Poster 44E (shown at IDWeek 2022, Poster 469)

Healthcare Resource Utilization among Hospitalized Patients with Candidemia and Invasive Candidiasis without Candidemia Who Received Definitive Treatment with an Echinocandin Across United States (US) Hospitals

Thomas P. Lodise[1], Kevin W. Garey[2], Brian H. Nathanson[3]

[1]Albany College of Pharmacy and Health Sciences, Albany, NY, [2] University of Houston College of Pharmacy, Houston, TX, [3]OptiStatim, LLC, Longmeadow, MA

Contact:
Tom Lodise, Pharm.D., Ph.D.
Professor, Pharmacy Practice
Albany College of Pharmacy
Albany, New York, 12208-3492
Email: thomas.lodise@acphs.edu

ABSTRACT

Background: Guidelines recommend Echinocandins (EC) as preferred agents for candidemia (C) and invasive candidiasis without candidemia (IC). Although studies demonstrate that the ECs are safe and effective for the treatment of patients with C/IC, there are scant healthcare resource utilization data [HRU] (e.g., costs, length of stay) among hospitalized adult patients (pts) who received an EC for C/IC. This study sought to describe current EC use patterns and outcomes for C/IC across US hospitals.

Methods: A retrospective, multi-centered observational study was performed using the Premier Healthcare Database (1/2016-4/2019). Inclusion criteria: hospitalized; age ≥ 18 years, presence of *Candida sp*. on clinical culture consistent with C/IC; and received ≥3 days of an EC between -2 days of index culture to discharge. Patients were stratified by presence of C/IC. Baseline characteristics and treatment patterns (EC received, receipt of EC in relation to index culture, and EC duration) were assessed. Outcomes: discharge status (in-hospital death vs discharge location), hospital length of stay (LOS) post index culture, and hospital costs (overall and component costs) post index culture.

Results: 1,865 pts met study criteria. The mean (SD) age was 58.9 (19), 48% were female, mean (SD) Charlson Comorbidity Index was 3.4 (2.7) and 55% resided in the ICU at index culture. The most common *Candida sp*. were *C. albicans* (37%), *C. glabrata* (28%), *C. parapsilosis* (11%), and *C. tropicalis* (10%). Most pts had C (66%). Baseline characteristic and treatment patterns were largely similar between C/IC pts except for *Candida sp.,* EC received, and EC duration (**Table 1**). In-hospital mortality was higher in pts with C vs IC (**Figure 1**). Mean HRU was greater in pts with IC vs C (**Figures 2-4**). No differences in HRU were observed in pts who died vs survived. Most pts with C/IC received additional medical care post-discharge and pts with IC vs C were more likely to be discharged to a home health agency.

Conclusions: Hospital costs associated with C/IC are substantial, with most attributable to room and board costs. In-hospital mortality was considerable for pts with C/IC and many pts with C/IC required additional medical care in a long-term care facility or with a home health agency post-discharge. New treatment options are needed to mitigate the costs and outcomes associated with daily receipt of EC for pts with C/IC.

BACKGROUND

- Expert guidelines recommend echinocandins (EC) as first-line agents for candidemia (C) and invasive candidiasis without candidemia (IC) due to their superior response rates, increasing prevalence of azole-resistant *Candida sp*., favorable safety profiles, and few drug-drug interactions (PMID: 26679628).
- While randomized clinical trials demonstrate that the echinocandins are highly effective and safe for the treatment of patients with C/IC, there are scant healthcare resource utilization data among hospitalized adult pts who received an EC for C/IC.
- Limited data are available on in-hospital mortality rates and discharge destination locations of survivors among hospitalized patients with C/IC who received definitive treatment with an EC.

OBJECTIVES

- Determine current EC use patterns among hospitalized patients with C/IC who received definitive treatment with an EC.
- Quantify healthcare utilization and costs among hospitalized patients with C/IC who received definitive treatment with an EC.
- Determine in-hospital mortality rates and hospital discharge destinations of survivors among hospitalized patients with C/IC who received definitive treatment with an EC.

METHODS

Study Design and Population

- Retrospective, multi-centered observational study of hospitalized adult patients in the Premier Healthcare Database

Study Criteria

- Age ≥ 18 years old
- Inpatient; discharged between 1/2016 to 4/2019
- Identification of a *Candida sp.* on a clinical culture consistent with C/IC
- Receipt of ≥ 3 days of an EC starting on or after -2 days from index C/IC culture collection day
 - For patients with ≥1 C/IC admissions with EC treatment, only the first admission was analyzed

Clinical Characteristics

- Demographics
 - Age and sex
- Clinical characteristics
 - Charlson Comorbidity Index (CCI index and individual conditions)
 - Hospital length of stay (LOS) prior to index C/IC culture collection day
 - Residence in ICU on index C/IC culture collection day
 - Infection type (C vs IC)
 - *Candida sp.* on index culture
 - Timing of EC treatment initiation in relation to index C/IC culture
 - Empiric (-2-2 days of index culture)
 - Early Targeted (3-6 days post index culture)
 - Late Targeted (7 -10 days post index culture)
 - Salvage (11+ days post index culture)
 - Duration of EC treatment

Outcomes

- Hospital length of stay (LOS) post index C/IC culture
- Hospital costs (overall and component costs) post index C/IC culture
- In-hospital mortality
- Discharge destination among survivors

Descriptive Analyses

- Baseline characteristics and outcomes were reported by presence of C vs IC.
 - Mean costs were stratified by timing of EC in relation to index C/IC culture

RESULTS

Table 1. Comparison of Baseline Characteristics and Treatment Patterns and Outcomes Between Patients with C/IC

Baseline Characteristics/ EC Patterns	Candidemia N = 1,239	IC N = 626
Age; Mean (SD)	58.3 (16.6)	60.2 (14.6)
Female	590 (47.6%)	311 (49.7%)
Mean (SD) Charlson Comorbidity Index	3.5 (2.7)	3.3 (2.6)
Mean (SD) hospital LOS Prior to C/IC	7.7 (10.9)	7.8 (9.8)
Residence in ICU on Index IC/C Day	678 (54.7%)	351 (56.1%)
Candida sp.		
C. albicans	409 (33.0%)	281 (28.9%)
C. glabrata	343 (27.7%)	181 (28.9%)
C. parapsilosis	176 (14.2%)	38 (6.1%)
C. tropicalis	126 (10.2%)	59 (9.4%)
Other	211 (17.0%)	137 (21.9%)
Echinocandin Received		
Caspofungin	289 (23.3%)	125 (20.0%)
Micafungin	902 (72.8%)	500 (79.9%)
Anidulafungin	57 (4.6%)	4 (0.6%)
EC Treatment Initiation Relative to Index C/IC Day		
Empiric (-2-2 days of C/IC)	867 (70.0%)	310 (49.5%)
Early Targeted (3-6 days post C/IC)	342 (27.6%)	231 (36.9%)
Late Targeted (7-10 days post C/IC)	22 (1.8%)	49 (7.8%)
Salvage (11+ days post C/IC)	8 (0.7%)	36 (5.8%)
Mean (SD) EC Days Post Index Culture	8.9 (7.4)	10.0 (9.0)

Figure 1. Comparison of Discharge Destination Between Patients with C/IC

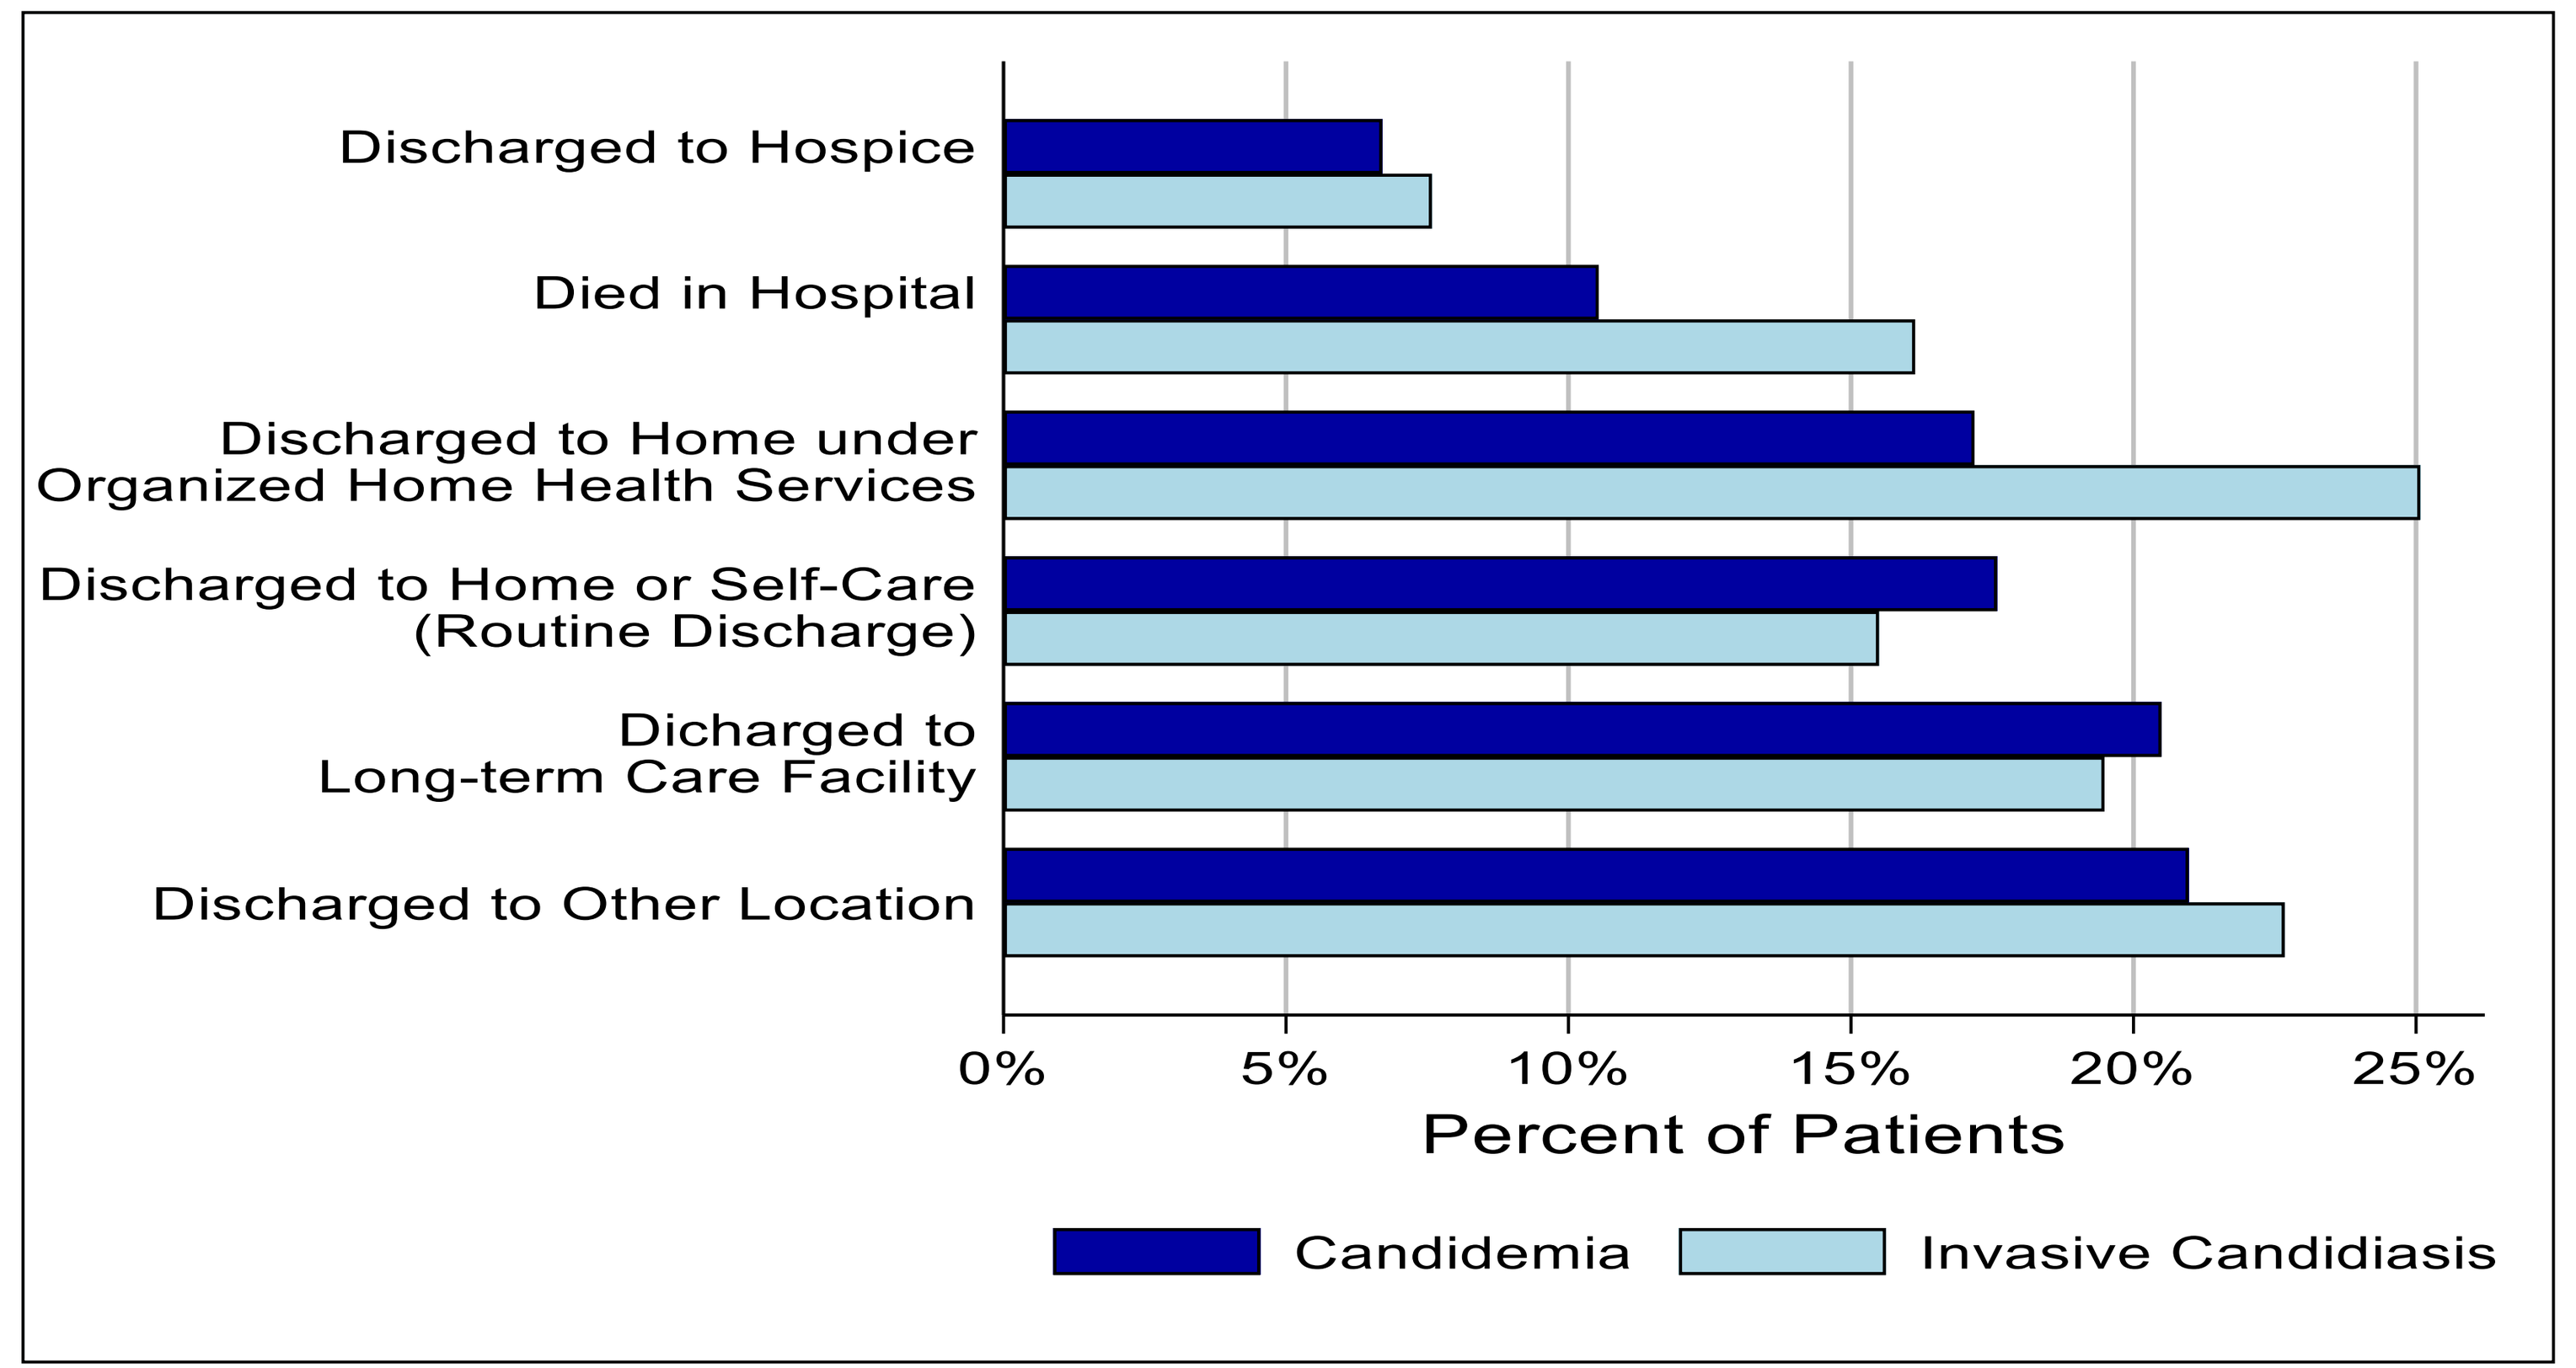

Discharged to Hospice
Died in Hospital
Discharged to Home under Organized Home Health Services
Discharged to Home or Self-Care (Routine Discharge)
Dicharged to Long-term Care Facility
Discharged to Other Location
0%
5%
10%
15%
20%
25%
Percent of Patients
Candidemia
Invasive Candidiasis

Figure 2. Comparison of Mean Hospital Length of Stay Post Index Culture Day Between Patients with Candidemia and Invasive Candidiasis without Candidemia

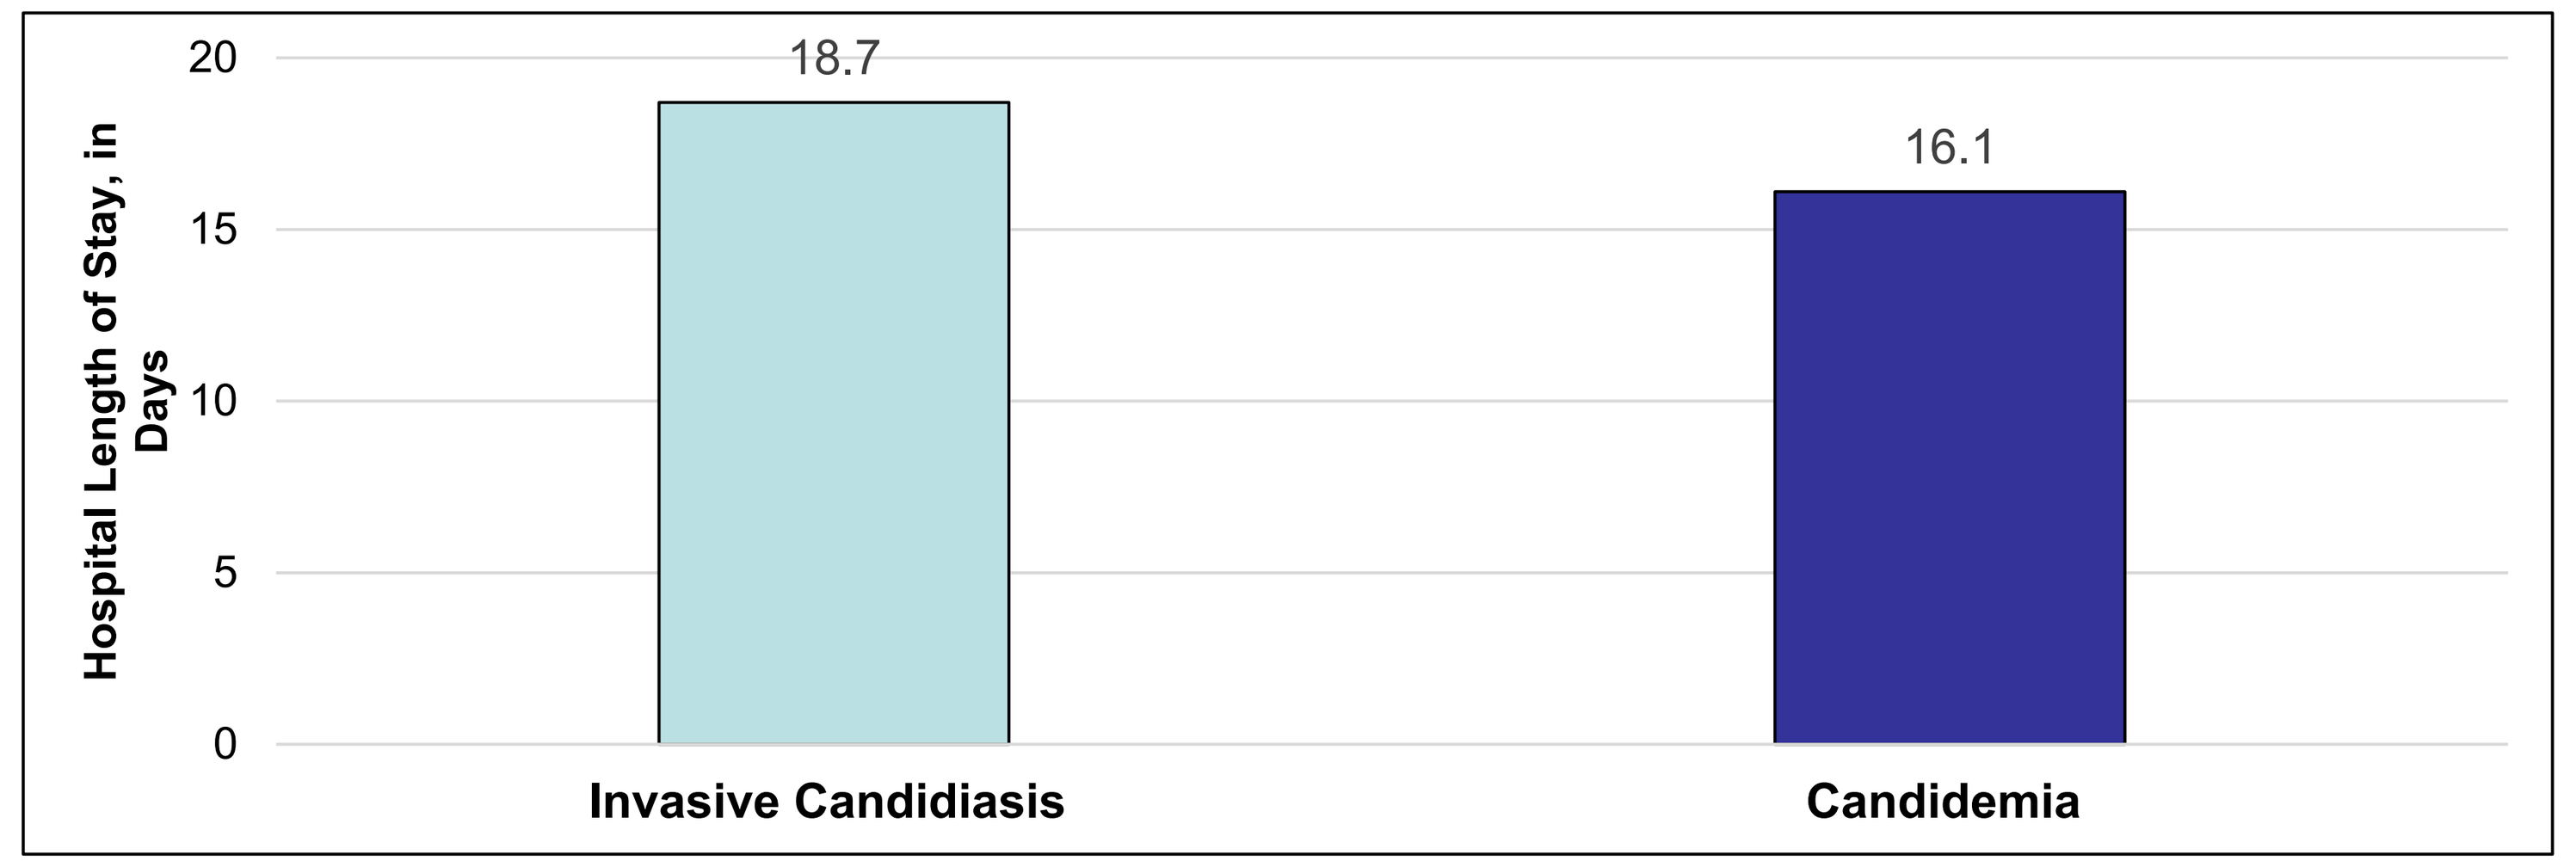

Hospital Length of Stay, in Days
0
5
10
15
20
18.7
16.1
Invasive Candidiasis
Candidemia

Figure 3. Comparison of Mean Healthcare Cost Post Index Culture Day Between Patients with Candidemia and Invasive Candidiasis without Candidemia

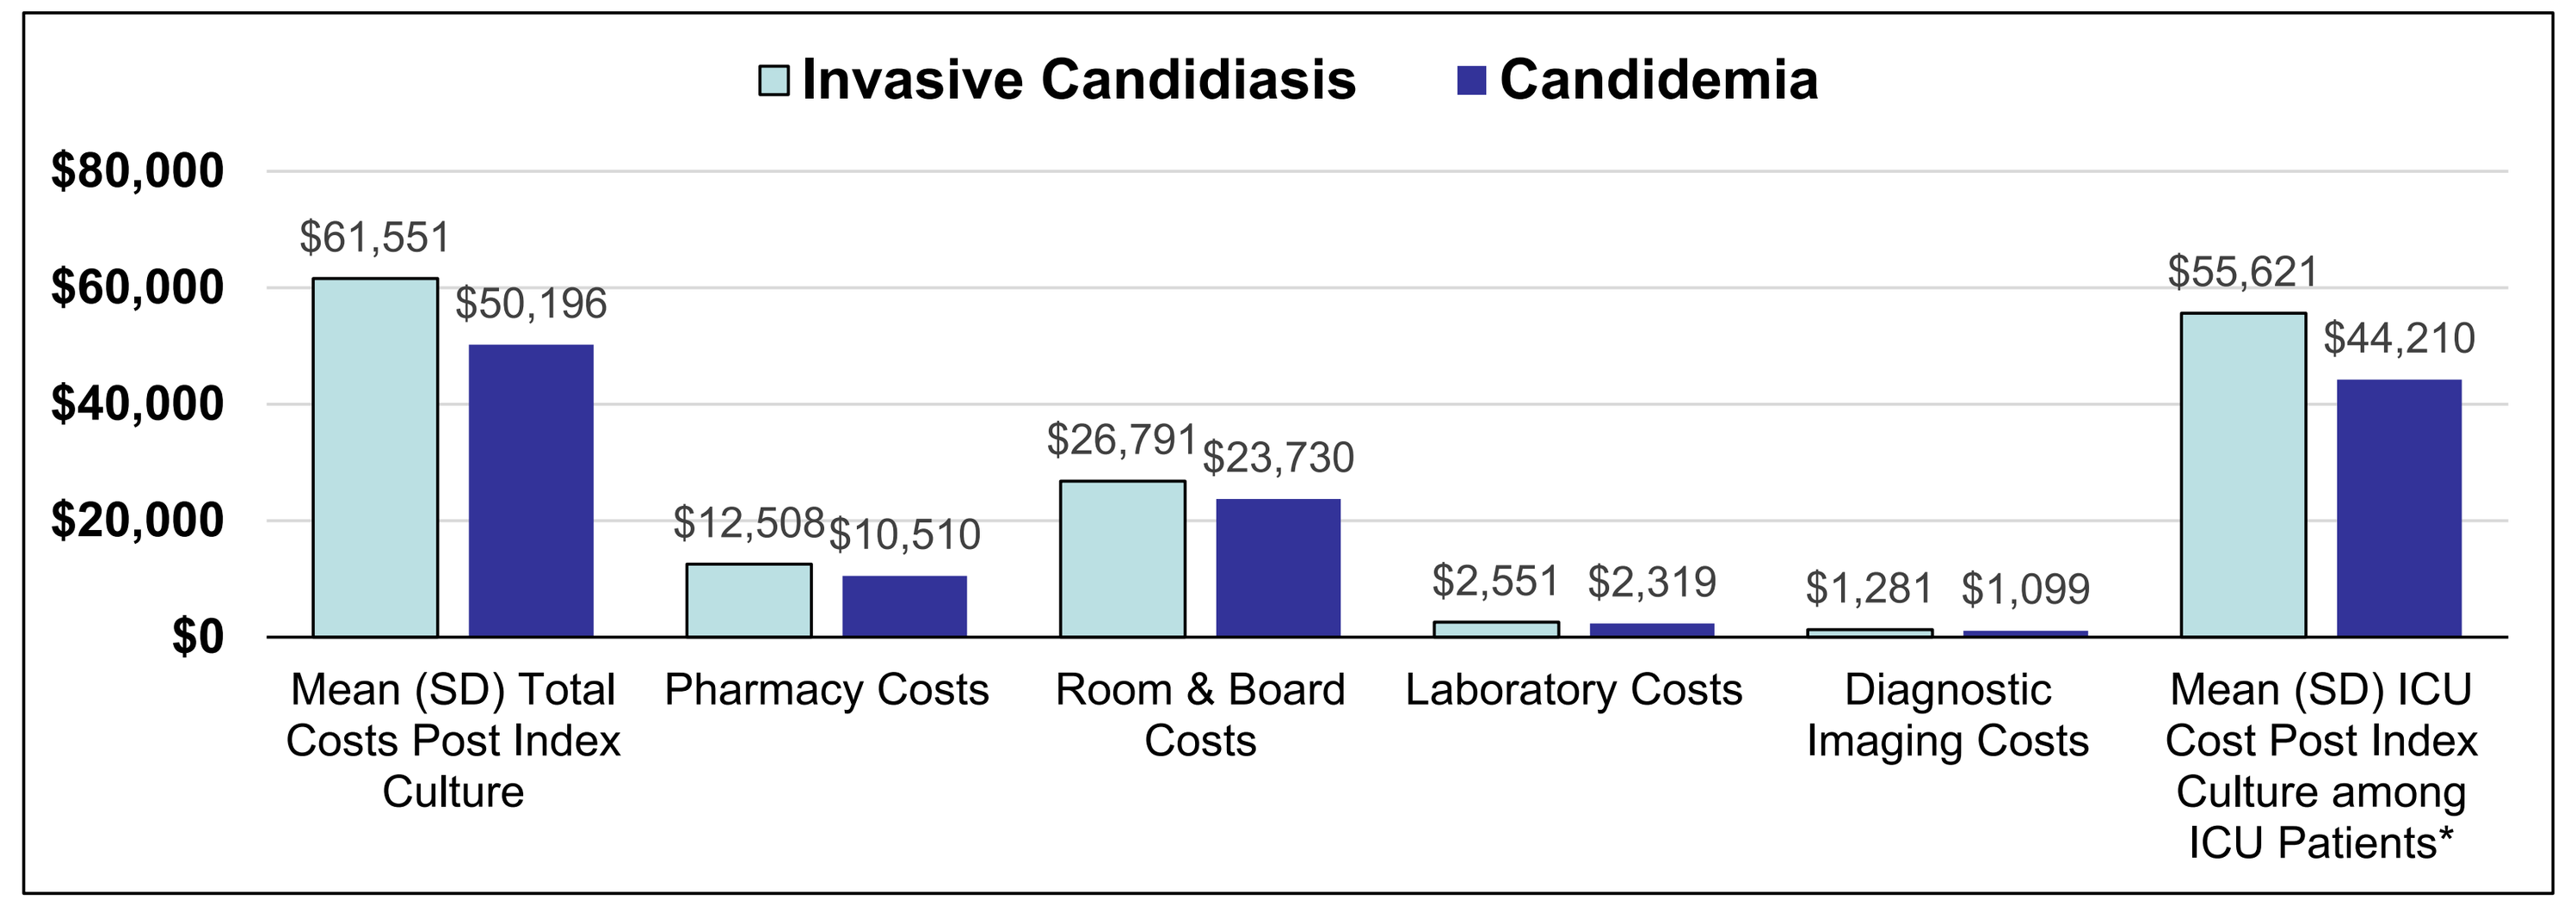

Invasive Candidiasis
Candidemia
$80,000
$60,000
$40,000
$20,000
$0
$61,551
$50,196
$12,508
$10,510
$26,791
$23,730
$2,551
$2,319
$1,281
$1,099
$55,621
$44,210
Mean (SD) Total Costs Post Index Culture
Pharmacy Costs
Room & Board Costs
Laboratory Costs
Diagnostic Imaging Costs
Mean (SD) ICU Cost Post Index Culture among ICU Patients*

Figure 4. Comparison of Mean Healthcare Cost Post Index Culture Day by Timing of EC Between Patients with Candidemia and Invasive Candidiasis without Candidemia

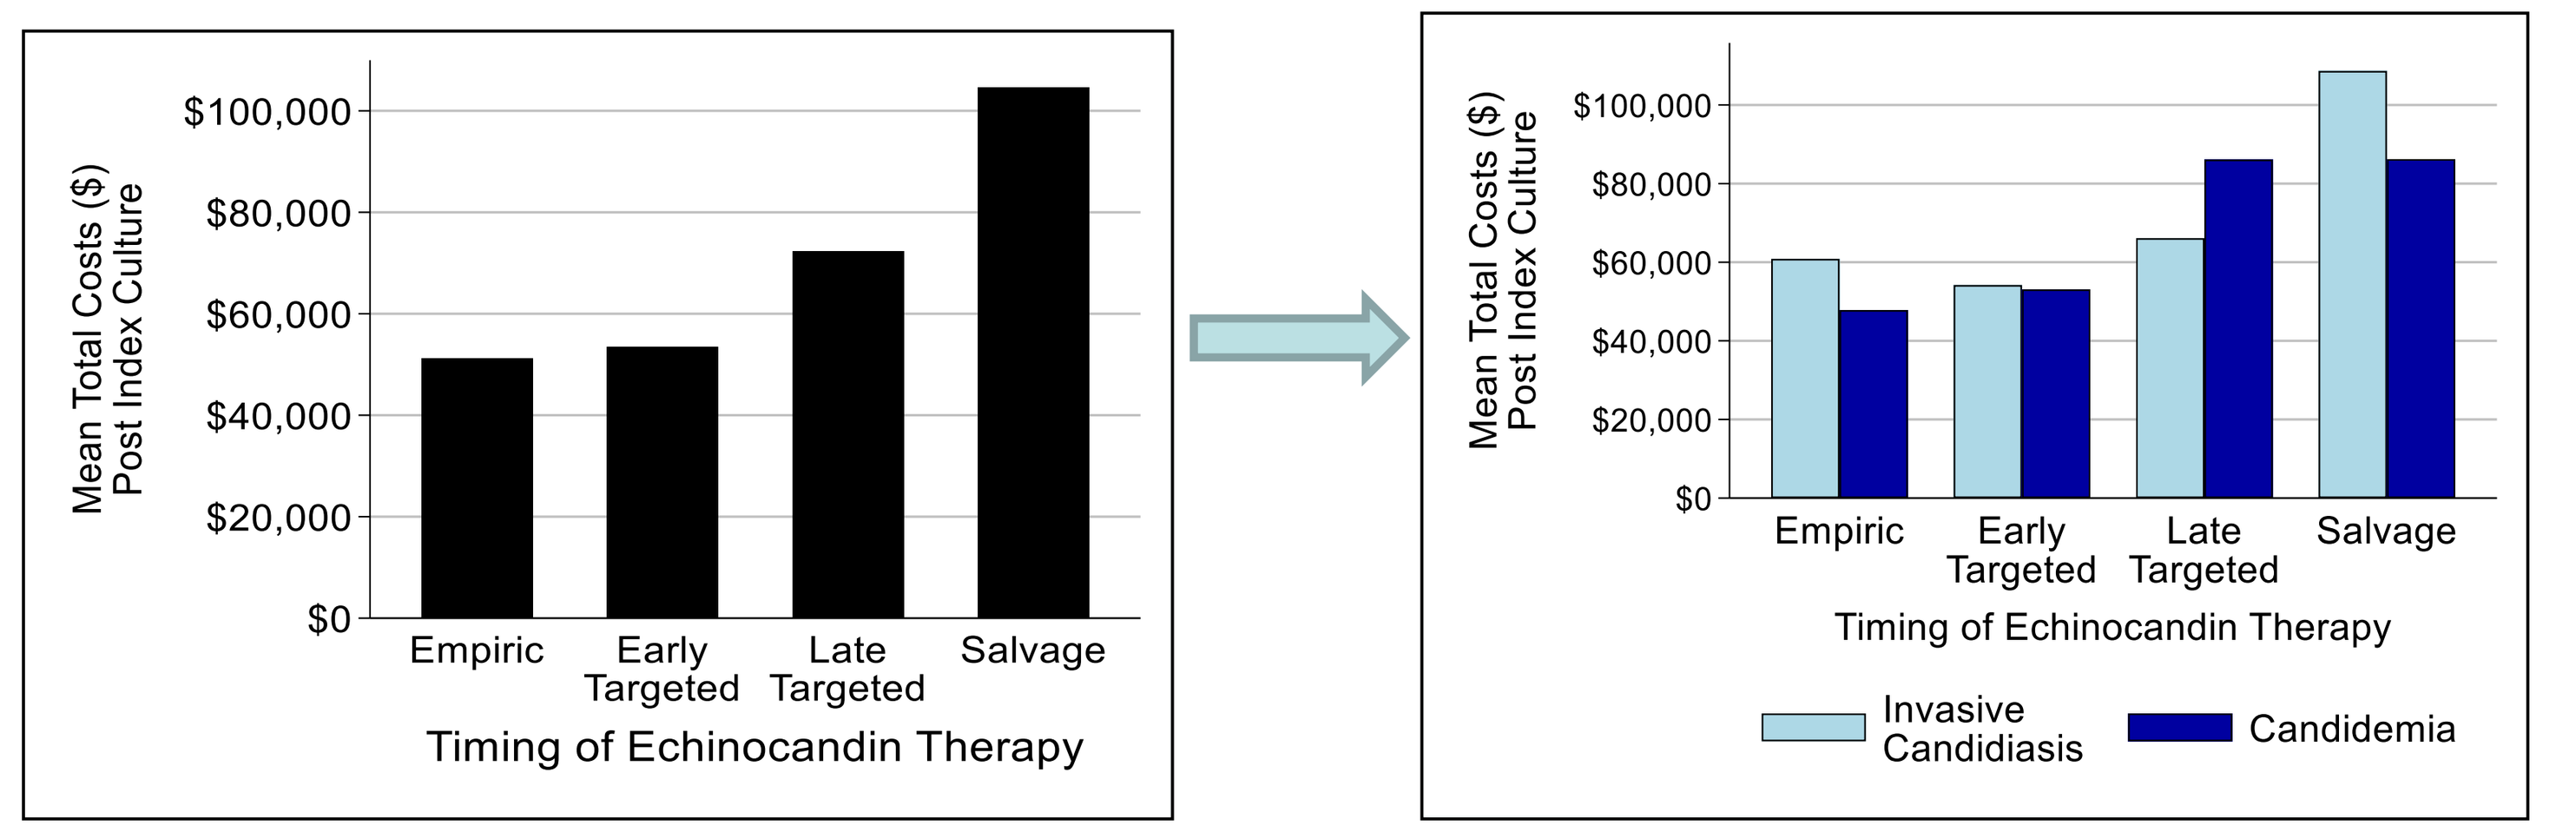

Mean Total Costs ($) Post Index Culture
$100,000
$80,000
$60,000
$40,000
$20,000
$0
Empiric
Early Targeted
Late Targeted
Salvage
Timing of Echinocandin Therapy
Invasive Candidiasis
Candidemia

CONCLUSIONS

- Hospital costs associated with C/IC are substantial, with ICU room and board costs representing the largest component.
- Hospital costs were higher among patients who received delayed EC treatment relative to those who received early EC therapy.
- In-hospital mortality was considerable for pts with C/IC and many pts with C/IC required additional medical care in a long-term care facility or with a home health agency post-discharge.
- New treatment options are needed to mitigate the costs and outcomes associated with daily receipt of EC for pts with C/IC.

Research Reported Herein was Supported by Cidara Therapeutics