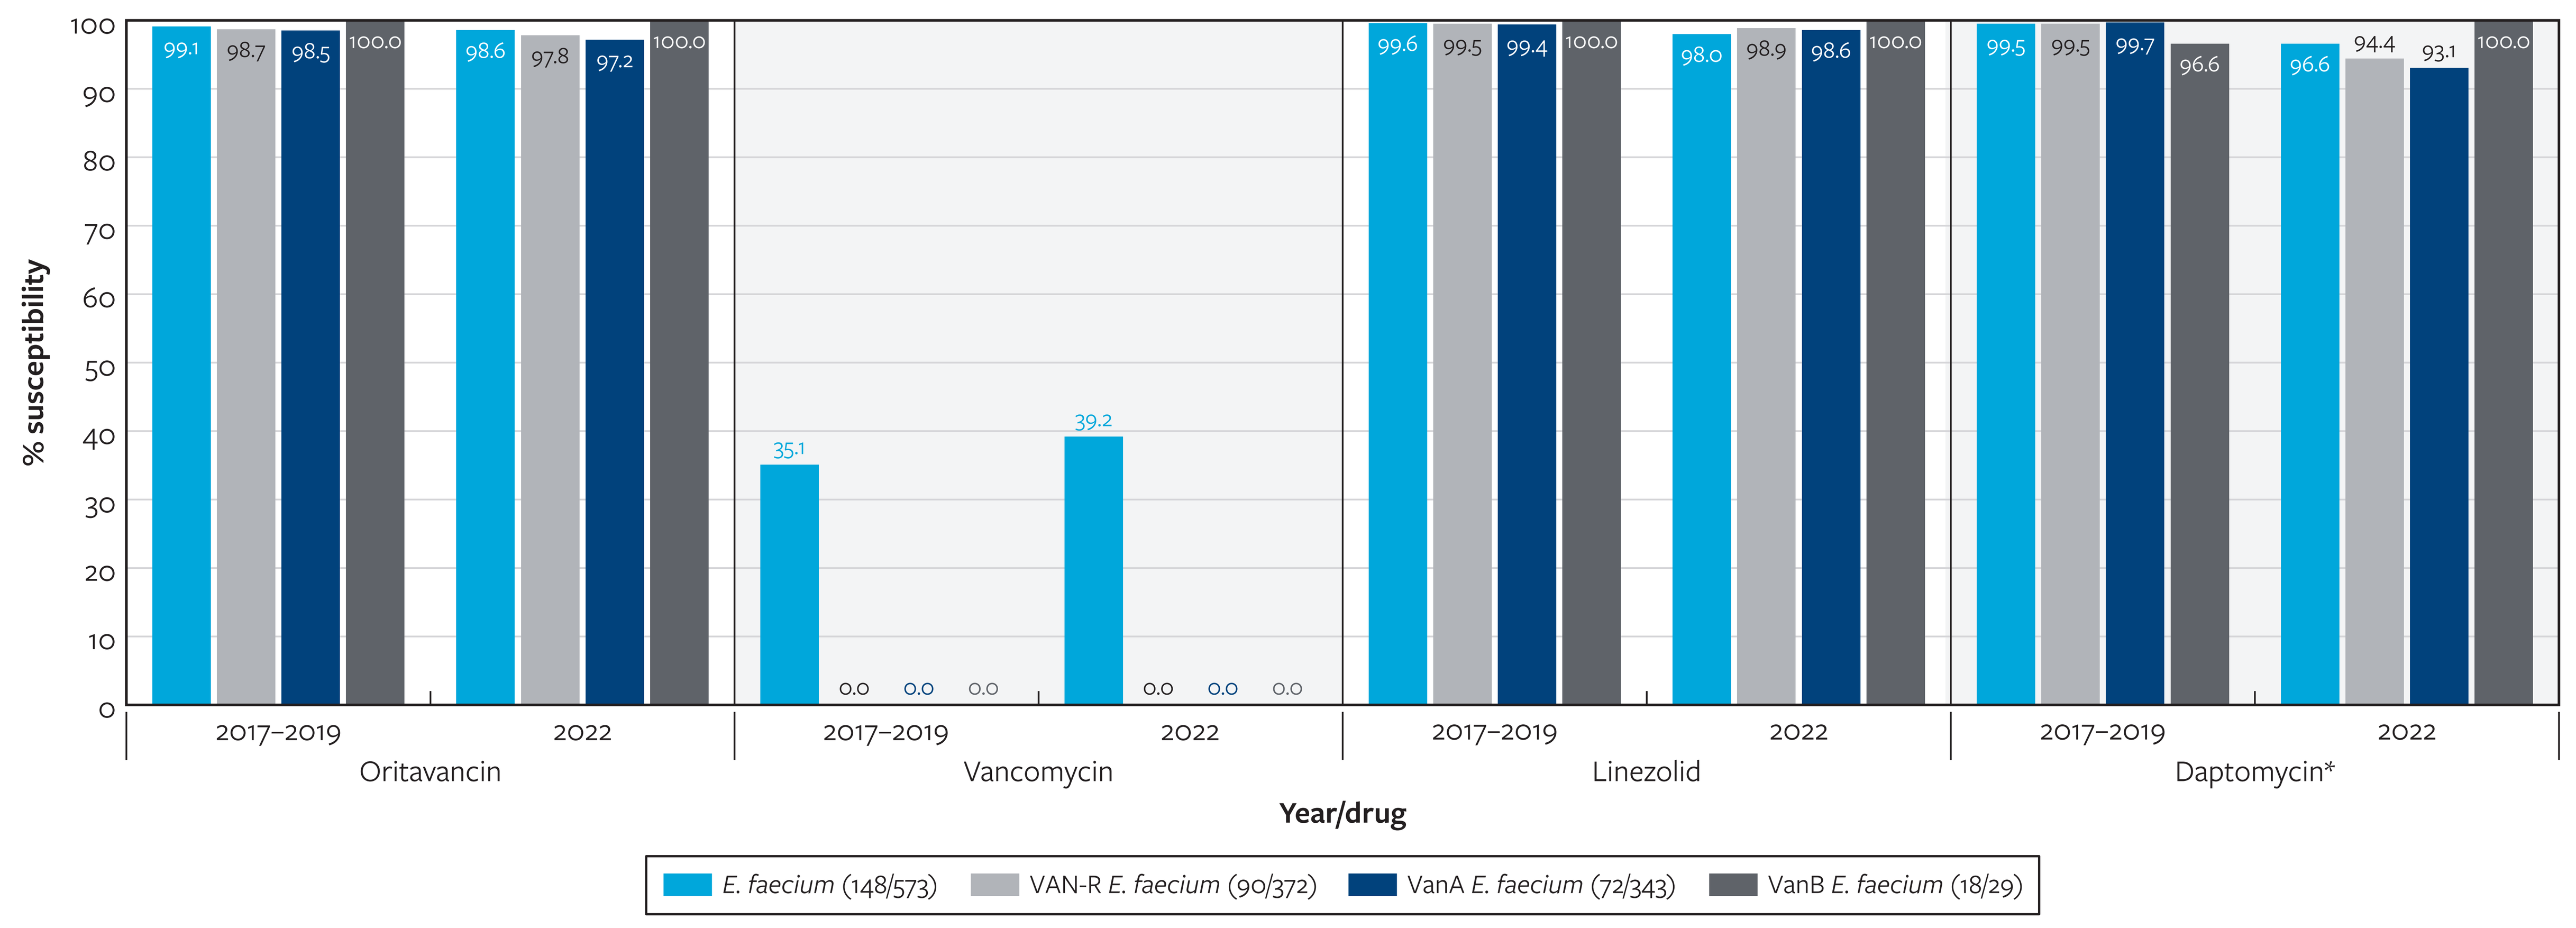
% susceptibility
100
90
80
70
60
50
40
30
20
10
0
99.1
98.7
98.5
100.0
98.6
97.8
97.2
100.0
35.1
0.0
0.0
0.0
39.2
0.0
0.0
0.0
99.6
99.5
99.4
100.0
98.0
98.9
98.6
100.0
99.5
99.5
99.7
96.6
96.6
94.4
93.1
100.0
2017–2019
2022
Oritavancin
Vancomycin
Linezolid
Daptomycin*
Year/drug
E. faecium (148/573)
VAN-R E. faecium (90/372)
VanA E. faecium (72/343)
VanB E. faecium (18/29)

* Daptomycin bars represent the susceptible-dose dependent (SDD) category of MIC values ≤4 mg/L.

Table 1. Activity of oritavancin and comparator agents against *Enterococcus* spp collected from US medical centers in 2017–2019 and 2022

Organism group (no. of isolates 2017–2019/2022)	2017–2019								2022							
	Oritavancin		Vancomycin		Linezolid		Daptomycin		Oritavancin		Vancomycin		Linezolid		Daptomycin	
	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}	MIC_{50}	MIC_{90}
Enterococcus spp. (1,936/408)	0.015	0.06	1	>16	1	2	1	2	0.015	0.03	1	>16	1	2	1	2
E. faecalis (1,283/246)	0.015	0.03	1	2	1	2	0.5	1	0.015	0.03	1	2	1	2	1	1
VAN-R *E. faecalis* (32/7)	0.12	0.5	>16	>16	1	2	0.5	1	0.12	—	>16	—	1	—	0.5	—
VanA *E. faecalis* (30/6)	0.12	0.5	>16	>16	1	2	0.5	1	0.12	—	>16	—	1	—	0.5	—
VanB *E. faecalis* (2/1)	0.015	—	>16	—	1	—	0.5	—	0.015	—	>16	—	0.5	—	0.5	—
E. faecium (573/148)	0.015	0.06	>16	>16	1	2	1	2	0.008	0.06	>16	>16	1	2	1	2
VAN-R *E. faecium* (372/90)	0.03	0.06	>16	>16	1	2	1	2	0.015	0.06	>16	>16	1	2	1	2
VanA *E. faecium* (343/72)	0.03	0.06	>16	>16	1	2	1	2	0.03	0.06	>16	>16	1	2	1	2
VanB *E. faecium* (29/18)	0.008	0.03	>16	>16	1	2	1	2	0.004	0.015	>16	>16	1	1	1	2
Other *Enterococcus* spp. (80[a]/14[b])	0.008	0.015	0.5	8	1	2	1	2	0.004	0.015	0.5	4	1	2	0.5	0.5

[a] Organisms include in 2017–2019: *Enterococcus avium* (31), *E. casseliflavus* (16), *E. gallinarum* (17), *E. hirae* (3), and *E. raffinosus* (13).
[b] Organisms include in 2022: *Enterococcus avium* (8), *E. casseliflavus* (1), *E. gallinarum* (2), and *E. raffinosus* (3).

Table 2. Oritavancin and comparator agents' susceptibility rates against *Enterococcus* spp. and subgroups

Organism group (no. of isolates 2017–2019/2022)	2017–2019				2022			
	CLSI[a] %S				CLSI[a] %S			
	ORI[c]	VAN	LZD	DAP	ORI[c]	VAN	LZD	DAP
Enterococcus spp. (1,936/408)	98.9	78.6	99.6	[b]	97.5	76.2	99.0	[b]
E. faecalis (1,283/246)	98.7	97.5	99.8	99.4	96.7	97.2	99.6	99.2
VAN-R *E. faecalis* (32/7)	53.1	0	100	100	57.1	0	100	100
VanA *E. faecalis* (30/6)	50	0	100	100	50	0	100	100
VanB *E. faecalis* (2/1)	100	0	100	100	100	0	100	100
E. faecium (573/148)	99.1	35.1	99.6	99.5[d]	98.6	39.2	98.0	96.6[d]
VAN-R *E. faecium* (372/90)	98.7	0	99.5	99.5[d]	97.8	0	98.9	94.4[d]
VanA *E. faecium* (343/72)	98.5	0	99.4	99.7[d]	97.2	0	98.6	93.1[d]
VanB *E. faecium* (29/18)	100	0	100	96.6[d]	100	0	100	100.0[d]
Other *Enterococcus* spp. (80[e]/14[f])	100	87.5	100	97.5	100	100	100	100

Abbreviations: ORI, oritavancin; VAN, vancomycin; LZD, linezolid; DAP, daptomycin
[a] Criteria as published by CLSI (2023).
[b] Values are not shown because breakpoints are different among organism species (CLSI 2023).
[c] Vancomycin-susceptible *E. faecalis* breakpoints have been applied to all *Enterococcus*.
[d] Using daptomycin susceptible-dose dependent (SDD) category of MIC values ≤4 mg/L.
[e] Organisms include in 2017–2019: *Enterococcus avium* (31), *E. casseliflavus* (16), *E. gallinarum* (17), *E. hirae* (3), and *E. raffinosus* (13).
[f] Organisms include in 2022: *Enterococcus avium* (8), *E. casseliflavus* (1), *E. gallinarum* (2), and *E. raffinosus* (3).

Acknowledgments

The authors thank all the participant centers for their work in providing isolates.

References

Carvalhaes CG, Sader HS, Streit JM, Castanheira M, Mendes RE. Activity of Oritavancin against Gram-Positive Pathogens Causing Bloodstream Infections in the United States over 10 Years: Focus on Drug-Resistant Enterococcal Subsets (2010–2019). Antimicrob Agents Chemother. 2022 Feb 15;66(2):e0166721.

CLSI. 2023. M100Ed33. Performance standards for antimicrobial susceptibility testing: 33rd informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.

Hoover RK, Krsak M, Molina KC, Shah K, Redell M. Kimyrsa, An Oritavancin-Containing Product: Clinical Study and Review of Properties. Open Forum Infect Dis. 2022 Mar 23;9(5):ofac090.

Riccardi N, Monticelli J, Antonello RM, Di Lallo G, Frezza D, Luzzati R, Di Bella S. Therapeutic Options for Infections due to vanB Genotype Vancomycin-Resistant Enterococci. Microb Drug Resist. 2021 Apr;27(4):536-545.

Contact

element

Cecilia Carvalhaes, MD, Ph.D., D(ABMM)
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia.carvalhaes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2023_23-MEL-04_P3_Orita_Enterococcus.pdf

Charges may apply. No personal information is stored.

IDWeek 2023, October 11–15, 2023, Boston, Massachusetts