1866

Meropenem-Vaborbactam (VABOMERE) vs. Piperacillin-Tazobactam in TANGO I (a Phase 3, Randomized, Double-blind Trial): Outcomes by Baseline MIC in Adults with cUTI or AP

Thomas J. Walsh[1], Tanaya Bhowmick[2], Rabih O. Darouiche[3], Valerii I. Zaitsev[4], Evangelos J. Giamarellos-Bourboulis[5], Andrew Shorr[6], Elena Fedosiuk[7], Thomas M. File[8], Jeff Loutit[9], Olga Lomovskaya[9], Mike Dudley[9], David S. Perlin[10]

[1]Weill Cornell Medicine of Cornell University, New York, NY, USA; [2]Rutgers Robert Wood Johnson Medical School, New Brunswick, NJ, USA; [3]Baylor College of Medicine, Houston, TX, USA; [4]Bukovinian State Medical University, Chernivtsi, Ukraine; [5]National and Kapodistrian University of Athens, Medical School, Athens, Greece; [6]Medstar Washington Hospital Center/Medstar Georgetown University, Washington, DC, USA; [7]Brest Regional Hospital, Brest, Belarus; [8]Northeast Ohio Medical University, Rootstown, OH, USA; [9]The Medicines Company, San Diego, CA, USA; [10]Rutgers – New Jersey Medical School, Newark, NJ, USA

The Medicines Company
3013 Science Park Rd.
San Diego, CA 92121 USA
8 Sylvan Way
Parsippany, NJ 07054 USA

Abstract

Background

Meropenem-vaborbactam (M-V) is being developed for gram-negative infections, including complicated urinary tract infections (cUTIs) and multidrug-resistant bacterial infections. TANGO I was a Phase 3, multicenter, double-blind, randomized study of M-V vs. piperacillin-tazobactam (P-T) for treatment of adults with cUTI/acute pyelonephritis (AP). Superiority of M-V over P-T at the FDA primary endpoint has been reported. The effect of baseline MICs of Enterobacteriaceae to both study drugs and by susceptibility vs. non-susceptibility to P-T on clinical/microbiological outcome at end of intravenous treatment (EOIVT) was investigated.

Methods

MICs were conducted on baseline urinary isolates using CLSI methods. Susceptibility to P-T was defined as MIC ≤16 µg/mL (FDA/CLSI).

Results

Of 550 subjects randomized, 374 (68.0%) were included in m-MITT (165/192 [85.9%] in M-V and 154/182 [84.6%] in P-T groups had baseline Enterobacteriaceae). Mean duration of IV therapy was 8 days. Clinical outcomes were >90% across all MICs (Table).

Table. Outcomes by Baseline MIC for Enterobacteriaceae at EOIVT (m-MITT)

M-V			P-T*		
MIC (µg/mL)	Clinical Cure n/N' (%)	Microbial Eradication n/N' (%)	MIC (µg/mL)	Clinical Cure n/N' (%)	Microbial Eradication n/N' (%)
≤0.06	146/149 (98.0)	146/149 (98.0)	≤0.05	10/10 (100)	10/10 (100)
0.12	12/12 (100)	11/12 (91.7)	1	14/15 (93.3)	14/15 (93.3)
0.25	2/2 (100)	2/2 (100)	2	57/60 (95.0)	56/60 (93.3)
0.5	1/1 (100)	1/1 (100)	4	21/22 (95.5)	20/22 (90.9)
32	1/1 (100)	1/1 (100)	8	6/6 (100)	6/6 (100)
			16	10/11 (90.9)	10/11 (90.9)
			32	10/11 (90.9)	8/11 (72.7)
			64	2/2 (100)	2/2 (100)
			>64	17/17 (100)	16/17 (94.1)
Totals	162/165 (98.2)	161/165 (97.6)		147/154 (95.5)	142/154 (92.2)

* FDA/CLSI non-susceptibility breakpoint of Enterobacteriaceae to P-T is >16 µg/mL.

Although 19.5% of baseline Enterobacteriaceae isolates were non-susceptible to P-T, clinical and microbiologic outcomes were similar for subjects with susceptible and non-susceptible isolates (*P*>.1).

Conclusion

At EOIVT, clinical cure and microbial eradication rates were >90% for both groups with no trend in responses according to baseline study drug MIC. M-V is highly active against Enterobacteriaceae *in vitro* and in patients, and is a potential new treatment option for cUTI/AP.

Background

- Meropenem-vaborbactam (M-V; VABOMERE) has been developed for the treatment of serious gram-negative infections, including complicated urinary tract infections (cUTIs).
- TANGO I was a Phase 3, multicenter, double-blind, randomized study evaluating the efficacy, safety, and tolerability of M-V with that of piperacillin-tazobactam (P-T) for the treatment of adults with cUTI/acute pyelonephritis (AP). The study schema is described in **Figure 1**.

Figure 1. TANGO I Study Schema

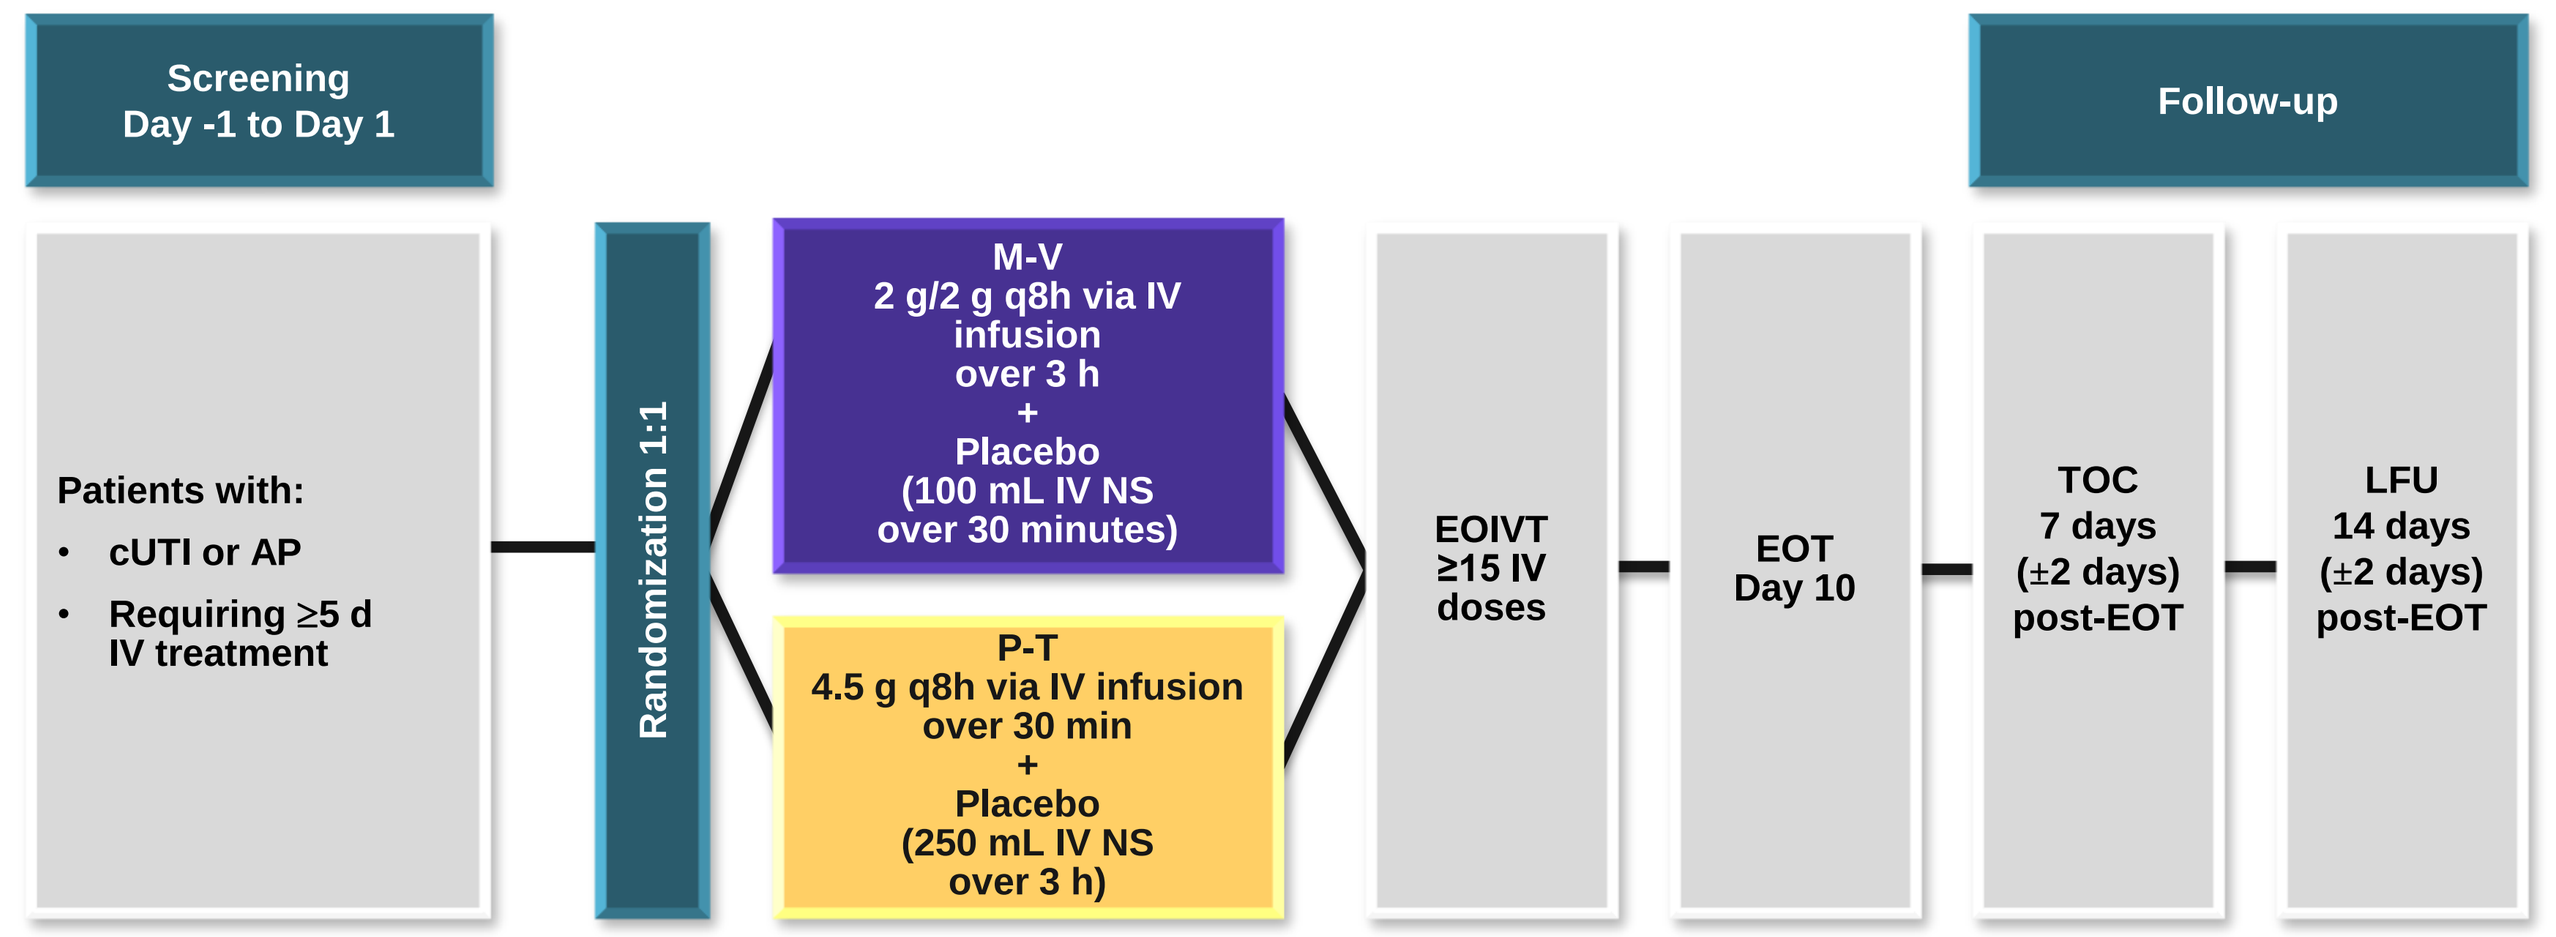

Screening
Day -1 to Day 1
Follow-up
Patients with:
cUTI or AP
Requiring ≥5 d IV treatment
Randomization 1:1
M-V
2 g/2 g q8h via IV infusion over 3 h
+
Placebo
(100 mL IV NS over 30 minutes)
P-T
4.5 g q8h via IV infusion over 30 min
+
Placebo
(250 mL IV NS over 3 h)
EOIVT
≥15 IV doses
EOT
Day 10
TOC
7 days
(±2 days)
post-EOT
LFU
14 days
(±2 days)
post-EOT

EOIVT, end of IV treatment; EOT, end of treatment; TOC, test of cure; LFU, last follow-up.
Note: For patients with cUTI, isolation of pathogens was obtained after any indwelling urinary catheter or instrumentation was removed or replaced (if removal was not clinically acceptable).

- Noninferiority was met for the Food and Drug Administration (FDA) primary endpoint of overall success in the microbiologic modified intent-to-treat (m-MITT) population at End of Intravenous Treatment (EOIVT) and for the European Medicines Agency (EMA) coprimary endpoints of microbial eradication at Test of Cure (TOC) in the m-MITT and microbiologic evaluable (ME) populations.
 - In this randomized noninferiority trial that included 550 patients, the difference in the composite outcome of complete resolution or improvement of symptoms along with microbial eradication met the noninferiority margin of 15% when comparing M-V vs. P-T (98.4% vs 94.0%).
- To assess the impact of baseline pathogen susceptibility, the effect of baseline minimum inhibitory concentrations (MICs) of Enterobacteriaceae to both study drugs, along with susceptibility vs. non-susceptibility to P-T on clinical/microbial outcome at EOIVT, were investigated.

Methods

- Patients with at least 1 baseline pathogen at ≥10^5 CFU/mL were included in the m-MITT population.
- Using Clinical and Laboratory Standards Institute (CLSI) methods, MICs were conducted on baseline urinary isolates.
- Susceptibility to P-T was defined as MIC ≤16 µg/mL (FDA/CLSI criteria).
- M-V dose and extended infusion optimizes the target attainment for pathogens with MIC up to 8 µg/mL.
- FDA assigned a breakpoint of 4 µg/mL based on extent of clinical cases seen in the Phase 3 studies.
- Clinical cure was defined (at EOIVT) as the complete resolution or significant improvement of the baseline signs and symptoms of cUTI or AP. Symptom resolution did not necessarily include baseline symptoms associated with anatomic abnormalities that predisposed to cUTI, such as symptoms associated with the presence of an indwelling urinary catheter.
- Microbial eradication was defined as meeting the following criteria:
 - Baseline bacterial pathogen(s) was reduced to <10^4 CFU/mL on urine culture (FDA criteria) or <10^3 CFU/mL (EMA criteria).

AND

 - A negative blood culture for an organism that was identified as a uropathogen (if repeated after positive at baseline blood culture).
- Differences in proportions were calculated by Chi square test or Fisher's exact test, as appropriate.

Results

- Of 550 randomized patients, 374 (68.0%) had ≥1 baseline pathogen, comprising the m-MITT population (192 in the M-V group and 182 in the P-T group).
- Mean total treatment duration in the m-MITT population was 10 days in both groups; mean duration of IV therapy was 8 days in both groups.
- Baseline pathogens in the m-MITT population are shown in **Table 1**.
 - 165/192 (85.9%) in the M-V group had baseline Enterobacteriaceae.
 - 154/182 (84.6%) in the P-T group had baseline Enterobacteriaceae.

Table 1. Baseline Pathogens in the m-MITT Population

Pathogen	M-V (N=192) n (%)	P-T (N=182) n (%)
Escherichia coli	125 (65.1)	117 (64.3)
Klebsiella pneumoniae	30 (15.6)	28 (15.4)
Enterococcus faecalis	13 (6.8)	14 (7.7)
Proteus mirabilis	6 (3.1)	12 (6.6)
Enterobacter cloacae sp. complex	10 (5.2)	5 (2.7)
Pseudomonas aeruginosa	5 (2.6)	10 (5.5)
Citrobacter freundii sp. complex	2 (1.0)	3 (1.6)
Staphylococcus saprophyticus	5 (2.6)	0 (0.0)
Morganella morganii	1 (0.5)	3 (1.6)
Acinetobacter baumannii-calcoaceticus sp. complex	1 (0.5)	2 (1.1)
Klebsiella oxytoca	2 (1.0)	1 (0.5)
Citrobacter koseri	1 (0.5)	1 (0.5)
Proteus vulgaris	1 (0.5)	1 (0.5)
Providencia rettgeri	0 (0.0)	2 (1.1)

Outcomes by baseline MIC for Enterobacteriaceae at EOIVT are shown in **Table 2**.

- The clinical cure rate at EOIVT in patients treated with P-T who had a non-susceptible Enterobacteriaceae at baseline was 29/30 (96.7%) vs. 118/124 (95.2%) in those with a susceptible organism.
- The microbial eradication rate at EOIVT in patients treated with P-T who had a non-susceptible Enterobacteriaceae at baseline was 26/30 (86.7%) vs. 116/124 (93.5%) in those with a susceptible organism (*P*>0.1).
- Of 30 Enterobacteriaceae at baseline with P-T MIC >16 µg/mL, 26 contained extended-spectrum β-lactamases (mainly CTX-M-15, but also SHV-12 in some cases), alone or in combination with broad-spectrum carbapenemases from class C (CMY, ACT) or class D (OXA-1).
- Clinical and microbial eradication rates at EOIVT in patients treated with M-V were also similar across the range of MICs.
- MICs of M-V were ≤0.05 µg/mL in >99% of all isolates, while MICs of P-T were <1.0 µg/mL in 16% of cases (95% CI: 0.006-0.31; *P*<.0001).

Table 2. Cure and Eradication Rates at End of IV Treatment in Patients with Enterobacteriaceae by Baseline MIC (m-MITT Population)

M-V			P-T		
MIC (µg/mL)	Clinical Cure n/N' (%)	Microbial Eradication n/N' (%)	MIC (µg/mL)	Clinical Cure n/N' (%)	Microbial Eradication n/N' (%)
≤0.06	146/149 (98.0)	146/149 (98.0)	≤0.05	10/10 (100)	10/10 (100)
0.12	12/12 (100)	11/12 (91.7)	1	14/15 (93.3)	14/15 (93.3)
0.25	2/2 (100)	2/2 (100)	2	57/60 (95.0)	56/60 (93.3)
0.5	1/1 (100)	1/1 (100)	4	21/22 (95.5)	20/22 (90.9)
32	1/1 (100)	1/1 (100)	8	6/6 (100)	6/6 (100)
			16	10/11 (90.9)	10/11 (90.9)
			32	10/11 (90.9)	8/11 (72.7)
			64	2/2 (100)	2/2 (100)
			>64	17/17 (100)	16/17 (94.1)
Totals	162/165 (98.2)	161/165 (97.6)		147/154 (95.5)	142/154 (92.2)

- **FDA/CLSI non-susceptibility breakpoint for P-T for Enterobacteriaceae is >16 µg/mL.**
- **Non-susceptibility breakpoint for M-V for Enterobacteriaceae is > 4 µg/mL.**

Conclusions

- At EOIVT, the FDA primary endpoint for overall success (clinical cure and microbial eradication) rates were >95% for both groups.
- The outcome of superiority of meropenem-vaborbactam over piperacillin-tazobactam for overall success at EOIVT was not driven by non-susceptibility to the comparator.
- There was no trend in response according to baseline study drug MIC or susceptibility.
- In patients with cUTI, baseline pathogen susceptibility testing may not be predictive of clinical outcomes.
- Meropenem-vaborbactam is highly active against Enterobacteriaceae *in vitro* and in patients.
- Meropenem-vaborbactam is a safe and effective new FDA-approved treatment option for cUTI/AP.

Disclosures

This project has been funded in part by The Medicines Company and the Department of Health and Human Services; Office of the Assistant Secretary for Preparedness and Response; Biomedical Advanced Research and Development Authority (BARDA), under Contract No. HHSO100201400002C with Rempex Pharmaceuticals, a wholly-owned subsidiary of The Medicines Company and Agreement No. HHSO100201600026C with The Medicines Company.

Clinicaltrials.gov Identifier: NCT02166476

References

Kaye KS, Bhowmick T, Metallidis S, et al. Effect of meropenem-vaborbactam versus piperacillin-tazobactam on clinical cure or improvement and microbial eradication among patients with complicated urinary tract infection: the TANGO I randomized clinical trial. Submitted.

The Medicines Company Announces Positive Top- Line Results for Phase 3 TANGO 1 Clinical Trial of CARBAVANCE. http://www.themedicinescompany.com/investors/news/medicines-company-announces-positive-top-line-results-phase-3-tango-1-clinical-trial. Accessed September 17, 2017.

Presented at IDWeek 2017, October 4-8, 2017, San Diego, CA