IDWeek 2023 | Poster #2115

Rezafungin Activity against Invasive Candidiasis Isolates Globally: Results from the 2022 Rezafungin Surveillance Program

Cecilia G. Carvalhaes, Paul R. Rhomberg, Abby Klauer, Mariana Castanheira
Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- Rezafungin is a new echinocandin approved by the US FDA to treat candidemia and invasive candidiasis.
- Rising fluconazole resistance rates is a concern to treat invasive candidiasis, and echinocandins are often used as first-line therapy.
- Rezafungin is a once-weekly echinocandin with a long half-life and front-loaded drug exposure that differs from the usual daily regimen of other echinocandins.
- We evaluated the *in vitro* activity of rezafungin, caspofungin, micafungin, anidulafungin, and azoles against a global collection of 500 *Candida* isolates causing invasive candidiasis worldwide.

Methods

- A total of 500 *Candida* isolates causing invasive infections were collected in 2022 from 52 medical centers located in Europe (*n*=203; 19 centers), North America (*n*=167; 14 centers), Asia-Pacific (*n*=63; 8 centers), and Latin America (*n*=67; 7 centers).
- Invasive candidiasis is defined as any serious infection caused by *Candida* species.
- Candidemia was the most prevalent infection (59.2%), followed by skin and skin structure (6.6%), pneumonia in hospitalized patients (4.4%), urinary tract (3.8%), and intra-abdominal infections (1.8%).
- The site of infection was not reported for 24.2% of isolates.
- Only 1 isolate per patient was included.
- Organisms included 233 *C. albicans* (46.6%), 102 *C. glabrata* (20.4%), 82 *C. parapsilosis* (16.4%), 41 *C. tropicalis* (8.2%), 18 *C. krusei* (3.6%), 15 *C. dubliniensis* (3.0%), and 9 *C. auris* (1.8%) isolates.
- Isolates were identified by MALDI-TOF MS and/or sequencing and susceptibility tested by CLSI broth microdilution.
- CLSI breakpoints (BP) were applied where available in the M27M44S document, including rezafungin susceptible only clinical BPs (Table 1).
- CLSI BP and CDC tentative BPs were used for rezafungin and other echinocandins against *C. auris*, respectively.

Results

C. albicans

- Rezafungin exhibited activity against *C. albicans* inhibiting 99.1% of the 233 isolates overall, and 98.6%, 99.0%, 100%, and 100% of isolates from North America, Europe, Asia-Pacific, and Latin America, respectively (Table 2).
- Only 1 *C. albicans* isolate (from US) displayed a fluconazole MIC value within the susceptible-dose dependent (SDD) category but was susceptible to rezafungin and other echinocandins (Table 3).
- Caspofungin, anidulafungin, and micafungin showed susceptibility rates of 99.1% against *C. albicans*.

C. glabrata

- All but 1 *C. glabrata* isolate was susceptible to rezafungin (99.0% susceptible).
 - The rezafungin-non susceptible *C. glabrata* isolate was resistant to all echinocandins.
- *C. glabrata* susceptibility rates to caspofungin, anidulafungin, and micafungin were 98.0%, 98.0%, and 97.1%, respectively.
- Fluconazole resistance was detected in 3.9% (4) of *C. glabrata* isolates.
 - All 4 fluconazole-resistant *C. glabrata* isolates were susceptible to rezafungin and 2/4 isolates were also susceptible to other echinocandins (Table 3).

C. parapsilosis

- All *C. parapsilosis* isolates were susceptible to rezafungin, caspofungin, and micafungin.
- However, a total of 13 isolates (15.9%) were non-susceptible to fluconazole, 10 of them were resistant to fluconazole (12.2%).
 - Rezafungin and other echinocandins inhibited all 10 fluconazole-resistant isolates (9 from Europe and 1 from US; Table 3).

Other *Candida* species

- All *C. tropicalis* and *C. krusei* isolates were susceptible to rezafungin and other echinocandins.
- All *C. tropicalis* were also susceptible to azoles, and 94.4% of *C. krusei* were susceptible to voriconazole.
- Rezafungin is the only echinocandin with clinical breakpoints available by CLSI against *C. dubliniensis* (86.7% susceptibility).
- All *C. auris* isolates (9 total; 4 from Europe, 3 North America, and 2 Latin America) were fluconazole-resistant (Table 3).
 - 2 *C. auris* isolates were non-susceptible to rezafungin (77.8% susceptible) by the CLSI susceptible only criteria of ≤0.5 mg/L. One of the 2 isolates was resistant to all echinocandins and carried R1354G alteration in Fks1-HS2 sequence.
 - 1 *C. auris* isolate was resistant to anidulafungin (≥ 4 mg/L), caspofungin (≥ 2 mg/L), and micafungin (≥ 4 mg/L) per CDC tentative breakpoint (88.9% susceptible).
 - 3 *C. auris* isolates were resistant to amphotericin B per CDC tentative breakpoint (66.7% susceptible).

Conclusions

- Rezafungin demonstrated potent *in vitro* activity against invasive candidiasis isolates from North America, Europe, Asia-Pacific and Latin America medical centers.
- Rezafungin and other echinocandins remained active against fluconazole-resistant *C. parapsilosis* and *C. glabrata* isolates.
- Most of the fluconazole-resistant *C. auris* isolates were susceptible to echinocandins.
- Based on MIC_{50} and MIC_{90} results, rezafungin has similar activity to the other echinocandins overall.

Funding

This study was supported by Melinta Therapeutics. CG Carvalhaes, PR. Rhomberg, A Klauer, and M Castanheira are employees of JMI Laboratories, which was paid consultant to Melinta in connection with the development of this poster.

Acknowledgments

The authors thank all the participant centers for their work in providing isolates.

Table 1. CLSI clinical breakpoints and CDC tentative breakpoints applied in this study

Candida species	Susceptible / Resistant breakpoint criteria (CLSI, 2022) [a]					
	Rezafungin	Anidulafungin	Caspofungin	Micafungin	Fluconazole	Voriconazole
C. albicans	≤0.25 / NA	≤0.25 / ≥1	≤0.25 / ≥1	≤0.25 / ≥1	≤2 [b] / ≥8	≤0.12 / ≥1
C. glabrata	≤0.5 / NA	≤0.12 / ≥0.5	≤0.12 / ≥0.5	≤0.06 / ≥0.25	≤32 [b] / ≥64	NA
C. parapsilosis	≤2 / NA	≤2 / ≥8	≤2 / ≥8	≤2 / ≥8	≤2 [b] / ≥8	≤0.12 / ≥1
C. tropicalis	≤0.25 / NA	≤0.25 / ≥1	≤0.25 / ≥1	≤0.25 / ≥1	≤2 [b] / ≥8	≤0.12 / ≥1
C. krusei	≤0.25 / NA	≤0.25 / ≥1	≤0.25 / ≥1	≤0.25 / ≥1	NA	≤0.5 / ≥2
C. dubliniensis	≤0.12 / NA	NA	NA	NA	NA	NA
C. auris	≤0.5 / NA	NA / ≥4	NA / ≥2	NA / ≥4	NA / ≥32	NA

NA, not available
[a] CDC tentative breakpoints were applied for all antifungal agents against *C. auris* but rezafungin. CDC tentative breakpoints are available at https://www.cdc.gov/fungal/candida-auris/c-auris-antifungal.html.
[b] Susceptible-dose dependent

Table 2. Activity of rezafungin and comparator agents against worldwide collection of *Candida* isolates causing invasive infection in 2022

Organism (no. of isolates)	MIC_{50}/MIC_{90}; CLSI [a] %S					
	RZF	ANF	CSF	MCF	FLC	VRC
C. albicans (233)	0.015/0.06	0.015/0.03	0.015/0.03	0.015/0.015	0.12/0.25	0.004/0.008
	99.1	99.1	99.1	99.1	99.6	99.6
C. glabrata (102)	0.03/0.06	0.06/0.06	0.03/0.06	0.015/0.03	4/8	0.06/0.12
	99.0	98.0	98.0	97.1	96.1 [b]	NA
C. parapsilosis (82)	1/2	2/2	0.25/0.25	1/1	0.5/8	0.008/0.12
	100.0	96.3	100.0	100.0	84.1	93.9
FLC-R *C. parapsilosis* (10)	1/2	2/2	0.25/0.5	1/1	16/32	0.12/0.5
	100.0	100.0	100.0	100.0	0.0	50.0
C. tropicalis (41)	0.015/0.03	0.015/0.03	0.015/0.03	0.015/0.03	0.25/0.5	0.015/0.03
	100.0	100.0	100.0	100.0	100.0	100.0
C. krusei (18)	0.015/0.03	0.03/0.06	0.12/0.12	0.12/0.12	32/64	0.25/0.5
	100	100	100	100	NA	94.4
C. dubliniensis (15)	0.03/0.5	0.03/0.5	0.03/1	0.015/1	0.12/0.25	0.004/0.008
	86.7	NA	NA	NA	NA	NA
C. auris (9)	0.5/—	0.5/—	0.12/—	0.25/—	>128/—	1/—
	77.8	88.9	88.9	88.9	0.0	NA

Abbreviations: RZF, rezafungin; ANF, anidulafungin; CSF, caspofungin; MCF, micafungin; FLC, fluconazole; VRC, voriconazole; R, resistant; NA, not available.
[a] CLSI (2022) breakpoint was applied, including RZF breakpoint against *C. auris*. *C. auris* CDC tentative breakpoints were applied to all other antifungal agents. Breakpoints are available at https://www.cdc.gov/fungal/candida-auris/c-auris-antifungal.html.
[b] Susceptible-dose-dependent.

Table 3. Activity of rezafungin and other echinocandins against worldwide fluconazole-nonsusceptible *Candida* isolates causing invasive infections (2022)

Organism	Continent	Country	MIC (mg/L); Category of susceptibility[a]							
			Rezafungin		Anidulafungin		Caspofungin		Micafungin	
C. albicans	North America	USA	0.004	S	0.008	S	0.008	S	0.015	S
C. auris	North America	USA	1	**NS**	1	S	0.06	S	0.25	S
C. auris	Latin America	Panama	>4	**NS**	4	**R**	>4	**R**	4	**R**
C. auris	North America	USA	0.5	S	0.25	S	0.12	S	0.12	S
C. auris	North America	USA	0.5	S	0.5	S	0.12	S	0.25	S
C. auris	Europe	Greece	0.5	S	0.25	S	0.25	S	0.25	S
C. auris	Europe	Greece	0.5	S	0.5	S	0.25	S	0.25	S
C. auris	Europe	Israel	0.5	S	0.5	S	0.06	S	0.12	S
C. auris	Latin America	Panama	0.25	S	0.25	S	0.06	S	0.12	S
C. auris	Europe	Greece	0.25	S	0.25	S	0.25	S	0.12	S
C. glabrata	North America	USA	0.25	S	0.25	**I**	0.25	**I**	0.06	S
C. glabrata	Europe	Germany	0.06	S	0.06	S	0.06	S	0.03	S
C. glabrata	Europe	Spain	0.06	S	0.12	S	0.12	S	0.12	**I**
C. glabrata	Europe	Israel	0.03	S	0.06	S	0.06	S	0.03	S
C. parapsilosis	North America	USA	1	S	1	S	0.12	S	0.5	S
C. parapsilosis	Europe	Israel	1	S	1	S	0.25	S	1	S
C. parapsilosis	Latin America	Brazil	2	S	2	S	0.5	S	1	S
C. parapsilosis	Europe	Italy	1	S	2	S	0.25	S	1	S
C. parapsilosis	Europe	Italy	1	S	2	S	0.25	S	1	S
C. parapsilosis	Europe	Italy	1	S	2	S	0.25	S	1	S
C. parapsilosis	Europe	Spain	1	S	2	S	0.5	S	1	S
C. parapsilosis	Europe	Italy	1	S	1	S	0.25	S	1	S
C. parapsilosis	Europe	Spain	1	S	2	S	0.5	S	1	S
C. parapsilosis	Europe	Spain	2	S	2	S	0.25	S	1	S
C. parapsilosis	North America	USA	2	S	2	S	0.25	S	2	S
C. parapsilosis	Europe	Turkey	1	S	2	S	0.25	S	1	S
C. parapsilosis	Europe	Turkey	0.5	S	1	S	0.25	S	1	S

[a] CLSI (2022) breakpoint was applied, including RZF breakpoint against *C. auris*. *C. auris* CDC tentative breakpoints were applied to all other antifungal agents. Breakpoints are available at https://www.cdc.gov/fungal/candida-auris/c-auris-antifungal.html.

Figure 1. Distribution of *Candida* species causing invasive infections collected worldwide in 2022

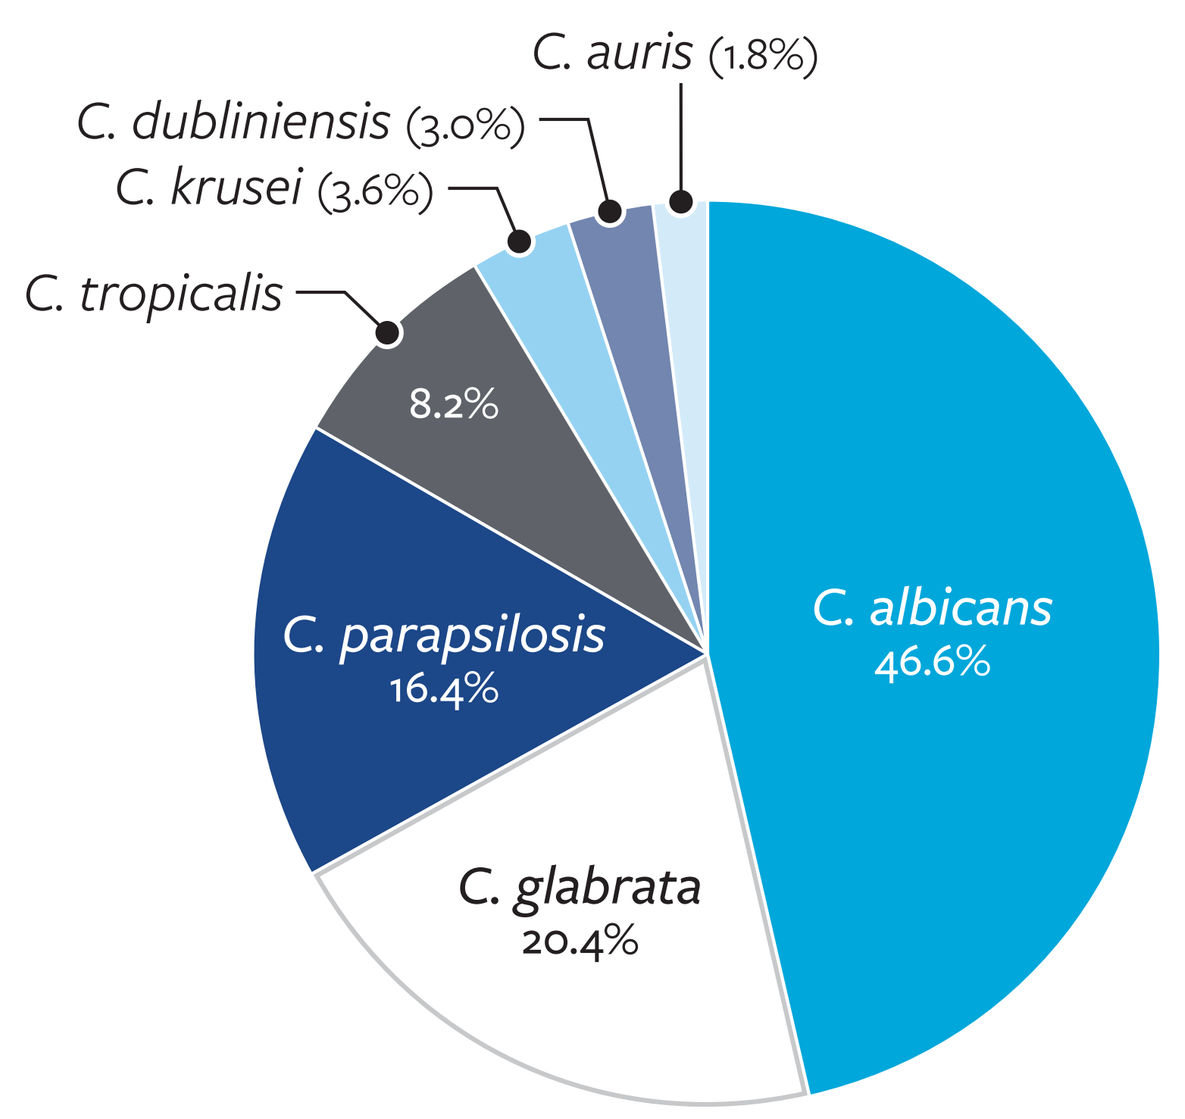
C. auris (1.8%)
C. dubliniensis (3.0%)
C. krusei (3.6%)
C. tropicalis
8.2%
C. parapsilosis
16.4%
C. glabrata
20.4%
C. albicans
46.6%

Figure 2. Distribution of invasive candidiasis isolates by region (2022)

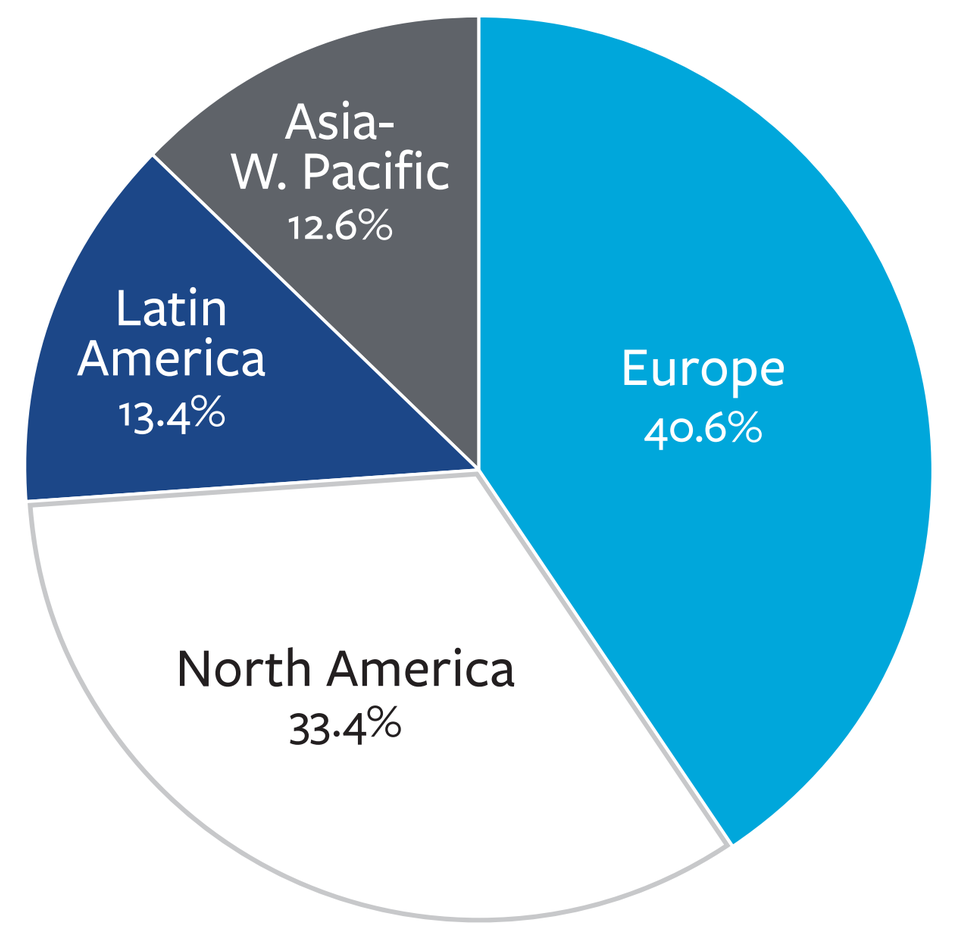
Asia-W. Pacific
12.6%
Latin America
13.4%
Europe
40.6%
North America
33.4%

Figure 3. Activity of rezafungin and other echinocandins against invasive candidiasis isolates (2022)

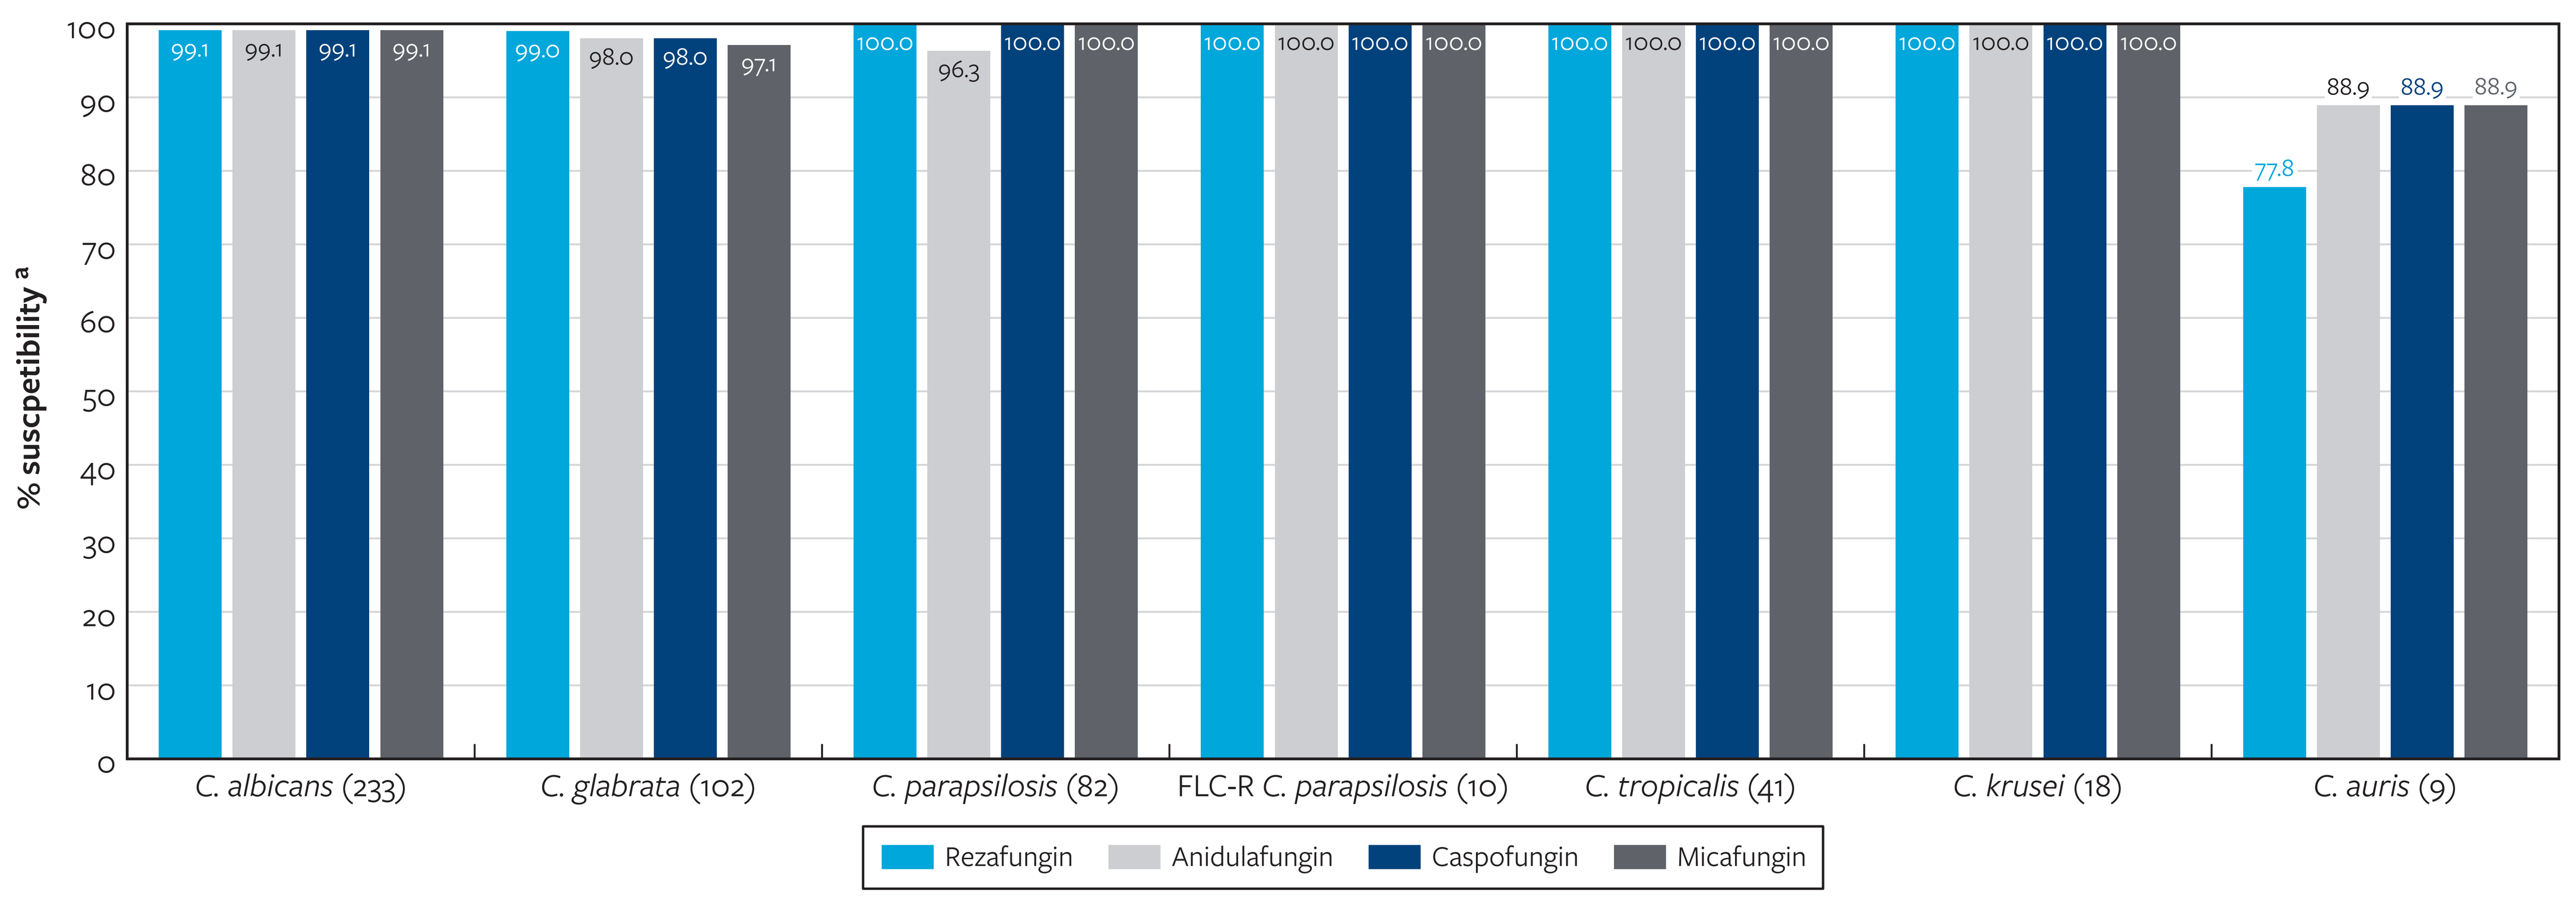
% susceptibility [a]
0
10
20
30
40
50
60
70
80
90
100
99.1 99.1 99.1 99.1
99.0 98.0 98.0 97.1
100.0 96.3 100.0 100.0
100.0 100.0 100.0 100.0
100.0 100.0 100.0 100.0
100.0 100.0 100.0 100.0
77.8 88.9 88.9 88.9
C. albicans (233)
C. glabrata (102)
C. parapsilosis (82)
FLC-R C. parapsilosis (10)
C. tropicalis (41)
C. krusei (18)
C. auris (9)
Rezafungin
Anidulafungin
Caspofungin
Micafungin

[a] CLSI (2022) breakpoint was applied, where available including RZF breakpoint against *C. auris*. *C. auris* CDC tentative breakpoints were applied to all other antifungal agents.

References

Ham YY, Lewis JS 2nd, Thompson GR 3rd. Rezafungin: a novel antifungal for the treatment of invasive candidiasis. Future Microbiol. 2021 Jan;16(1):27-36.

McCarty TP, White CM, Pappas PG. Candidemia and Invasive Candidiasis. Infect Dis Clin North Am. 2021 Jun;35(2):389-413.

Rezzayo™ package insert. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/217417s000lbl.pdf.

CLSI (2017). M27Ed4. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi. Wayne, PA.

CLSI (2022). M27M44SEd3. Performance standards for antifungal susceptibility testing of yeasts. Wayne, PA.

CDC *C. auris* tentative breakpoints available at https://www.cdc.gov/fungal/candida-auris/c-auris-antifungal.htm.

Contact

SCAN ME

Cecilia Carvalhaes, MD, Ph.D., D(ABMM)
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia.carvalhaes@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2023_23-MEL-03_P2_RZF_Surv_Candida.pdf

Charges may apply. No personal information is stored.

IDWeek 2023, October 11–15, 2023, Boston, Massachusetts