1694

COADMINISTRATION OF REZAFUNGIN DOES NOT IMPACT THE PHARMACOKINETICS OF CYCLOSPORINE, IBRUTINIB, MYCOPHENOLATE MOFETIL, OR VENETOCLAX

Taylor Sandison
tsandison@cidara.com
858.249.9459

Shawn Flanagan[1], Helen Walker[2], Voon Ong[1], Taylor Sandison[1]
[1]Cidara Therapeutics Inc., San Diego, CA [2]Mundipharma, Cambridge, UK

CIDARA THERAPEUTICS

BACKGROUND

Rezafungin is a novel once weekly echinocandin antifungal (Figure 1) in development for treatment of candidemia and invasive candidiasis, and for prevention of invasive fungal diseases caused by *Candida*, *Aspergillus*, and *Pneumocystis*. Vulnerable patient populations, such as those undergoing intensive chemotherapy or transplants may receive antifungal treatment or prophylaxis particularly with azole antifungals, with risks of drug-drug interactions (DDIs) that can significantly alter the pharmacokinetics (PK) of immunosuppressive or anticancer agents. This study was conducted to evaluate the effect of rezafungin on cyclosporine, mycophenolate mofetil, venetoclax, and ibrutinib.

Figure 1. Structure of rezafungin acetate

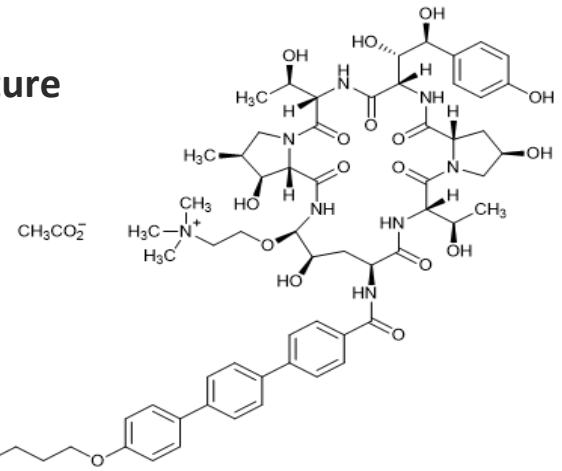

METHODS

A Phase 1, open-label study of 33 healthy subjects (17 males [Cohort 1], 16 females [Cohort 2]) was conducted to assess DDIs between rezafungin (as perpetrator) and cyclosporine 200 mg, mycophenolate mofetil 500 mg (males only), ibrutinib 280 mg, or venetoclax 50 mg (females only) as effectors. Each was given alone and with IV rezafungin (1 h infusion; 400 mg followed by once-weekly 200 mg) with a suitable washout period between dosing (Figure 2). Validated LC-MS/MS methods were used to determine plasma concentrations of mycophenolic acid, ibrutinib, venetoclax, and cyclosporine.

RESULTS

SAFETY

Overall, all study drugs were well tolerated. Ten subjects (29.4%) experienced adverse events (AEs) following the administration of the substrate drugs alone and 15 subjects (46.9%) experienced AEs for the substrate drugs with rezafungin. There were no deaths or serious adverse events. The most commonly reported AEs were headache and nausea. The majority of AEs were of mild intensity. Two subjects had one severe AE each (abdominal pain related to both rezafungin and venetoclax and esophagitis related to cyclosporine). No trends in safety laboratory results were identified.

Figure 2. Open-Label, Phase 1, DDI Study Design

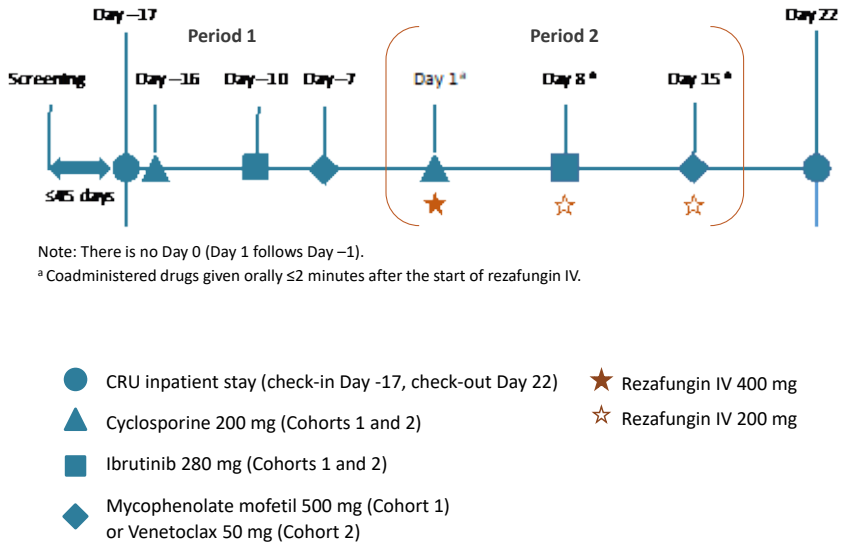

Day −17
Period 1
Period 2
Day 22
Screening
Day −16
Day −10
Day −7
Day 1ᵃ
Day 8ᵃ
Day 15ᵃ
≤45 days
Note: There is no Day 0 (Day 1 follows Day −1).
ᵃ Coadministered drugs given orally ≤2 minutes after the start of rezafungin IV.
CRU inpatient stay (check-in Day -17, check-out Day 22)
Cyclosporine 200 mg (Cohorts 1 and 2)
Ibrutinib 280 mg (Cohorts 1 and 2)
Mycophenolate mofetil 500 mg (Cohort 1) or Venetoclax 50 mg (Cohort 2)
Rezafungin IV 400 mg
Rezafungin IV 200 mg

PK

The PK of all 4 drugs were similar with and without rezafungin. Comparison of the exposure (AUC and C_{max}) of the drugs given alone or with rezafungin is presented in Table 1, with geometric mean ratio and 90% confidence intervals also shown in Figure 3. There were no clinically meaningful changes in exposure.

Parameter, unit	N: Without Rezafungin (reference)	N: With Rezafungin (test)	Intra-CV (%)	Geometric LSmeans: Without Rezafungin (reference)	Geometric LSmeans: With Rezafungin (test)	Geometric LSmeans Ratio (%)	90% CI (%): Lower Bound	90% CI (%): Upper Bound
Cyclosporine								
C_{max}, ng/mL	33	31	23.3	989.810	919.417	92.89	84.23	102.44
AUC_{0-t}, ng•h/mL	33	31	15.8	4781.06	4537.99	94.92	88.72	101.54
$AUC_{0-\infty}$, ng•h/mL	33	31	16.0	5001.93	4740.20	94.77	88.50	101.47
Mycophenolic Acid								
C_{max}, ng/mL	16	16	44.4	9541.20	7740.58	81.13	62.91	104.63
AUC_{0-t}, ng•h/mL	16	16	16.6	25299.7	25054.9	99.03	89.42	109.68
$AUC_{0-\infty}$, ng•h/mL	13	13	14.6	27954.2	28257.0	101.08	90.96	112.33
Ibrutinib								
C_{max}, ng/mL	32	31	35.3	78650.01	65547.61	83.34	71.92	96.58
AUC_{0-t}, ng•h/mL	32	31	21.2	274272.45	240725.76	87.77	80.19	96.07
$AUC_{0-\infty}$, ng•h/mL	32	31	20.6	277456.39	243135.30	87.63	80.25	95.69
Venetoclax								
C_{max}, ng/mL	16	14	20.0	253.512	240.437	94.84	83.08	108.26
AUC_{0-t}, ng•h/mL	16	14	19.3	3004.70	2693.83	89.65	78.92	101.85
$AUC_{0-\infty}$, ng•h/mL	16	14	18.3	3082.58	2781.57	90.24	79.91	101.89

N=Number; CI=confidence interval; CV=coefficient of variation expressed as a percentage; LSmeans = Least Squares Means.

RESULTS

Figure 3. Forest Plot of Rezafungin DDI Study Results
(ratio is drug + rezafungin/drug alone)

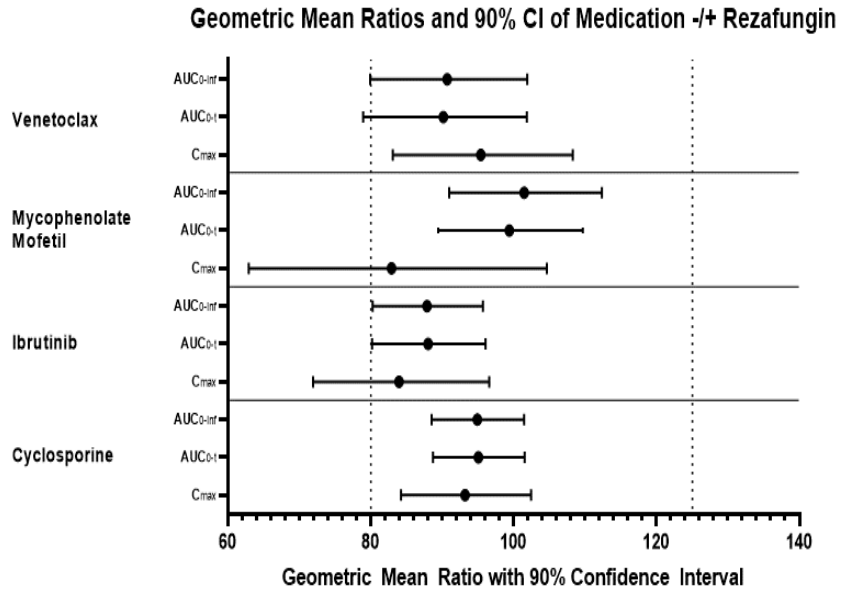

Geometric Mean Ratios and 90% CI of Medication -/+ Rezafungin
Venetoclax
Mycophenolate Mofetil
Ibrutinib
Cyclosporine
AUC0-inf
AUC0-t
Cmax
60
80
100
120
140
Geometric Mean Ratio with 90% Confidence Interval

CONCLUSIONS

Once weekly IV administration of rezafungin with single doses of cyclosporine, ibrutinib, mycophenolate mofetil and venetoclax did not result in any clinically meaningful change in the exposure of the concomitant medications. No dose adjustments of cyclosporine, mycophenolate mofetil, ibrutinib, or venetoclax are expected to be necessary when given in combination with rezafungin.

The administration of rezafungin 400 mg followed by 2 once weekly doses of 200 mg, coadministered with cyclosporine, ibrutinib, and mycophenolate mofetil or venetoclax, in healthy subjects was considered generally well-tolerated with an acceptable safety profile.

DISCLOSURES

This study was funded by Cidara Therapeutics.
SF, TS, and VO are employees and shareholders of Cidara Therapeutics. HW is a consultant of Mundipharma.